MGD019, a PD-1 x CTLA-4 Bispecific DART® Molecule, Provides Simultaneous Blockade of PD-1 and CTLA-4 Checkpoint Pathways

Poster 1012

Alexey Berezhnoy, Kurt Stahl, Kalpana Shah, Tim Gaynutdinov, Daorong Liu, Ross La Motte-Mohs R, Jessica Saul, Jonathan Li*, Douglas Smith*, Jill Rillema*, Sergey Gorlatov, Valentina Ciccarone, Ralph Alderson, Hua Li, James Tamura, Jennifer Brown, Bradley Sumrow, Jon Wigginton, Ezio Bonvini, Syd Johnson, Paul A. Moore

MacroGenics, Inc., Rockville, MD and *Brisbane, CA

NCT03761017

http://ir.macrogenics.com/events.cfm

Abstract

Combinatorial blockade of PD-1 and CTLA-4 has shown clinical benefit beyond that observed with individual mAbs, albeit with increased toxicity. MGD019 is a PD-1 x CTLA-4 tetravalent bispecific molecule with a human IgG4 backbone to limit effector function and designed to engage each target in a bivalent modality. MGD019 demonstrated independent blockade of CTLA-4/B7 and PD-1/PD-L(1/2) with a potency comparable to that achieved by replicas of the approved ipilimumab (ipi) and nivolumab (nivo). MGD019 enhanced antigen-driven in vitro T-cell activation to a level comparable to the combinatorial PD-1 plus CTLA-4 blockade. Tumor microenvironment models that recapitulate vascular or stromal compartments confirmed MGD019 induced in vitro immune response profiles comparable to those observed with replicas of ipi plus nivo. Multiplex ISH showed enrichment of PD-1/CTLA-dual positive cells in tumor-infiltrating lymphocytes (TILs) relative to normal tissues, where distinct populations expressed CTLA-4 or PD-1. MGD019 mediated enhanced blockade of CTLA-4 ligand interaction to CTLA4/PD1 dual-expressing cells compared to cells expressing CTLA-4 alone. MGD019 was well-tolerated in cynomolgus monkeys, with no mortality or significant adverse findings up to 100 mg/kg QWx4. T-cell proliferation in the periphery and expansion in lymphoid organs was observed, with increases in ICOS+ CD4 cells and memory T cells, findings attributable to the CTLA-4 blocking arm, since the anti-PD-1 mAb precursor was devoid of these activities. MGD019 offers the convenience of single molecule administration for dual checkpoint blockade. In addition to providing full blockade on cells expressing PD-1 or CTLA-4 individually, MGD019 exploits dual target avidity resulting in preferential engagement and enhanced blockade on cells that express both checkpoint molecules, a feature that could provide additional benefits given the preeminent co-expression of CTLA-4 and PD-1 by TILs. MGD019 was well tolerated in cynomolgus monkeys, while demonstrating biological effects of CTLA-4 antagonism. Taken together, these data support clinical testing of MGD019 in cancer patients.

Introduction

T Cells Co-expressing PD-1 and CTLA-4 are More Prevalent Among TILs Compared to Healthy Tissues

