Abstract #2794

Determinants of Response of HER2+ Gastric Cancer vs Gastroesophageal Junction Adenocarcinoma to Margetuximab plus Pembrolizumab post Trastuzumab

H. Park[1], H.E. Uronis[2], Y-K. Kang[3], M. Ng[4], P.C. Enzinger[5], K.W. Lee[6], S. Rutella[7], S. Church[8], J. Nordstrom[9], Y. Yang[9], P. Moore[9], D. Li[9], K. Knutson[10], C. Erskine[10], T. Wu[9], J. Yen[11], A. Franovic[11], J. Muth[9], M. Rosales[9], J. Vadakekolathu[7], J. Davidson-Moncada[9], Y.J. Bang[12], D.V.T. Catenacci[13]

[1]Washington University Medical School, St. Louis, MO, USA; [2]Duke Cancer Center, Durham, NC, USA; [3]Asan Medical Center, University of Ulsan College of Medicine, Seoul, Republic of Korea; [4]National Cancer Centre Singapore, Singapore; [5]Dana-Farber Cancer Institute, Cambridge, MA, USA; [6]Seoul National University Bundang Hospital, Seongnam, Republic of Korea; [7]Nottingham Trent University-Clifton Campus, Nottingham, United Kingdom; [8]Nanostring, Seattle, WA, USA; [9]MacroGenics, Inc., Rockville, MD, USA; [10]Mayo Clinic, Jacksonville, FL, USA; [11]Guardant Health, Inc., Redwood City, CA, USA; [12]Seoul National University Hospital, Seoul, Republic of Korea; [13]University of Chicago Medical Center, Chicago, IL, USA

NCT02689284

Background

- Trastuzumab + chemotherapy is standard treatment in 1st line advanced HER2+ gastroesophageal adenocarcinoma (GEA); however patients tend to progress in 6–8 months
- Up to 40% show loss of HER2 expression post trastuzumab, likely underlying the lack of efficacy of anti-HER2 agents in 2nd line therapy
- Margetuximab is an investigational next generation anti-HER2 monoclonal antibody with an engineered Fc domain that confers enhanced Fc-dependent antitumor activities across all FcγRIIIA (CD16A) genotypes
- Margetuximab has demonstrated single agent antitumor activity in patients with HER2+ GEA in a Phase 1 study
- We report herein a clinical update and biomarker analysis of an ongoing study in patients receiving margetuximab plus pembrolizumab, a chemotherapy-free treatment, in HER2+ GEA patients in 2nd line post trastuzumab

Margetuximab: Fc-engineered to Activate Immune Responses

Trastuzumab

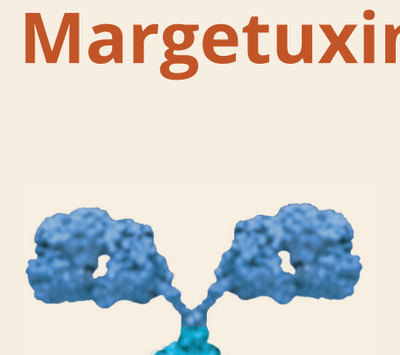

Fab:
- Binds HER2 with high specificity
- Disrupts signaling that drives cell proliferation and survival

Fc:
- Wild-type immunoglobulin G1 (IgG1) immune effector domains
- Binds and activates immune cells

Margetuximab[1,2]

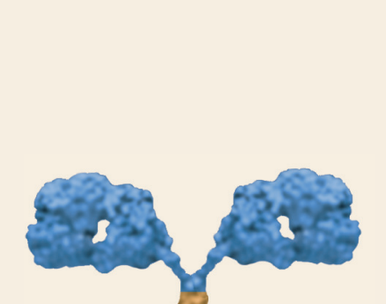

Fab:
- Same specificity and affinity
- Similarly disrupts signaling

Fc engineering:
- ↑ Affinity for activating FcγRIIIA (CD16A)
- ↓ Affinity for inhibitory FcγRIIB (CD32B)

Margetuximab Binding to FcγR Variants

Receptor Type	Receptor	Allelic Variant	Relative Fc Binding	Affinity Fold-Change
Activating	CD16A	158F	Lower	6.6 x ↑
		158V	Higher	4.7 x ↑
	CD32A	131R	Lower	6.1 x ↓
		131H	Higher	↔
Inhibitory	CD32B	232I/T	Equivalent	8.4 x ↓

[1]Nordstrom JL, et al. *Breast Cancer Res.* 2011;13(6):R123. [2]Stavenhagen JB, et al. *Cancer Res.* 2007;67(18):8882-8890.

Margetuximab Enhances Innate Immunity In Vitro

Greater relative cytotoxicity of margetuximab with NK cells from CD16A-158F allele carriers

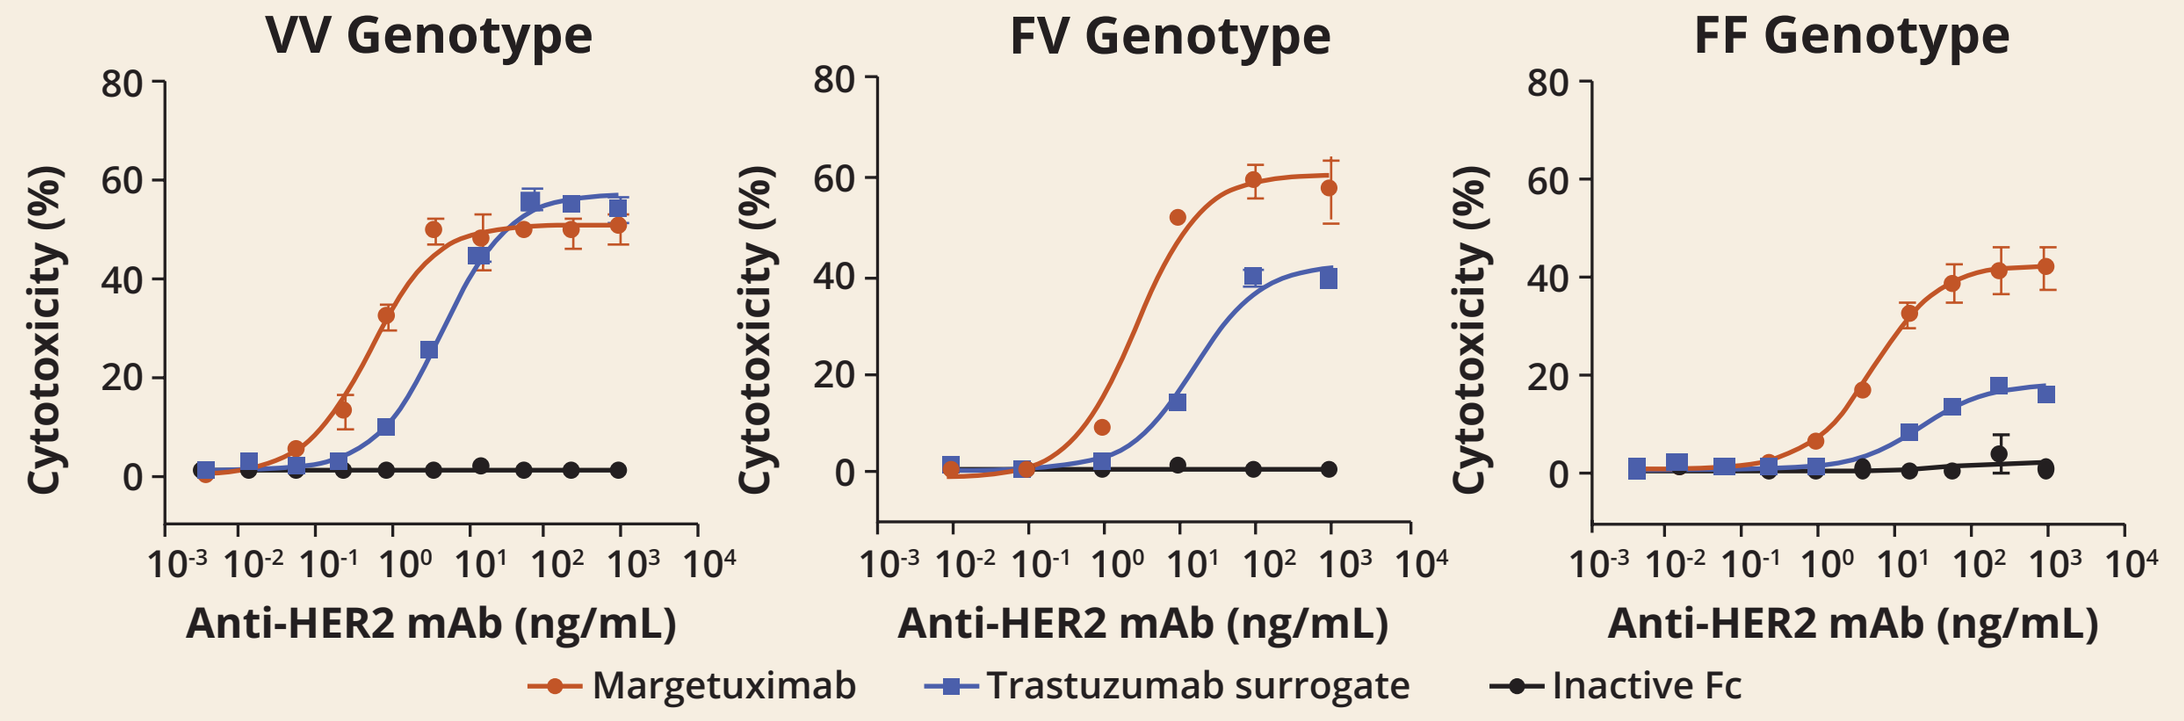
VV Genotype
FV Genotype
FF Genotype
Cytotoxicity (%)
Anti-HER2 mAb (ng/mL)
Margetuximab
Trastuzumab surrogate
Inactive Fc

Preclinical Assay of Antibody-Dependent Cellular Cytotoxicity (ADCC)[1]

- **Effector Cells:** Human NK cells from donors with CD16A genotypes 158VV, 158FV, and 158FF
- **Target Cells:** JIMT-1 HER2+ breast cancer cell line resistant to trastuzumab antiproliferative activity
- **Cellular Assay:** 3:1 Effector:Target ratio; 24-hour incubation time; endpoint: % lactate dehydrogenase release

[1]Nordstrom JL, et al. Breast Cancer Res. 2011;13(6):R123. mAb: monoclonal antibody; NK: natural killer.

Margetuximab Enhances HER2-specific Adaptive Immunity[1,2]

- Phase 1 margetuximab monotherapy study in 66 pretreated patients with HER2+ carcinomas[3,4]:
 - Four (17%) confirmed responses in 24 evaluable patients with HER2+ MBC[3]
 - Three patients continue on margetuximab at least 4 to 6 years, as of 15 May 2019[4]
- Enhanced HER2-specific T- and B-cell responses after margetuximab monotherapy[5]

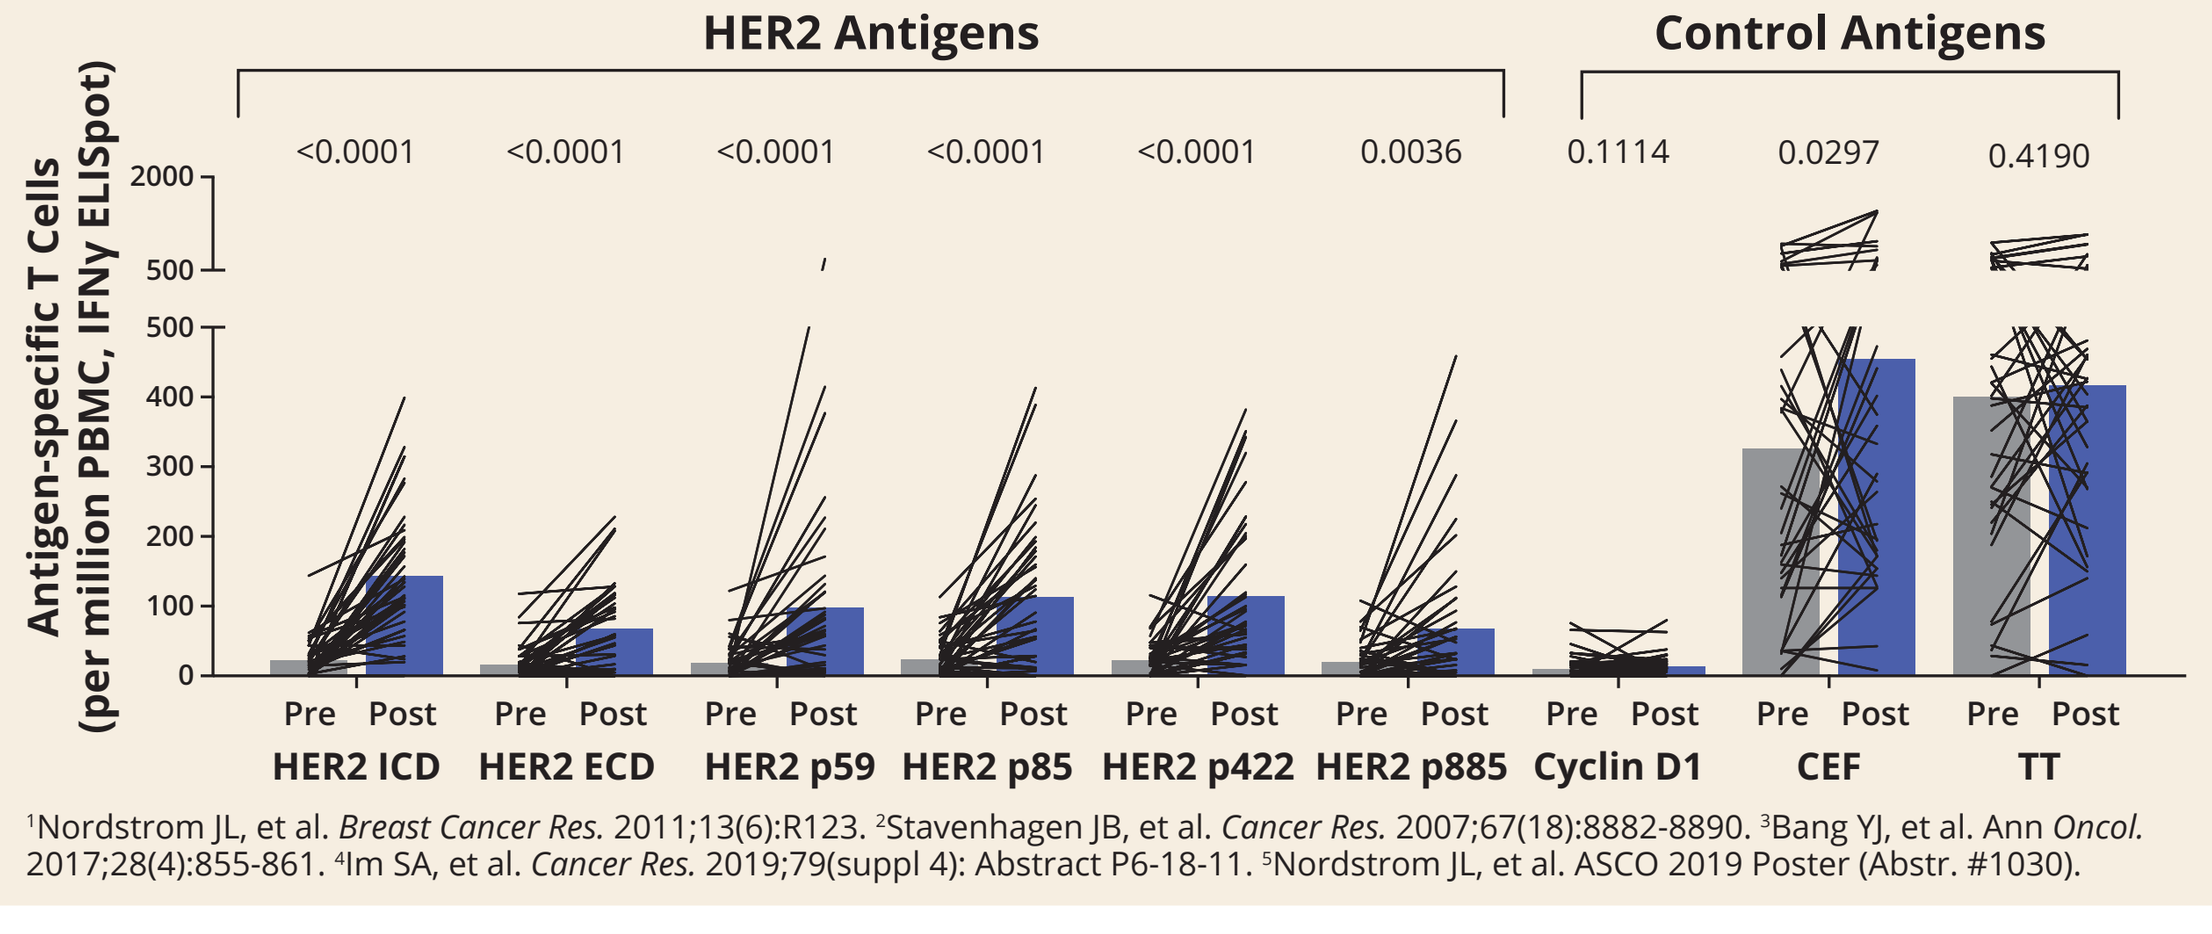
HER2 Antigens
Control Antigens
<0.0001 <0.0001 <0.0001 <0.0001 <0.0001 0.0036 0.1114 0.0297 0.4190
Antigen-specific T Cells (per million PBMC, IFNγ ELISpot)
Pre Post
HER2 ICD
HER2 ECD
HER2 p59
HER2 p85
HER2 p422
HER2 p885
Cyclin D1
CEF
TT

[1]Nordstrom JL, et al. *Breast Cancer Res.* 2011;13(6):R123. [2]Stavenhagen JB, et al. *Cancer Res.* 2007;67(18):8882-8890. [3]Bang YJ, et al. Ann *Oncol.* 2017;28(4):855-861. [4]Im SA, et al. *Cancer Res.* 2019;79(suppl 4): Abstract P6-18-11. [5]Nordstrom JL, et al. ASCO 2019 Poster (Abstr. #1030).