Ovarian Cancer | Normal Tonsil

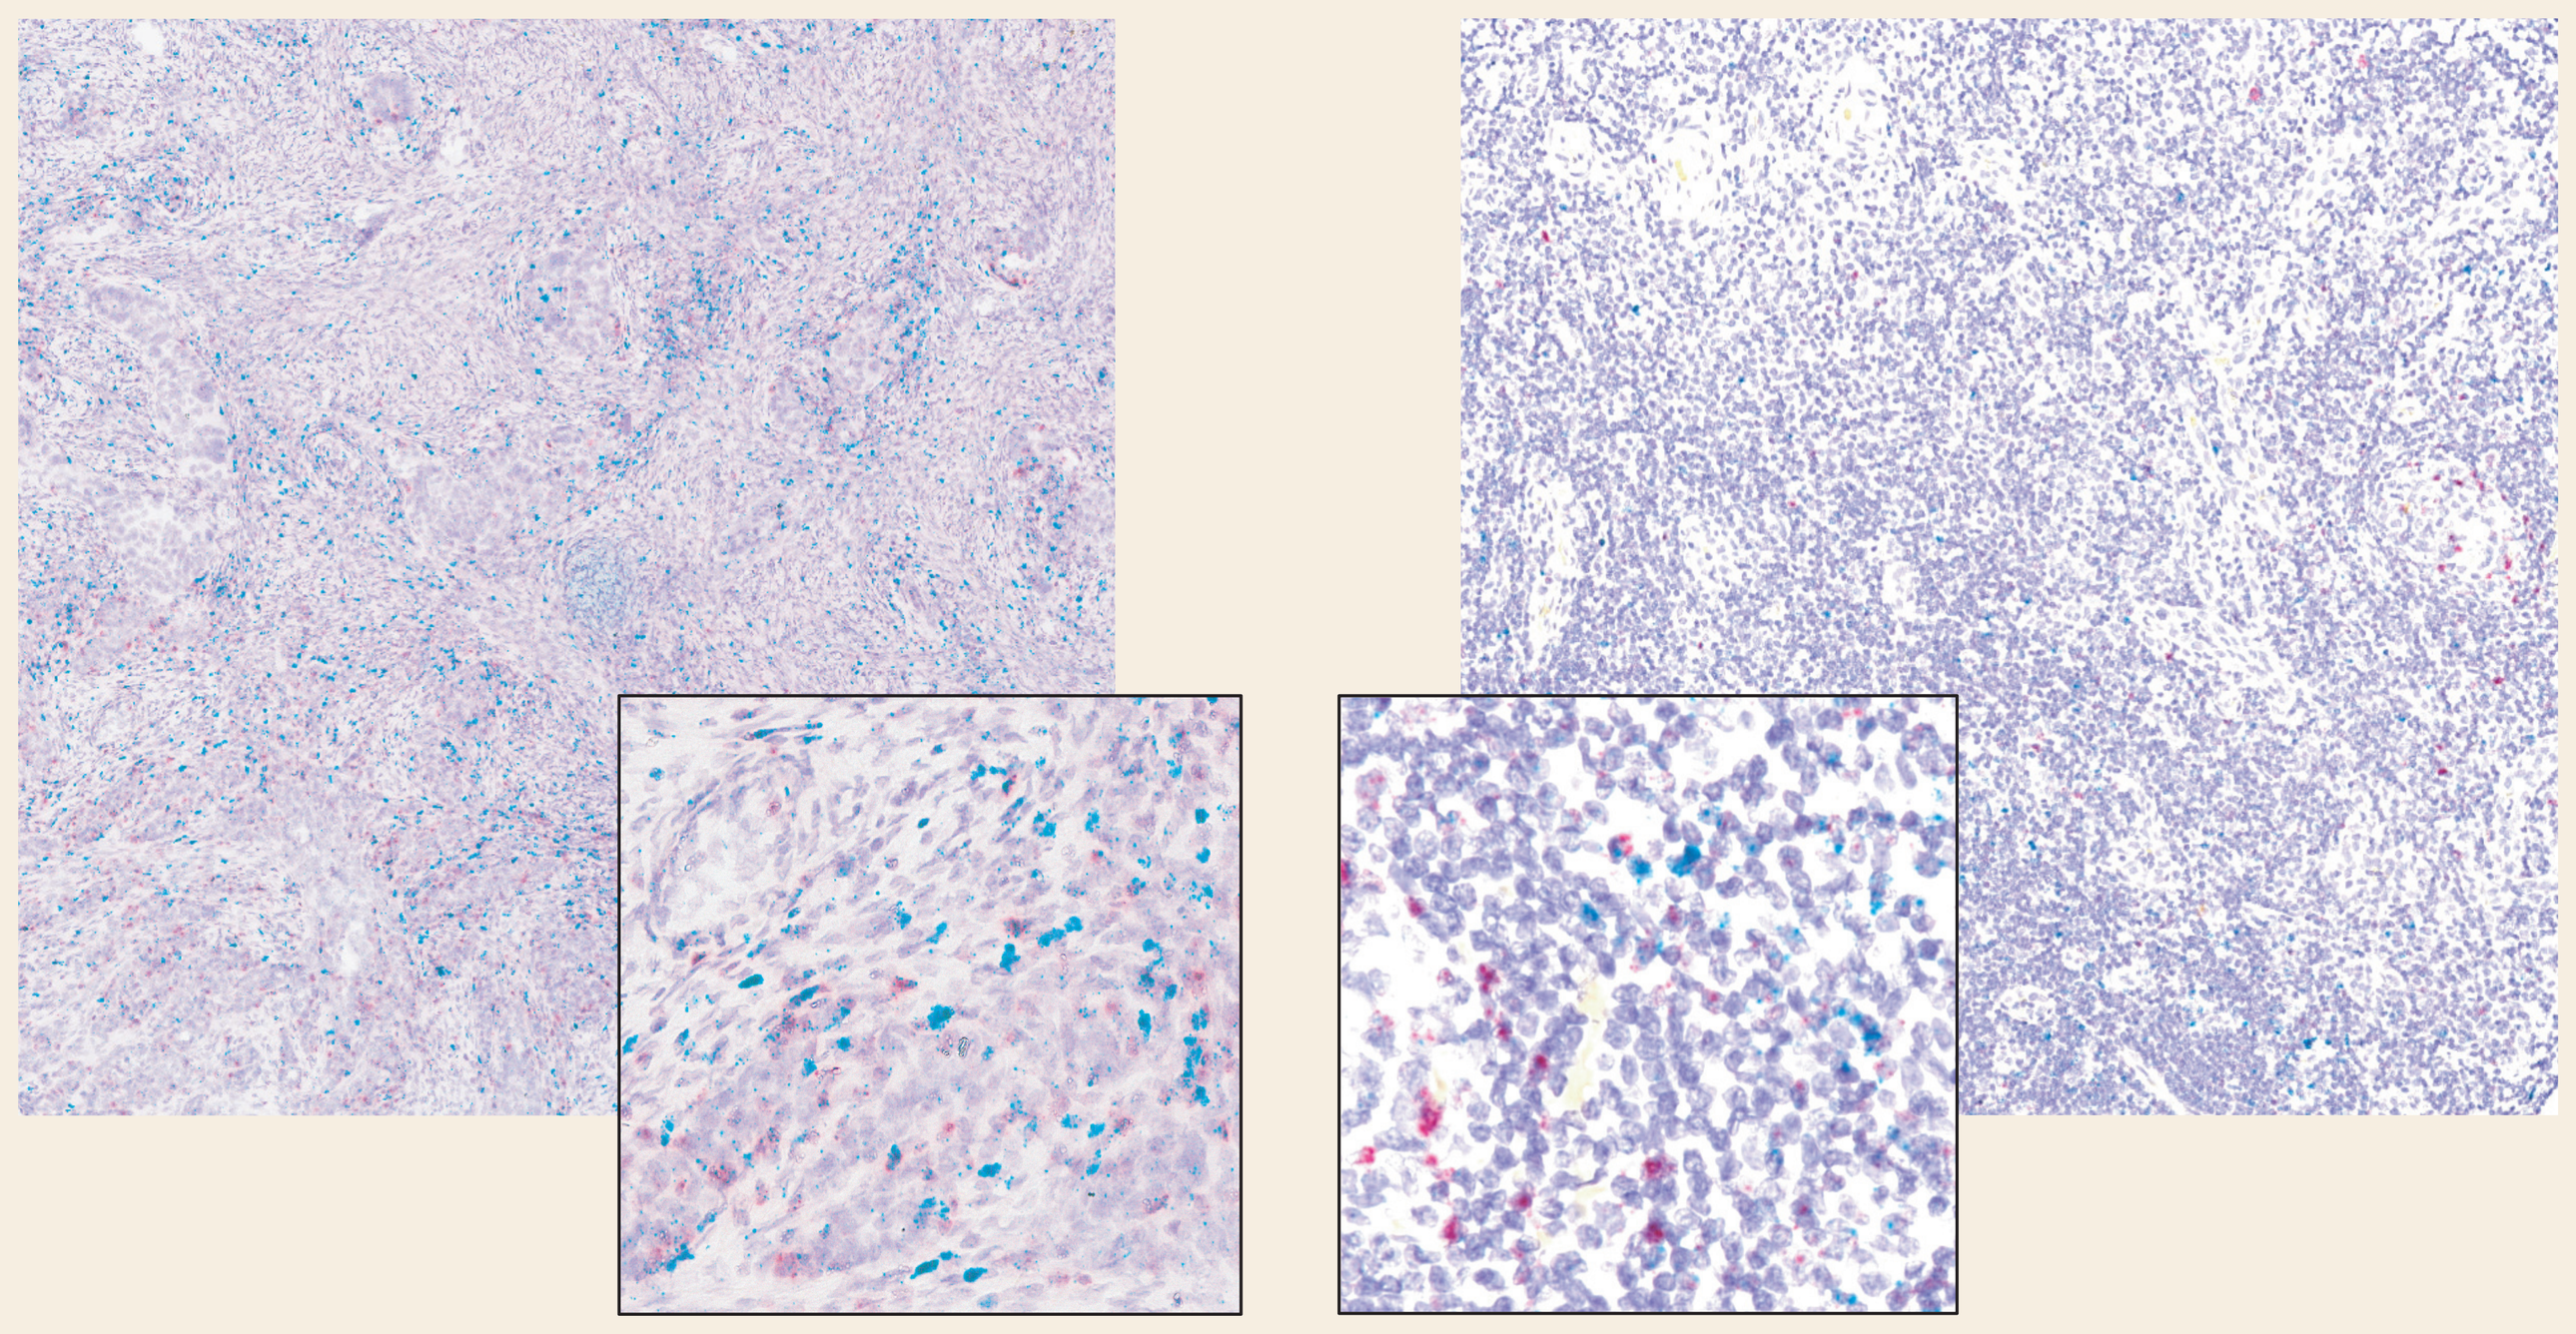

Expression of PD-1 (red) and CTLA-4 (blue) mRNA in tissue samples of ovarian cancer (left) or normal human tonsils (right) visualized by RNAscope™.