Anti-PD-1 Enhances Margetuximab-mediated NK Cell Cytolytic Potential In Vitro

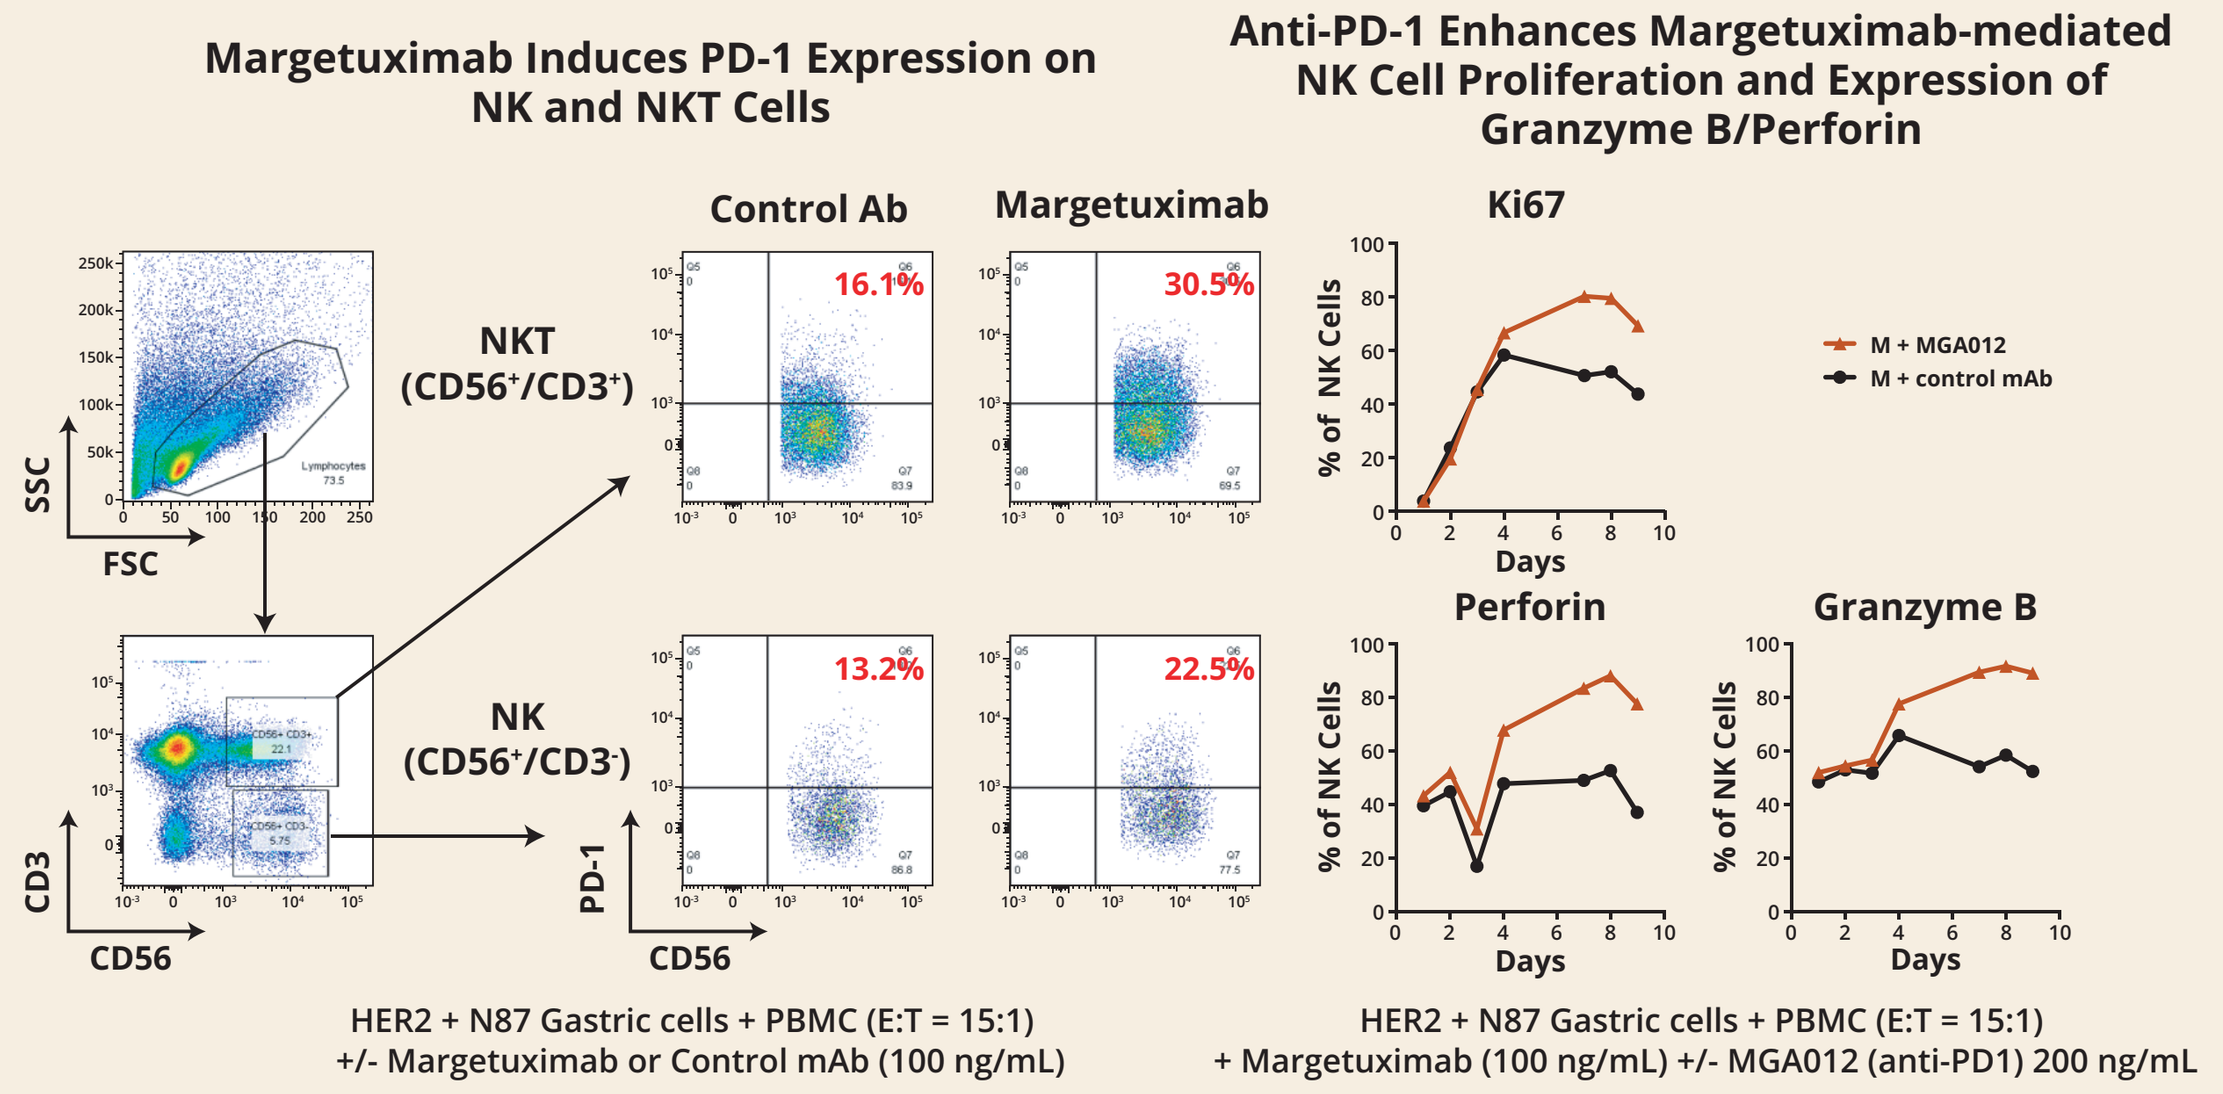
Margetuximab Induces PD-1 Expression on NK and NKT Cells
Control Ab
Margetuximab
NKT (CD56+/CD3+)
NK (CD56+/CD3-)
16.1%
30.5%
13.2%
22.5%
SSC
FSC
CD3
CD56
PD-1
HER2 + N87 Gastric cells + PBMC (E:T = 15:1)
+/- Margetuximab or Control mAb (100 ng/mL)
Anti-PD-1 Enhances Margetuximab-mediated NK Cell Proliferation and Expression of Granzyme B/Perforin
Ki67
Perforin
Granzyme B
% of NK Cells
Days
M + MGA012
M + control mAb
HER2 + N87 Gastric cells + PBMC (E:T = 15:1)
+ Margetuximab (100 ng/mL) +/- MGA012 (anti-PD1) 200 ng/mL

Proposed Margetuximab and Pembrolizumab Synergistic Mechanisms of Action

Margetuximab engages the innate immune system and activates the adaptive immune system supporting combination with checkpoint inhibitors

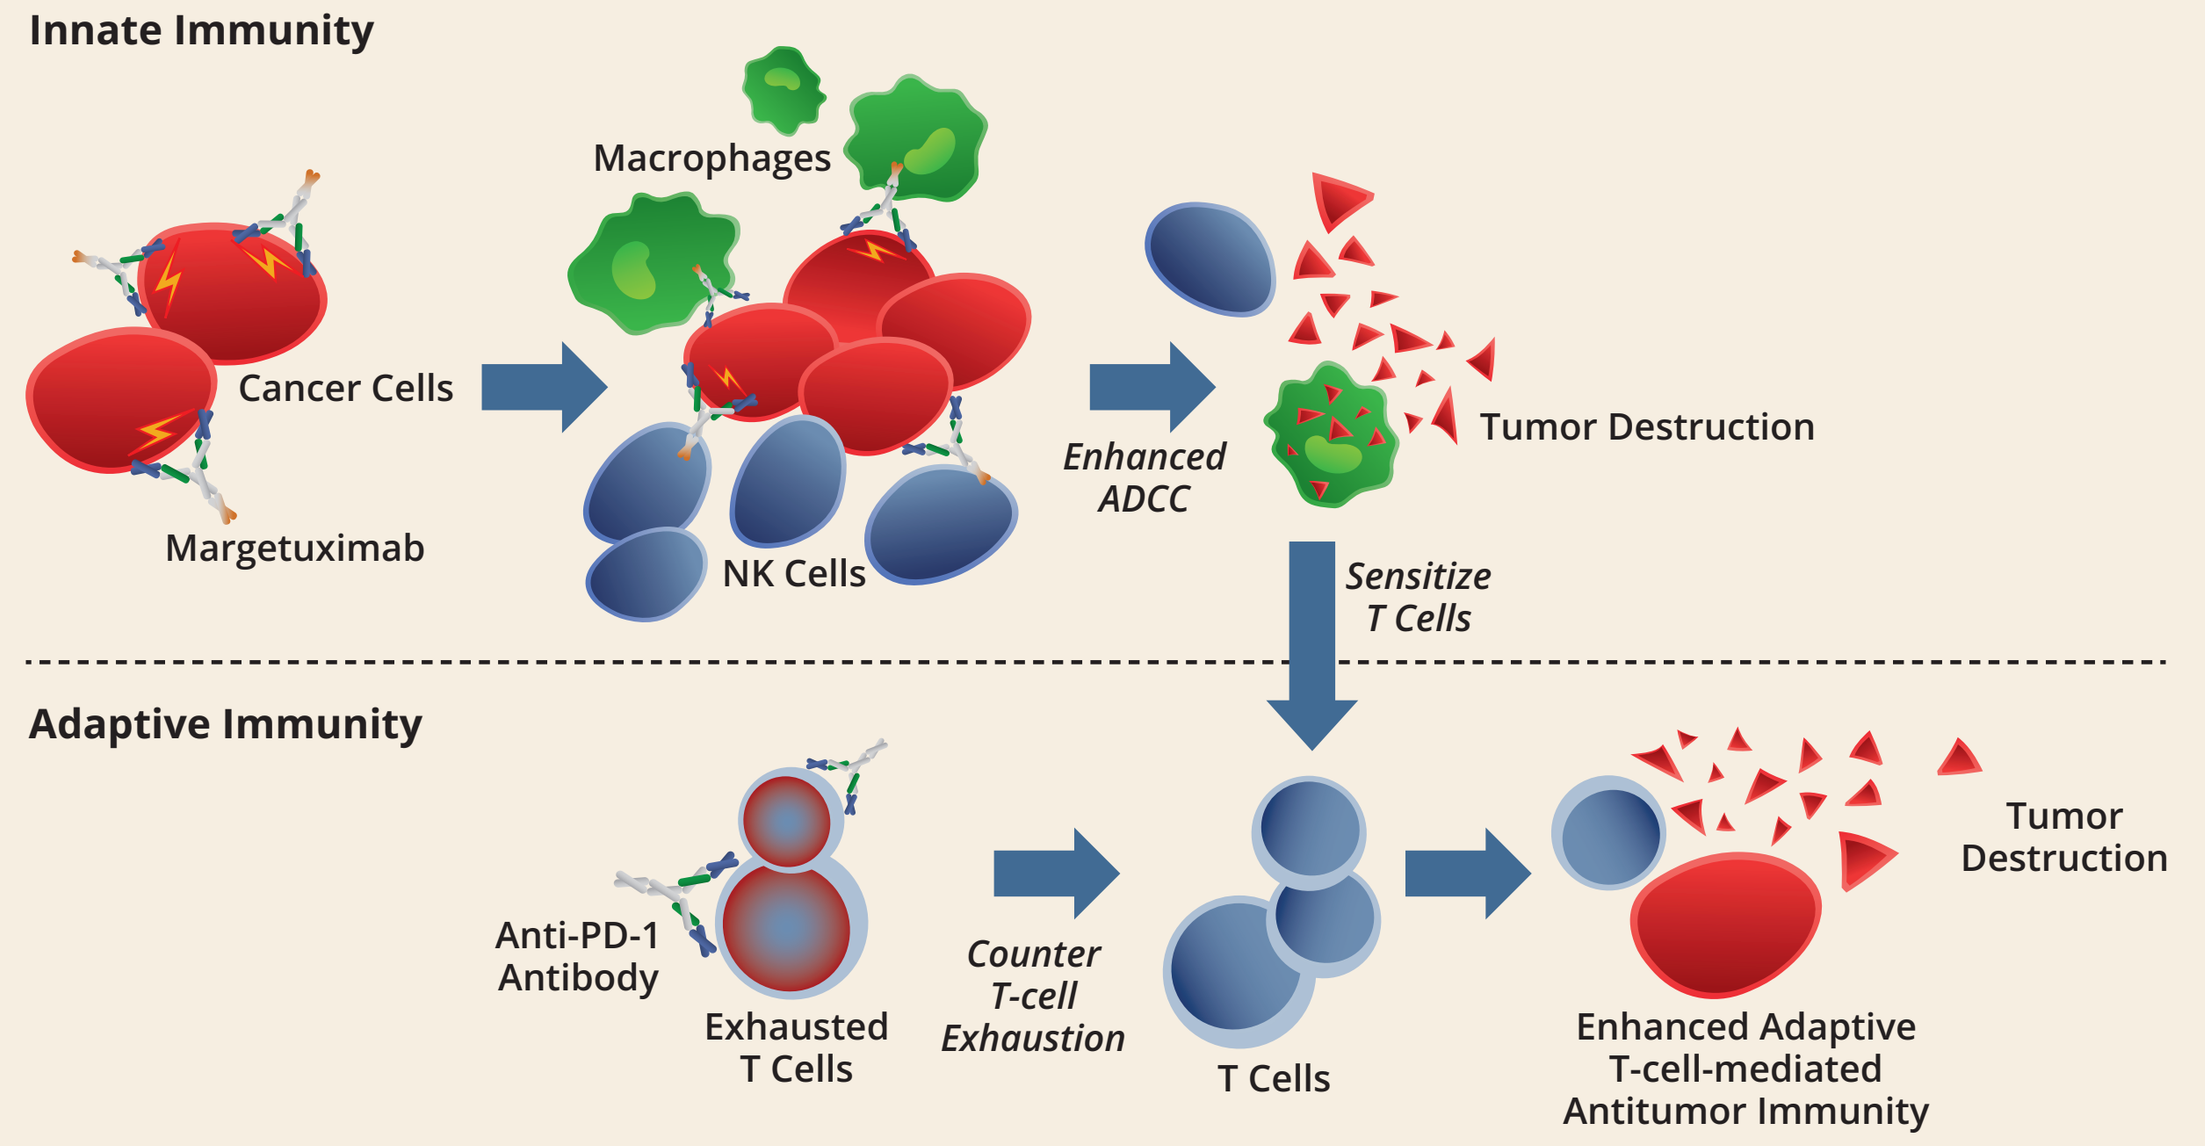
Innate Immunity
Cancer Cells
Margetuximab
Macrophages
NK Cells
Enhanced ADCC
Tumor Destruction
Sensitize T Cells
Adaptive Immunity
Anti-PD-1 Antibody
Exhausted T Cells
Counter T-cell Exhaustion
T Cells
Enhanced Adaptive T-cell-mediated Antitumor Immunity
Tumor Destruction