MGD019, a Tetravalent Bispecific DART Molecule

MGD019 Molecular Structure

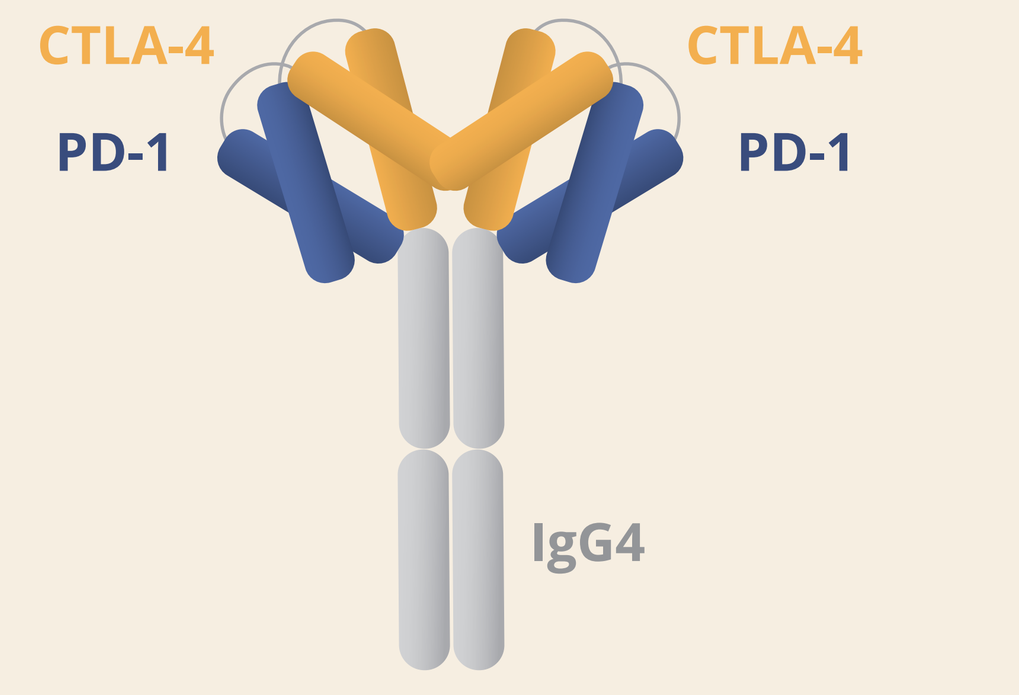
CTLA-4
CTLA-4
PD-1
PD-1
IgG4

Rationale for PD-1xCTLA-4 Dual Checkpoint Targeting Strategy

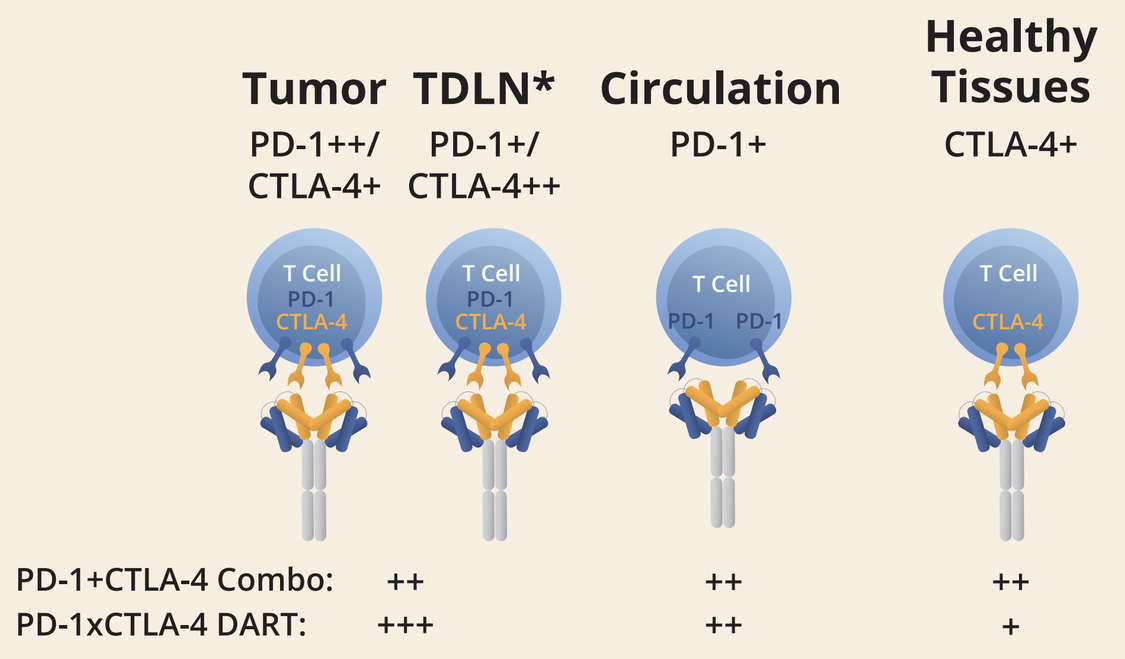
Tumor
PD-1++/ CTLA-4+
TDLN*
PD-1+/ CTLA-4++
Circulation
PD-1+
Healthy Tissues
CTLA-4+
T Cell
PD-1+CTLA-4 Combo: ++ ++ ++
PD-1xCTLA-4 DART: +++ ++ +

*TDLN: Tumor-draining lymph node.

MGD019 Co-engages PD-1 and CTLA-4

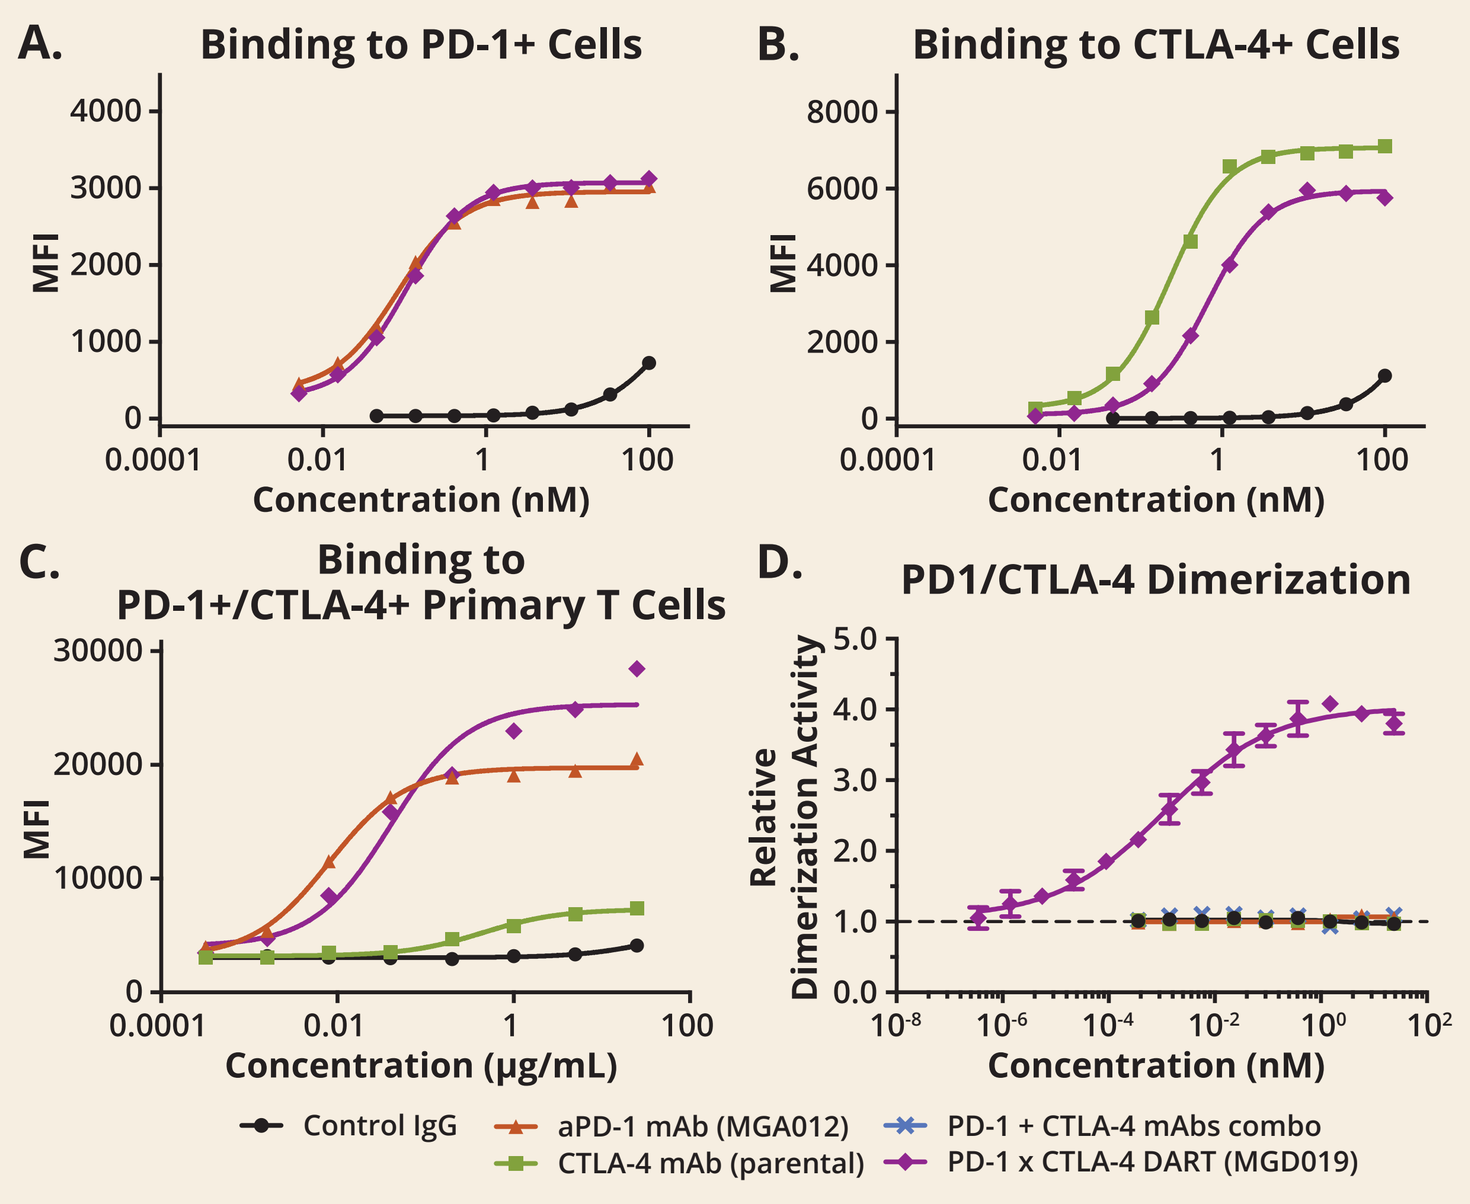
A. Binding to PD-1+ Cells
B. Binding to CTLA-4+ Cells
C. Binding to PD-1+/CTLA-4+ Primary T Cells
D. PD1/CTLA-4 Dimerization
MFI
Relative Dimerization Activity
Concentration (nM)
Concentration (μg/mL)
Control IgG
aPD-1 mAb (MGA012)
CTLA-4 mAb (parental)
PD-1 + CTLA-4 mAbs combo
PD-1 x CTLA-4 DART (MGD019)