Methods

Study Design

Fully Enrolled Phase 2 Study in Advanced HER2+ Gastric Carcinoma

Dose Escalation (n=3–6 per margetuximab dose)	Dose Expansion #1 (margetuximab 15 mg/kg q3W + pembrolizumab 200 mg q3W)	Dose Expansion #2 (margetuximab 15 mg/kg q3W + pembrolizumab 200 mg q3W)
Margetuximab 10 mg/kg (n=3), 15 mg/kg (n=6) q3w + pembrolizumab 200 mg q3w	Gastric and Gastroesophageal (n=60)	Gastric (HER2 3+) (n=25)

- HER2-positive (archival IHC3+, or ICH2+/FISH positive), PD-L1-unselected 2nd line GEA pts post trastuzumab
 - 92 patients treated at recommended Phase 2 dose (RP2D) of 15 mg/kg margetuximab + 200 mg pembrolizumab included in analysis (data cut 10 July 2019)

Primary Endpoint:
- Safety, tolerability, overall response rate (ORR)

Secondary Endpoints:
- Progression-free survival (PFS) and overall survival (OS); PFS and OS at 6 months

Exploratory Endpoints:
- Disease control rate (DCR) = proportion of patients with complete response (CR) + partial response (PR) + stable disease (SD) for a minimum of 12 weeks
- HER2-amplification (post-trastuzumab) was confirmed by NGS of circulating-tumor DNA (ctDNA) for *ERBB2*amp (Guardant360®) as a surrogate for HER2 expression; previously presented data showed ~80% concordance
- PD-L1 tested on archival tissue by IHC (Clone 22C3 pharmDx); Combined Positive Score (per standard FDA approved assay)
- Anti-HER2 T-cell immunity measured by ELISPOT on PBMCs
- Gene expression profile performed on archival FFPE biopsies by NanoString PanCancer IO360™

Results

Patient Characteristics

Characteristic		All Patients (n=92)*
Age	Mean ± SD	60.2 ± 12.83
	Median (Range)	61.0 (19, 85)
Gender [n (%)]	Male	75 (81.5)
	Female	17 (18.5)
Race [n (%)]	Asian	51 (55.4)
	White	34 (37.0)
	Other	4 (4.3)
	Black or African American	3 (3.3)
ECOG Status [n (%)]	0	33 (35.9)
	1	59 (64.1)
Diagnosis [n (%)]	Gastric Cancer	61 (66.3)
	GEJ Cancer	31 (33.7)
Microsatellite Stable [n (%)]		84 (91.3)

*Data cutoff 10 July 2019.

Safety

- Treatment with combination of margetuximab and pembrolizumab demonstrated acceptable tolerability
- 63% of patients experienced treatment-related AE (TRAE), irrespective of grade
 - 19.6% of patients with TRAE ≥ Grade 3
 - Most common TRAE is pruritis in 17.4%
- 7 drug-related serious adverse events reported: autoimmune hepatitis (2), hyponatremia, dehydration, diabetic ketoacidosis, infusion-related reaction, and pneumonitis (1 each)
- 18 adverse events of special interest reported: infusion-related reaction (11), autoimmune hepatitis (2), endocrinopathy, LVEF dysfunction, pneumonitis (1 each); others (3)

Adverse Event	All Related AE: All (N=92)	All Related AE: ≥ Gr 3
TOTAL	**58 (63%)**	**18 (19.6)**
Pruritus	16 (17.4)	
Diarrhoea	14 (15.2)	
Infusion related reaction	12 (13.0)	2 (2.2)
Fatigue	12 (13.0)	
Rash	8 (8.7)	
Rash maculo-papular	5 (5.4)	
Anaemia	7 (7.6)	4 (4.3)
Lipase increased	4 (4.3)	1 (1.1)
Aspartate aminotransferase increased	4 (4.3)	1 (1.1)
Nausea	4 (4.3)	2 (2.2)
Chills	3 (3.3)	
Amylase increased	3 (3.3)	2 (2.2)
Hyperthyroidism	3 (3.3)	
Alanine aminotransferase increased	3 (3.3)	
Adrenal insufficiency	3 (3.3)	
Pain	2 (2.2)	
Abdominal pain	2 (2.2)	
Pyrexia	2 (2.2)	
Vomiting	2 (2.2)	2 (2.2)
Blood alkaline phosphatase increased	2 (2.2)	1 (1.1)
Ejection fraction decreased	2 (2.2)	
Autoimmune hepatitis	2 (2.2)	2 (2.2)
Pneumonitis	2 (2.2)	1 (1.1)
Peripheral neuropathy	2 (2.2)	
Hypotension	2 (2.2)	1 (1.1)

Data cutoff 10 July 2019. Events in ≥ 2 patients at 15 mg/kg margetuximab.

Best Response by HER2 Expression and Tumor Site

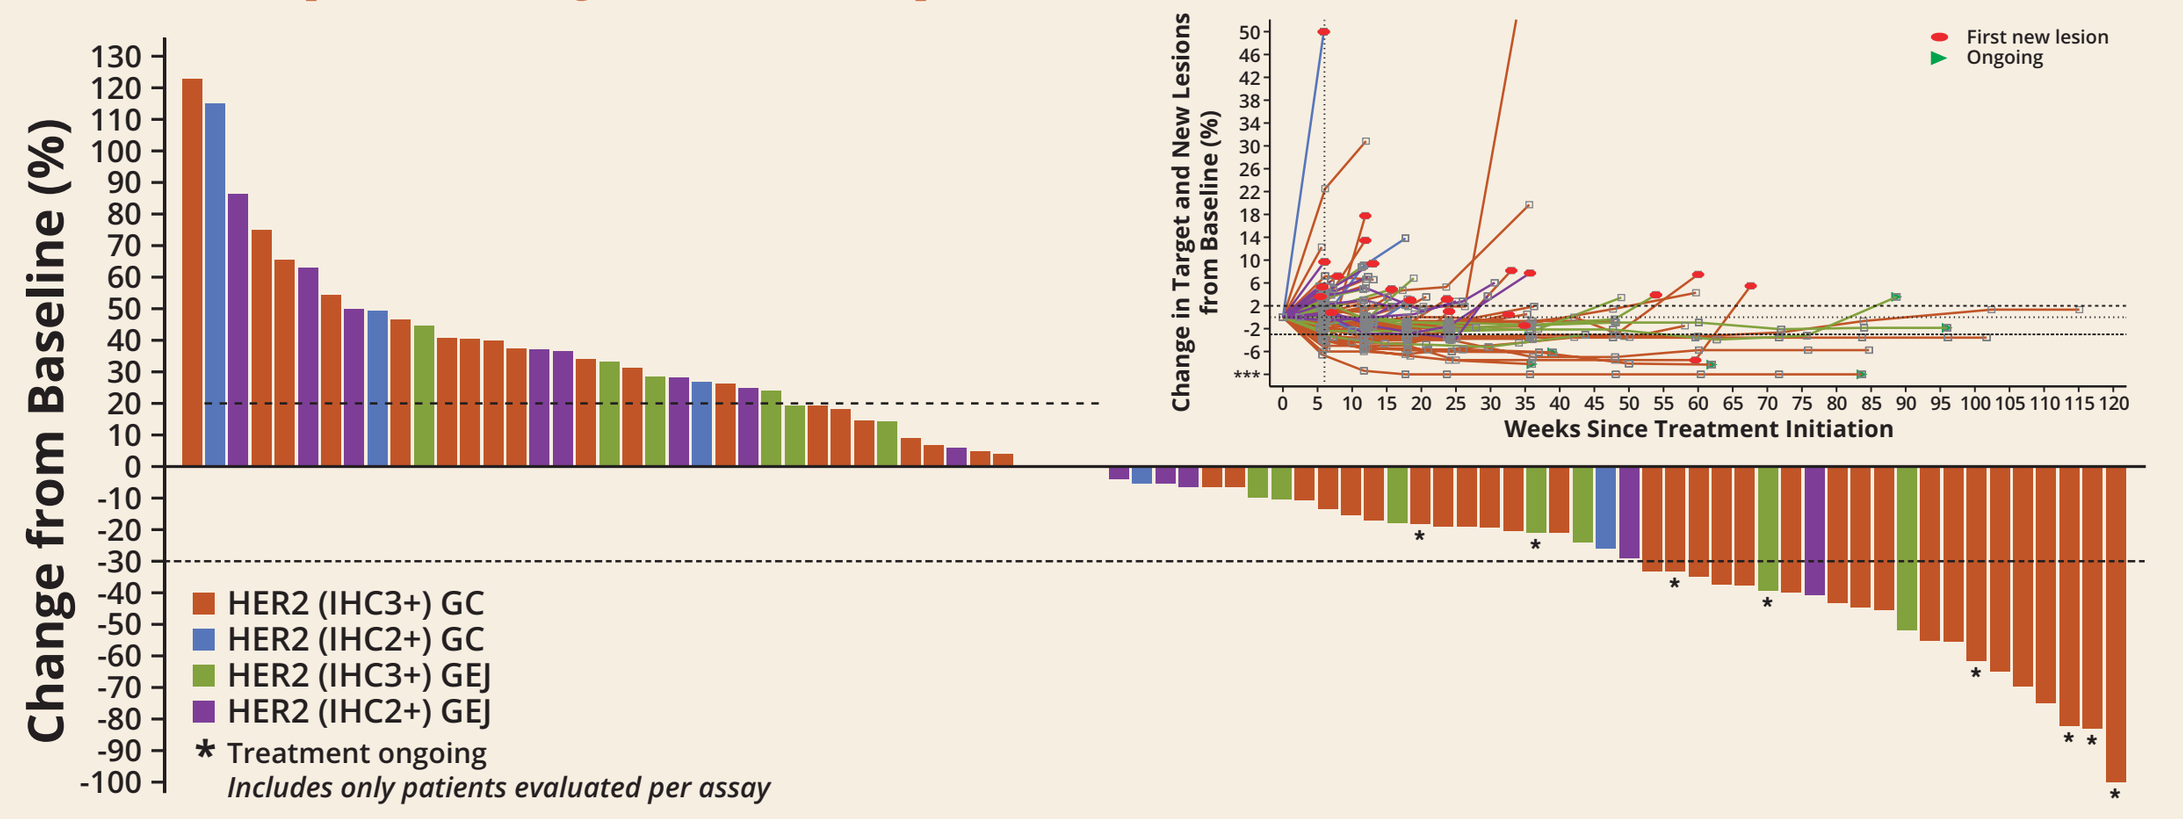
Change from Baseline (%)
HER2 (IHC3+) GC
HER2 (IHC2+) GC
HER2 (IHC3+) GEJ
HER2 (IHC2+) GEJ
* Treatment ongoing
Includes only patients evaluated per assay
Change in Target and New Lesions from Baseline (%)
Weeks Since Treatment Initiation
First new lesion
Ongoing

Targetable Biomarker Expression in Selected Populations

Positive Biomarker	All Patients*	Gastric Cancer	GEJ Cancer
*ERBB2*amp	48/82 (58.5%)	35/56 (62.5%)	13/26 (50.0%)
PD-L1+	33/76 (48.7%)	26/54 (48.1%)	7/22 (31.8%)
HER2 3+	71/92 (77.2%)	55/61 (90.2%)	16/31 (51.6%)
*ERBB2*amp/PD-L1+	18/39 (46.2%)	23/26 (88.5%)	1/13 (7.7%)

- Approximately 60% (32/53) of patients tested had retained HER2 expression post-trastuzumab as determined by *ERBB2*amp using ctDNA
- Approximately 49% of patients tested were PD-L1+ by IHC
- For both markers (PD-L1 and *ERBB2*amp), a higher rate of expression was observed in patients with GC

Data cutoff 10 July 2019. *Includes only patients evaluated per assay.

Higher Biomarker Expression in GC is Associated with Improved Clinical Activity

	N	ORR* (%, n)	DCR (%, n)	mPFS (months; 95% CI)	mOS (months; 95% CI)
Overall	92	21.7% (20/92)	54.4% (50/92)	2.73 (1.61, 4.34)	12.5 (9.07, 14.09)
Gastric Cancer	61	29.5% (18/61)	65.6% (40/61)	4.1 (2.60, 5.52)	13.9 (9.72, 20.47)
GEJ Cancer	31	6.5% (2/31)	32.3% (10/31)	1.4 (1.35, 3.61)	9.2 (4.96, 14.03)
Gastric Cancer HER2 IHC 3+	55	32.7% (18/55)	69.1% (38/55)	4.7 (2.66, 7.49)	14.6 (10.55, NR)
Gastric Cancer HER2 IHC 3+/PD-L1+	23	52.2% (12/23)	82.6% (19/23)	5.52 (2.60,13.90)	20.47 (8.08, NR)
Gastric Cancer HER2 IHC 3+/PD-L1+/*ERBB2*amp	14	71.4% (10/14)	92.9% (13/14)	6.60 (1.61, 5.54)	NR (6.74, NR)

*17 confirmed, 3 unconfirmed responses

HER2 (IHC3+) Gastric Cancer

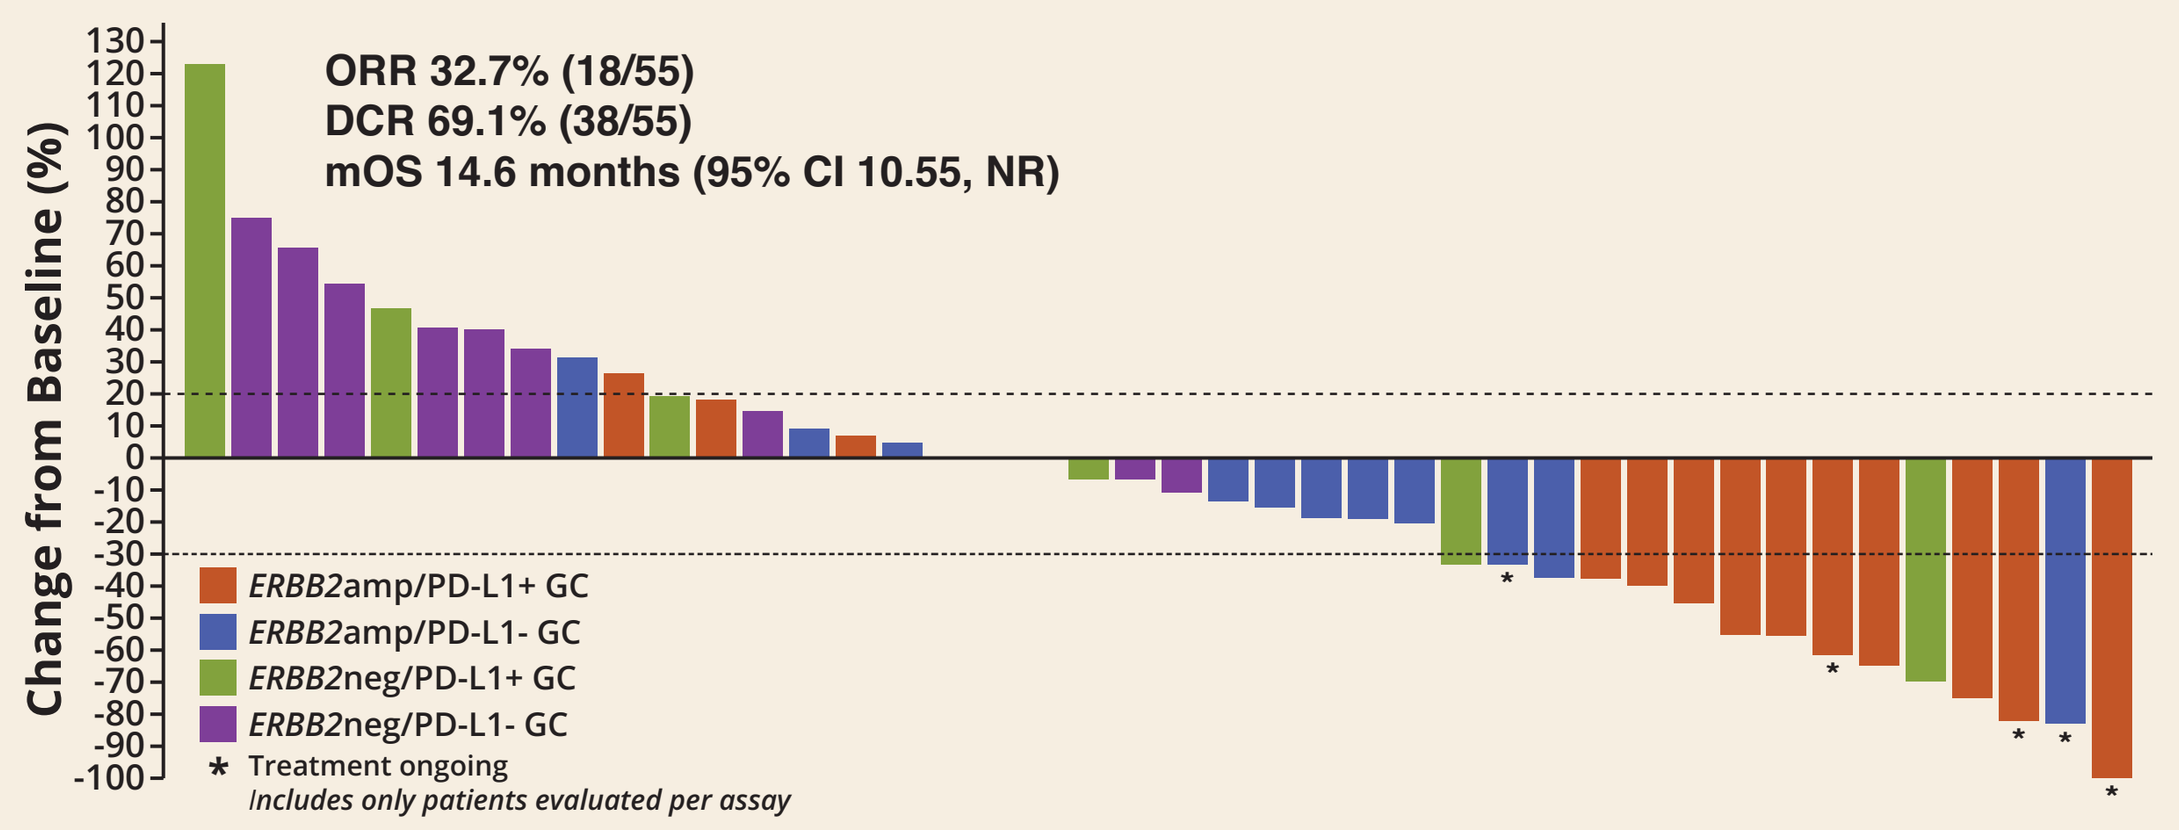
ORR 32.7% (18/55)
DCR 69.1% (38/55)
mOS 14.6 months (95% CI 10.55, NR)
Change from Baseline (%)
ERBB2amp/PD-L1+ GC
ERBB2amp/PD-L1- GC
ERBB2neg/PD-L1+ GC
ERBB2neg/PD-L1- GC
* Treatment ongoing
Includes only patients evaluated per assay

Data cutoff 10 July 2019.

Preliminary Correlative Studies: NanoString Gene Expression Analysis

Increasing expression of PD-L1 and ERBB2 is associated with response

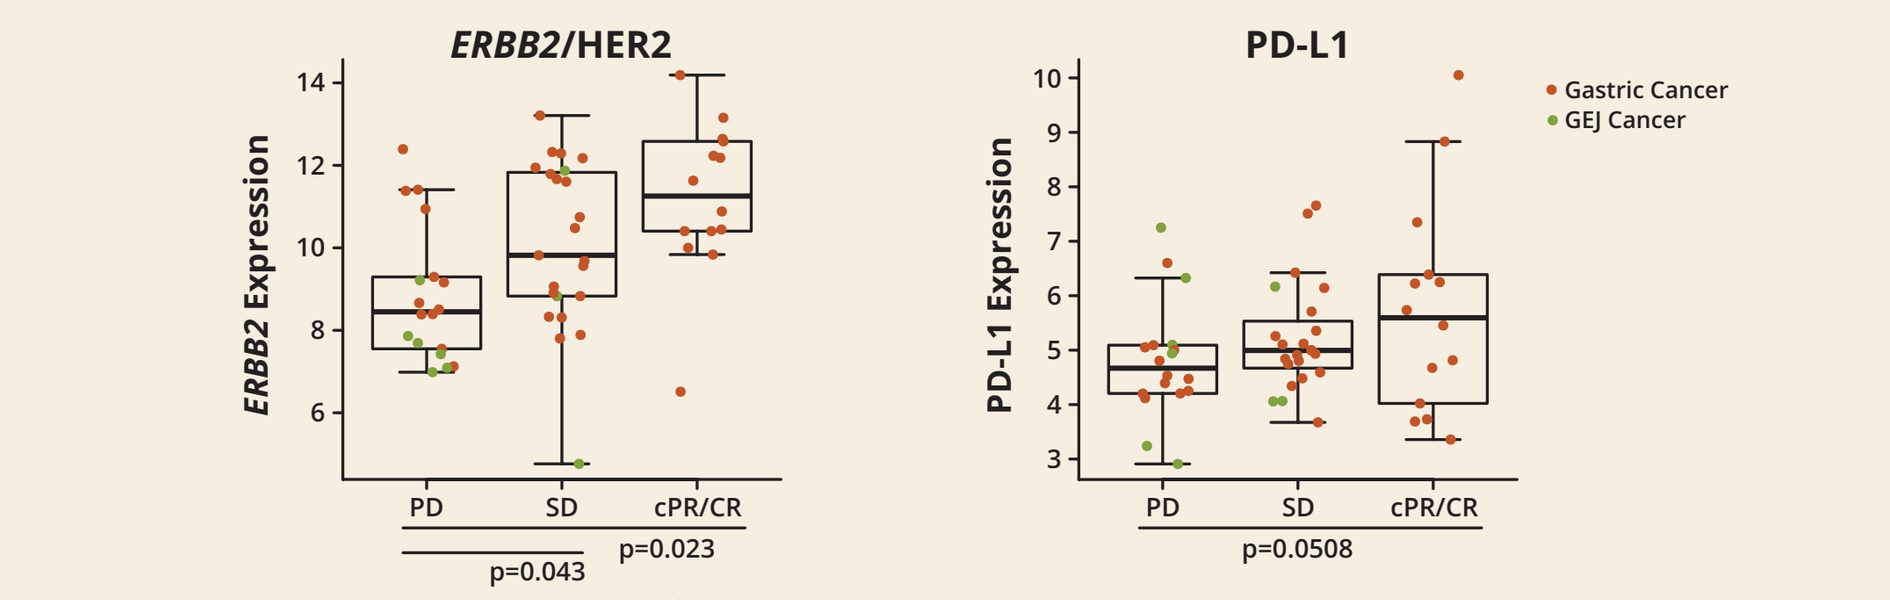
ERBB2/HER2
ERBB2 Expression
PD
SD
cPR/CR
p=0.043
p=0.023
PD-L1
PD-L1 Expression
p=0.0508
Gastric Cancer
GEJ Cancer

Patients treated at RP2D for HER2+GEA post-trastuzumab (n=52).

Increased intratumor NK cell abundance is associated with response

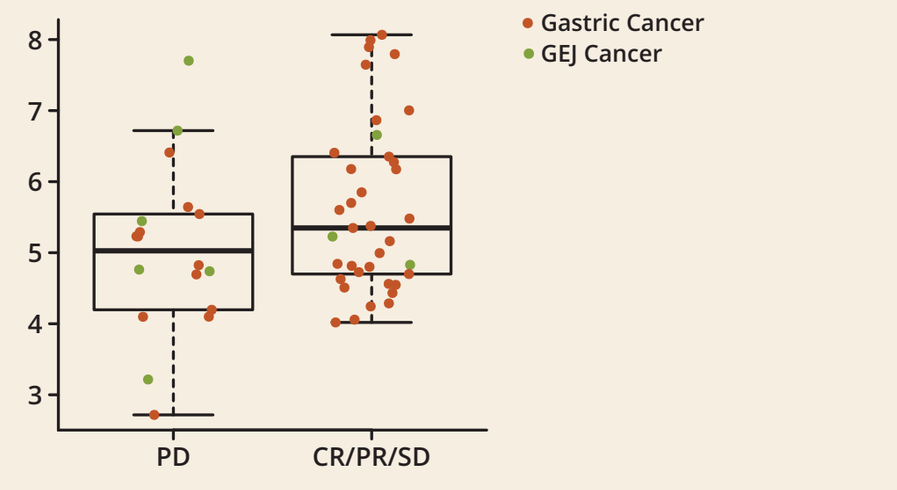
NK CD56dim Cells Abundance
PD
CR/PR/SD
Gastric Cancer
GEJ Cancer

Patients treated at RP2D for HER2+GEA post-trastuzumab (n=52). NK CD56[dim] genes: IL21R, KIR2DL3, KIR3DL1, KIR3DL2. Further analysis of patients on this study is presented "Evaluation of tumor microenvironment identifies immune correlates of response to combination immunotherapy with margetuximab (M) and pembrolizumab (P) in HER2+ gastroesophageal adenocarcinoma (GEA)" Abstract #2547.

Preliminary Correlative Studies: Anti-HER2 T-cell Immunity

Margetuximab activates the adaptive immunity as evidenced by increase in anti-HER2 specific T-cell immunity

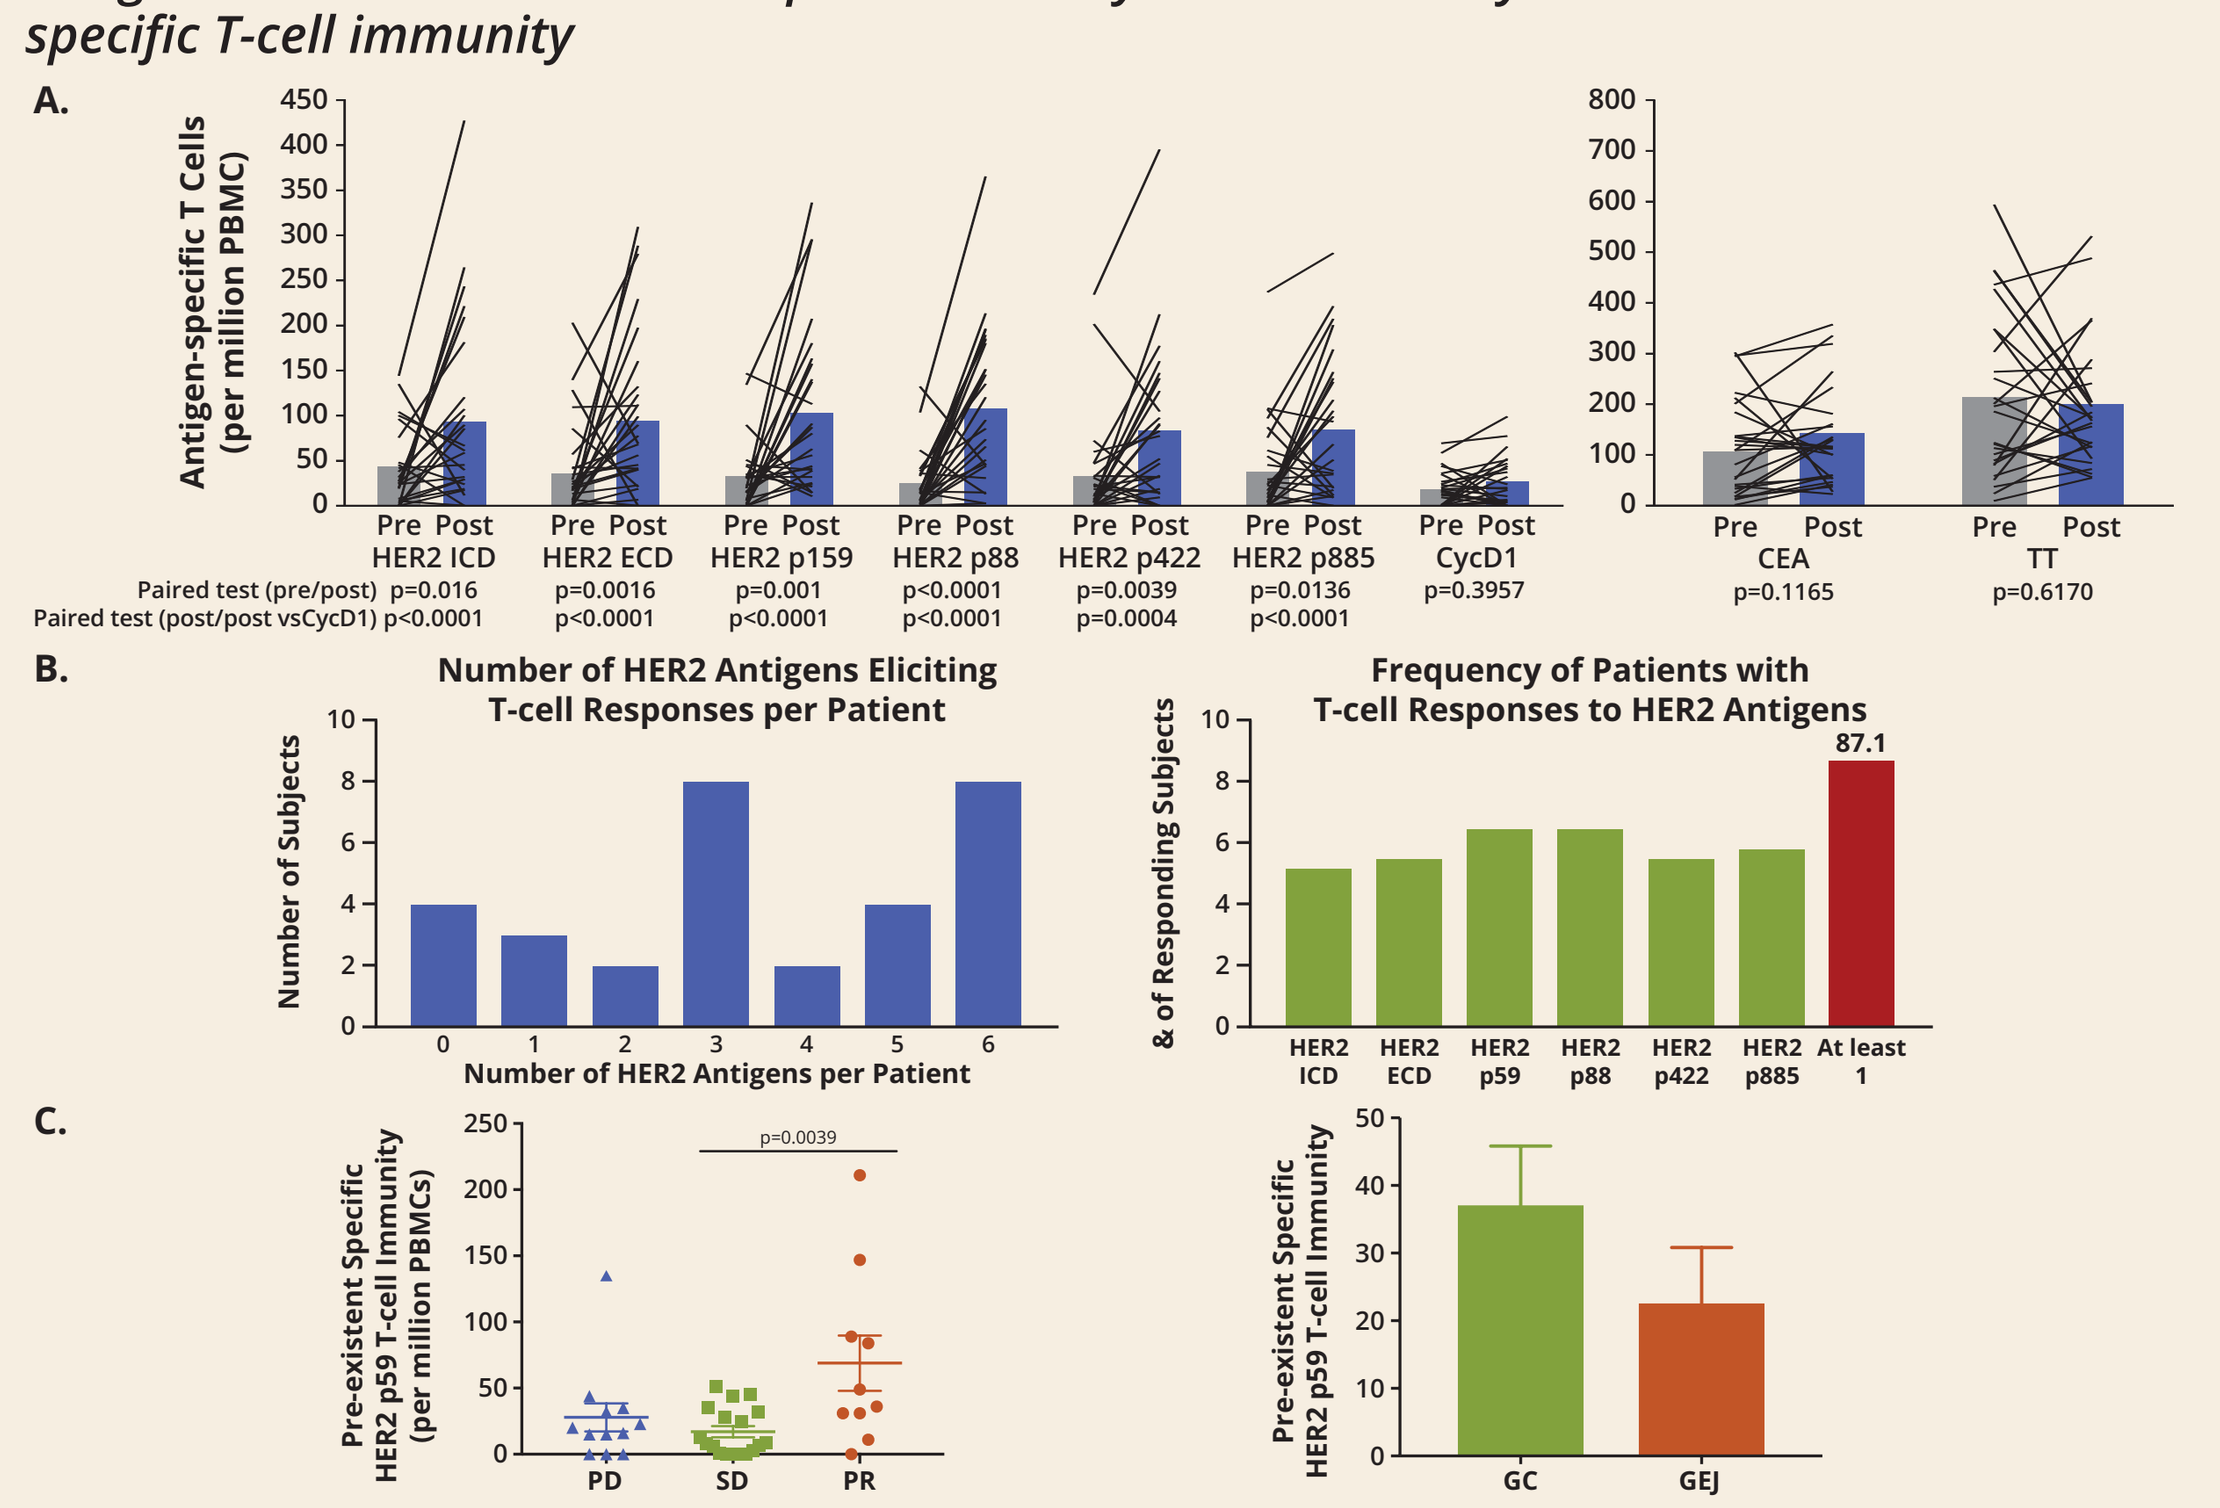
A.
Antigen-specific T Cells (per million PBMC)
Pre Post
HER2 ICD
HER2 ECD
HER2 p159
HER2 p88
HER2 p422
HER2 p885
CycD1
CEA
TT
Paired test (pre/post) p=0.016 p=0.0016 p=0.001 p<0.0001 p=0.0039 p=0.0136 p=0.3957 p=0.1165 p=0.6170
Paired test (post/post vsCycD1) p<0.0001 p<0.0001 p<0.0001 p<0.0001 p=0.0004 p<0.0001
B.
Number of HER2 Antigens Eliciting T-cell Responses per Patient
Number of Subjects
Number of HER2 Antigens per Patient
Frequency of Patients with T-cell Responses to HER2 Antigens
& of Responding Subjects
HER2 ICD
HER2 ECD
HER2 p59
HER2 p88
HER2 p422
HER2 p885
At least 1
87.1
C.
Pre-existent Specific HER2 p59 T-cell Immunity (per million PBMCs)
p=0.0039
PD
SD
PR
GC
GEJ

A: Left panel bars show the mean (n=40 patients) pre-treatment (C1D1, Pre) and the mean (n=33 patients) post-treatment (C4D1, Post) frequency of antigen-specific T cells frequencies (per million PBMC plated) that recognize vaccine antigens, HER2 ICD protein, HER2 ECD fragment (aa 22-122), HER2 p59 class II peptide, HER2 p88 class II peptide, HER2 p422 class II peptide and a pan class II binding cyclin D1 peptide. Right panel shows the mean pre-treatment (Pre) and highest post-vaccination frequency of CEA and tetanus toxoid (TT)-specific T cells for the same patients. Inset lines trace the pre and post responses for each unique patient for which there was a pre and post treatment value. p values (shown in the upper portion of the figure) were calculated using the Wilcoxon matched pairs ranked sum test for paired samples only (n=31). **B:** Paired pre- (C1D1) and post-treatment (C4D1) PBMC samples, obtained from 31 patients with HER2+ cancer, were subjected to IFN-γ ELISpot assays with different HER2 antigens. T cell responses were defined as positive if the number of antigen-specific T cells per million PBMC in the post-treatment sample increased by ≥ 2-fold compared to the pre-treatment baseline sample. **C:** Pre-treatment (C1D1, Pre) (n=40 patients) frequency of antigen-specific T cells frequencies (per million PBMC plated) that HER2 p59 class II peptide. Left panel shows the mean pre-treatment (Pre) in relations to M+P treatment outcome. Right panel shows the mean pre-treatment (Pre) in relation to tumor location.

Correlative studies further support the mechanism of action of margetuximab and pembrolizumab in the GEA patient population.

Margetuximab + Anti-PD-1 Data in 2nd Line Presents Opportunity to Advance to 1st Line

HER2+ gastric cancer benchmarks

	1st Line: SOC	2nd Line: SOC	2nd Line: Ongoing Phase 2 Study		2nd Line: Failed
Agent (Study)	Trastuzumab + Chemo[a] (TOGA)	Ramucirumab + Paclitaxel[b] (RAINBOW)	Margetuximab + Pembrolizumab[c]: IHC 3+	Margetuximab + Pembrolizumab[c]: IHC 3+/PD-L1+	Pembrolizumab[d] (KEYNOTE-61) ✖
ORR	47%	28%	33%	52%	15.8% (PD-L1+)
Median PFS	6.7 mos.	4.4 mos.	4.7 mos.	5.5 mos.	1.5 mos.
Median OS	**13.1 mos.**	9.6 mos.	**14.6 mos.**	**20.5 mos.**	9.1 mos
≥ Grade 3 TRAEs	**68%**	Overall: N/A 41% Neutropenia 15% Hypertension 12% Fatigue	**20%**	**20%**	14.3%
Gastric/GEJ Patient Mix	80/20%	80/20%	100%/0%	100%/0%	Not disclosed

[a]Data from Herceptin package insert; Bang, et al., *Lancet*, 2010. [b]Data from Cyramza package insert; Wilkes, et al., *Lancet Oncology*, 2014. [c]Grade 3 TRAE includes all GC and GEJ patients (n=92). [d]Data presented at ASCO 2018, Abstract 4062.

Conclusions

- Margetuximab is an Fc-engineered anti-HER2 antibody that mediates enhanced innate responses and leads to increased HER2-specific adaptive immune responses in patients with HER2+ gastric and breast carcinoma
- Margetuximab can upregulate the expression of PD-1 on NK and NKT cells, and anti-PD-1 antibody (MGA012) can further potentiate the enhancement of NK cell function by margetuximab in vitro
- The combination of margetuximab + pembrolizumab (M+P), as a chemotherapy-free regimen, demonstrated acceptable safety and tolerability in patients with HER2 GEA that have progressed/recurred after prior 1L therapy including trastuzumab
- The combination of M+P has demonstrated encouraging antitumor activity in patients with 2nd line HER2-positive, PD-L1 unselected GEA after treatment with trastuzumab plus chemotherapy
 - ORR that exceed historical experience with either margetuximab or checkpoint inhibitor alone
 - ORR further increased in gastric cancer patients whose tumors are HER2 IHC 3+ (33%)
 - ORR most pronounced in gastric cancer patients whose tumors are both HER2 IHC3+ and PD-L1+ (52%)
- Maturing data from this ongoing study suggest that the combination of margetuximab + checkpoint prolonged PFS, and in particular, overall survival* compared to historical experience with checkpoint inhibitor alone, or existing standard of care
- Exploratory biomarker studies suggest potential associations between *ERBB2* and PD-L1 expression in tumor microenvironment (TME), baseline NK infiltration at baseline in the TME, and pre-existing HER2 specific T-cell immunity with objective response to M+P, as well as evidence of enhancement on HER2 specific T-cell immunity with M+P
- Based on these observations, the combination of margetuximab + a checkpoint inhibitor could provide a potential chemotherapy-free regimen for the treatment of GEA and/or be used with chemotherapy to improve the clinical activity of existing 1L SoC
- A Phase 2/3 study (MAHOGANY) is being initiated to evaluate margetuximab in combination with a checkpoint inhibitor with or without chemotherapy in 1L GEA

*Margetuximab plus Pembrolizumab for Treatment of Patients with HER2-Positive Gastroesophageal Adenocarcinoma (GEA) Post-Trastuzumab: Survival Analysis, Abstract Number: 2812.

MAHOGANY Phase 2/3 Study: Registration Path in 1L Gastric and GEJ Cancer

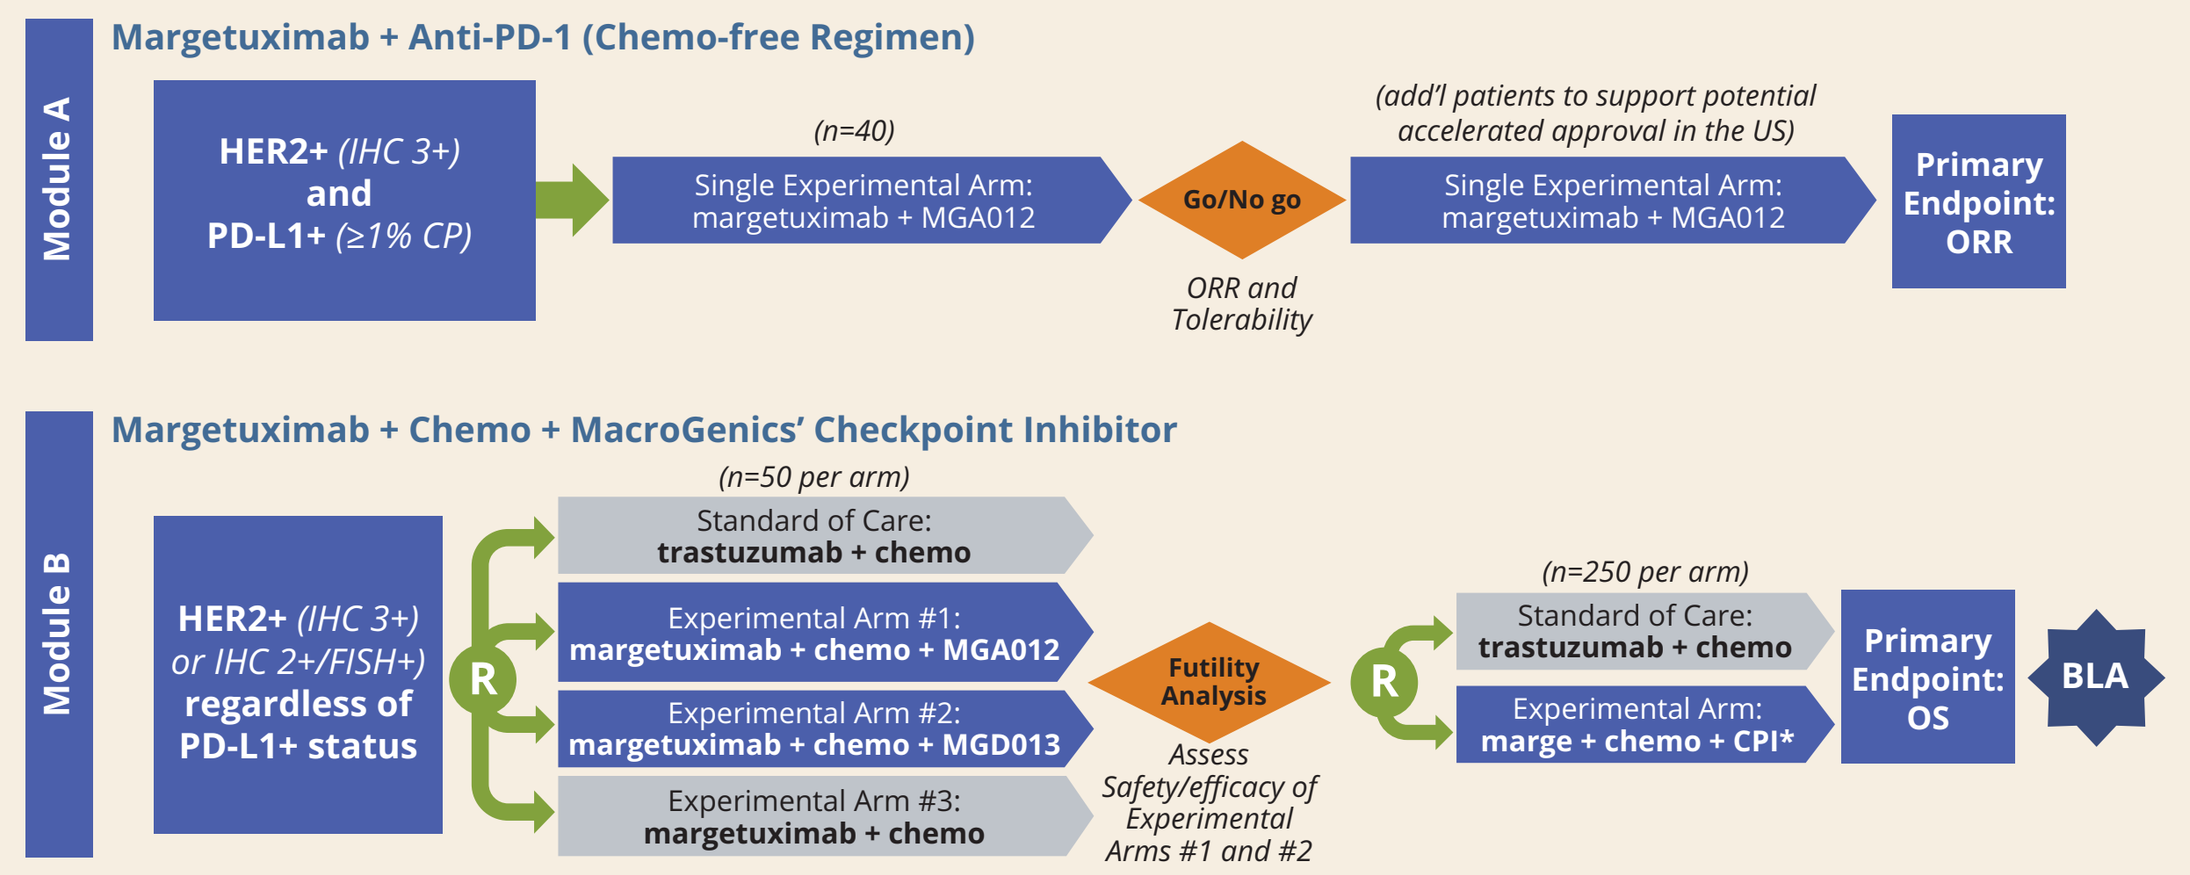
Module A
Margetuximab + Anti-PD-1 (Chemo-free Regimen)
HER2+ (IHC 3+) and PD-L1+ (≥1% CP)
(n=40)
Single Experimental Arm: margetuximab + MGA012
Go/No go
ORR and Tolerability
(add'l patients to support potential accelerated approval in the US)
Single Experimental Arm: margetuximab + MGA012
Primary Endpoint: ORR
Module B
Margetuximab + Chemo + MacroGenics' Checkpoint Inhibitor
HER2+ (IHC 3+) or IHC 2+/FISH+) regardless of PD-L1+ status
R
(n=50 per arm)
Standard of Care: trastuzumab + chemo
Experimental Arm #1: margetuximab + chemo + MGA012
Experimental Arm #2: margetuximab + chemo + MGD013
Experimental Arm #3: margetuximab + chemo
Futility Analysis
Assess Safety/efficacy of Experimental Arms #1 and #2
(n=250 per arm)
Standard of Care: trastuzumab + chemo
Experimental Arm: marge + chemo + CPI*
Primary Endpoint: OS
BLA

*Pending chronic tox study (if regimen with MGD013 is selected).

This study was sponsored by MacroGenics, Inc. Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Presented at the 2019 Annual Congress of the European Society for Medical Oncology, September 27–October 1, 2019, Barcelona, Spain

davidsonj@macrogenics.com