Binding of MGD019 or control molecules to Jurkat/PD-1 **(A)** or Jurkat/CTLA-4 **(B)** cells measured by flow cytometry. **C.** Binding of MGD019 or control molecules to in vitro activated primary human T cells by flow cytometry. **D.** Co-engagement of PD-1 and CTLA-4 on the cell surface by MGD019 measured by DiscoverX™ enzyme complementation assay.

Results

Enhanced Blockade of CTLA-4 by MGD019 on Dual Expressing Cells

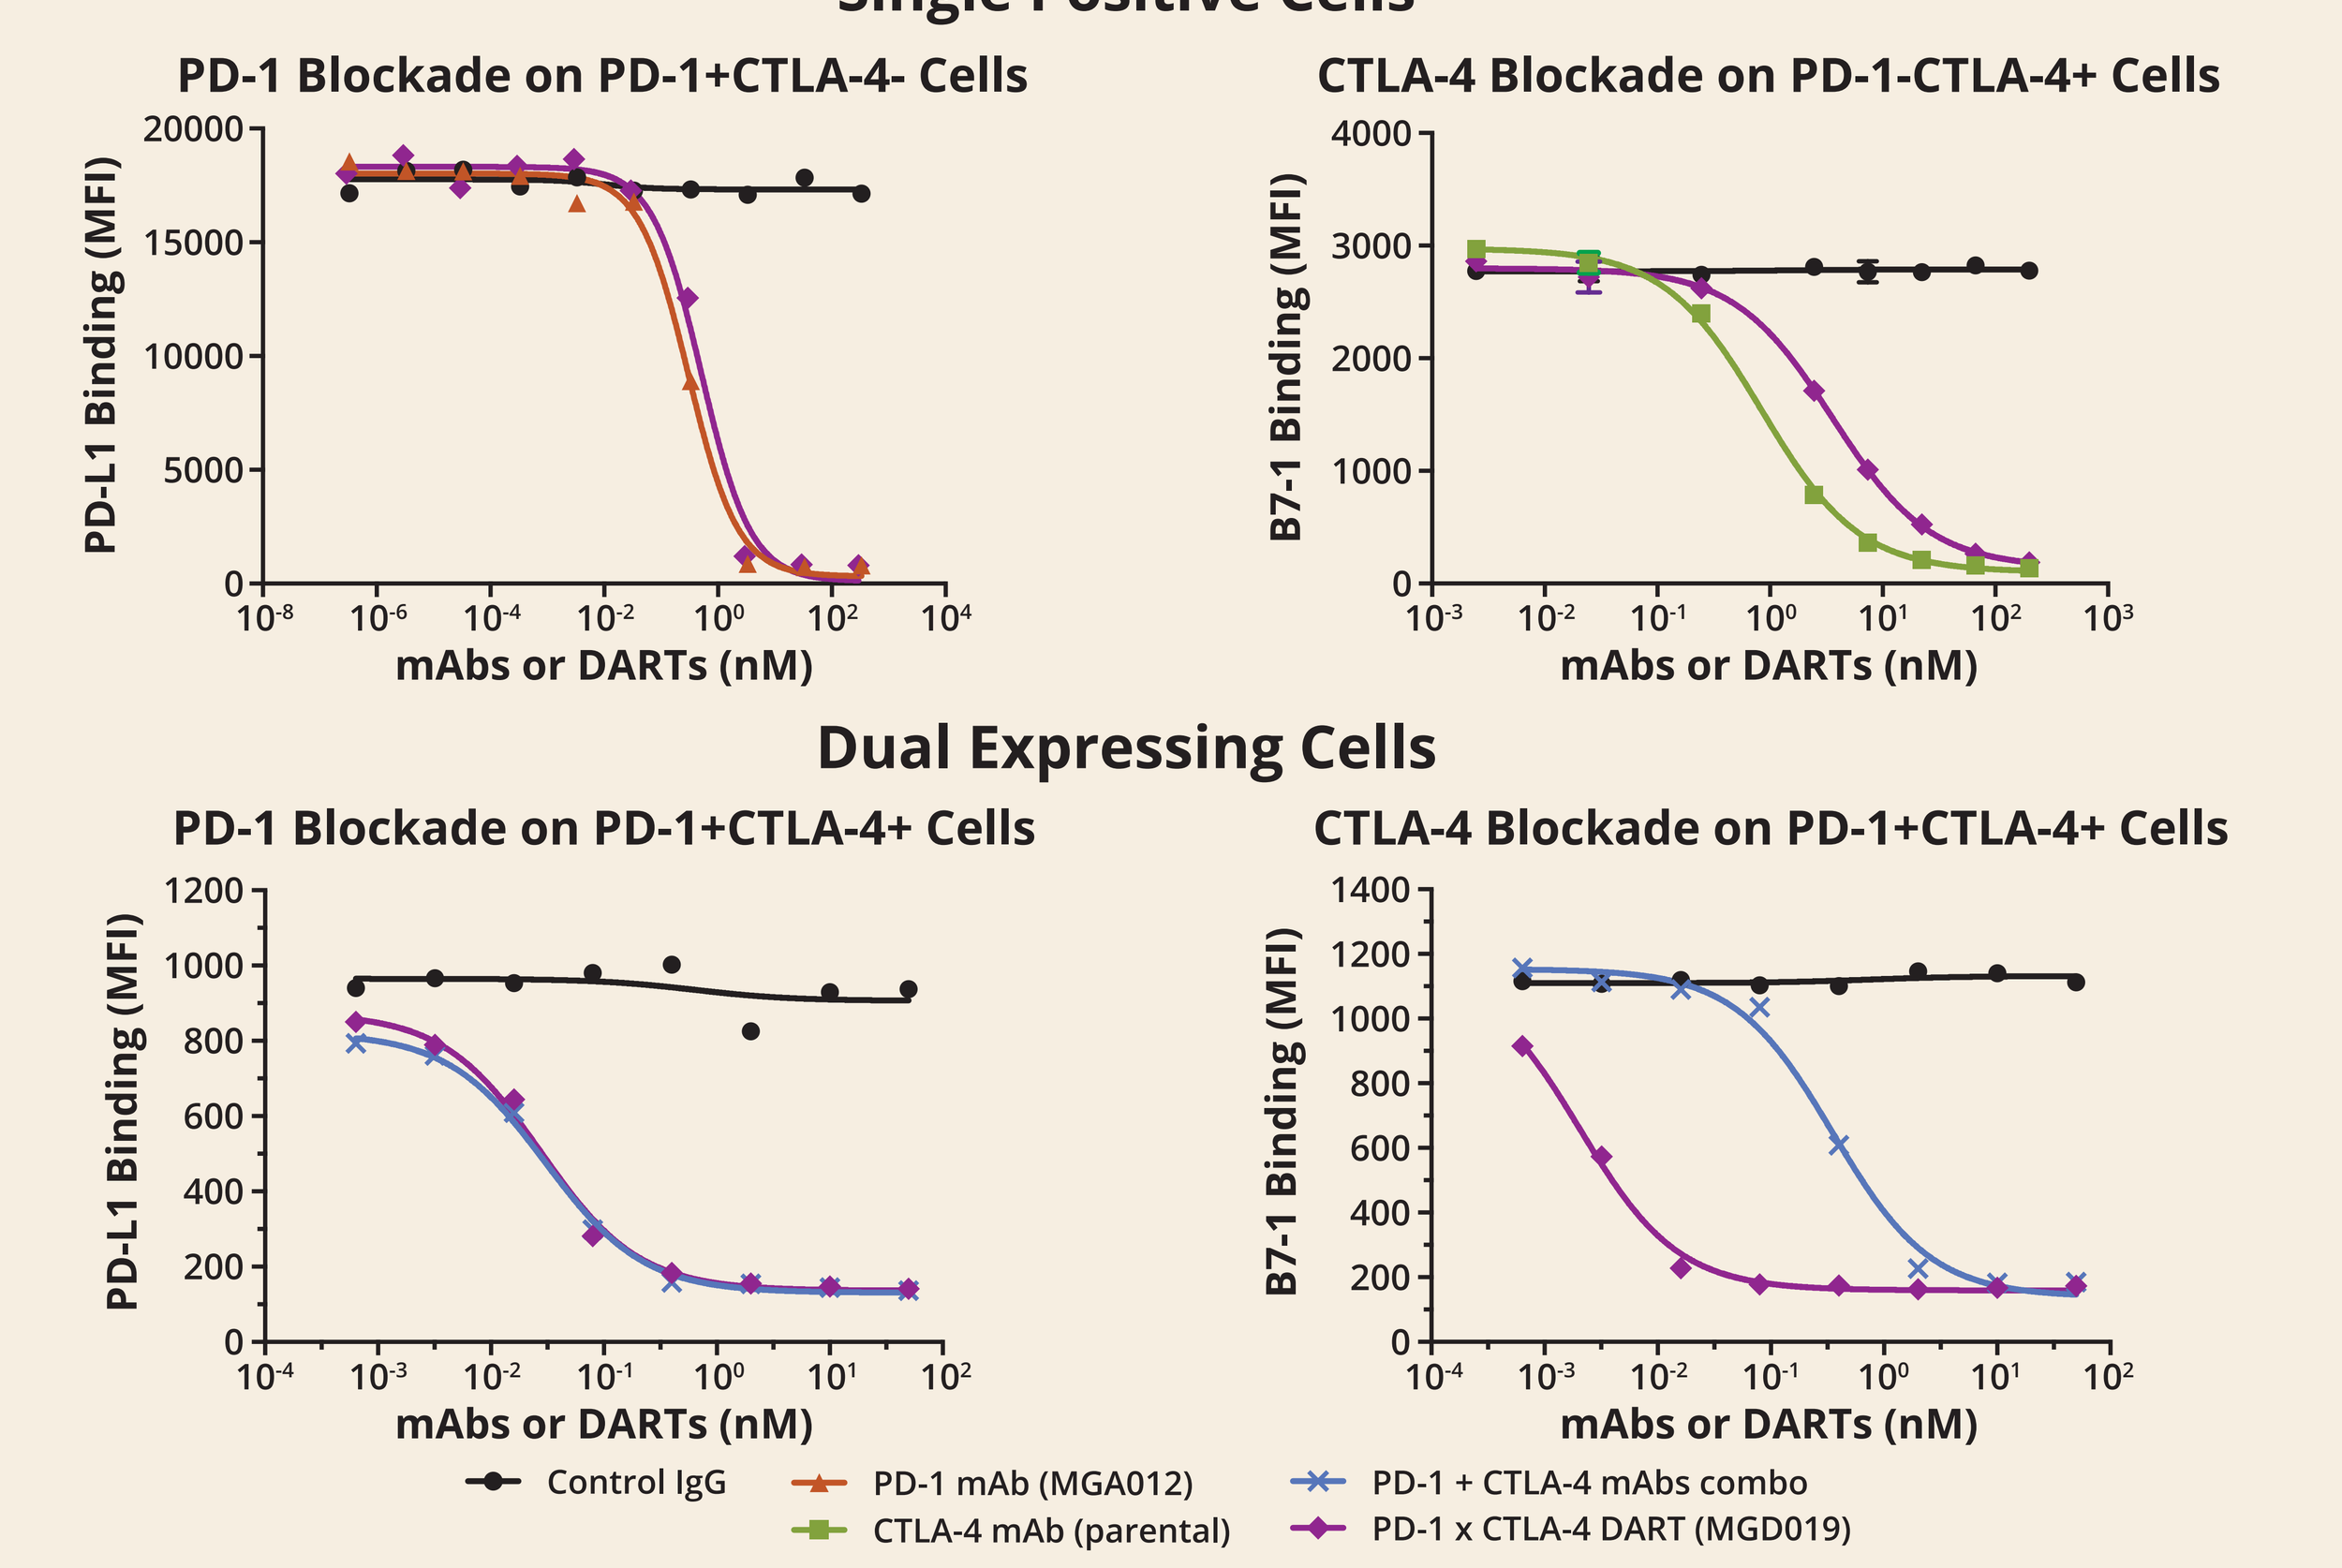
Single Positive Cells
PD-1 Blockade on PD-1+CTLA-4- Cells
CTLA-4 Blockade on PD-1-CTLA-4+ Cells
Dual Expressing Cells
PD-1 Blockade on PD-1+CTLA-4+ Cells
CTLA-4 Blockade on PD-1+CTLA-4+ Cells
PD-L1 Binding (MFI)
B7-1 Binding (MFI)
mAbs or DARTs (nM)
Control IgG
PD-1 mAb (MGA012)
CTLA-4 mAb (parental)
PD-1 + CTLA-4 mAbs combo
PD-1 x CTLA-4 DART (MGD019)

PD-1 or CTLA-4 binding to their respective soluble ligands on Jurkat cells engineered to express PD-1 and/or CTLA-4 was measured by flow cytometry.

MGD019 Enhances T-cell Activation

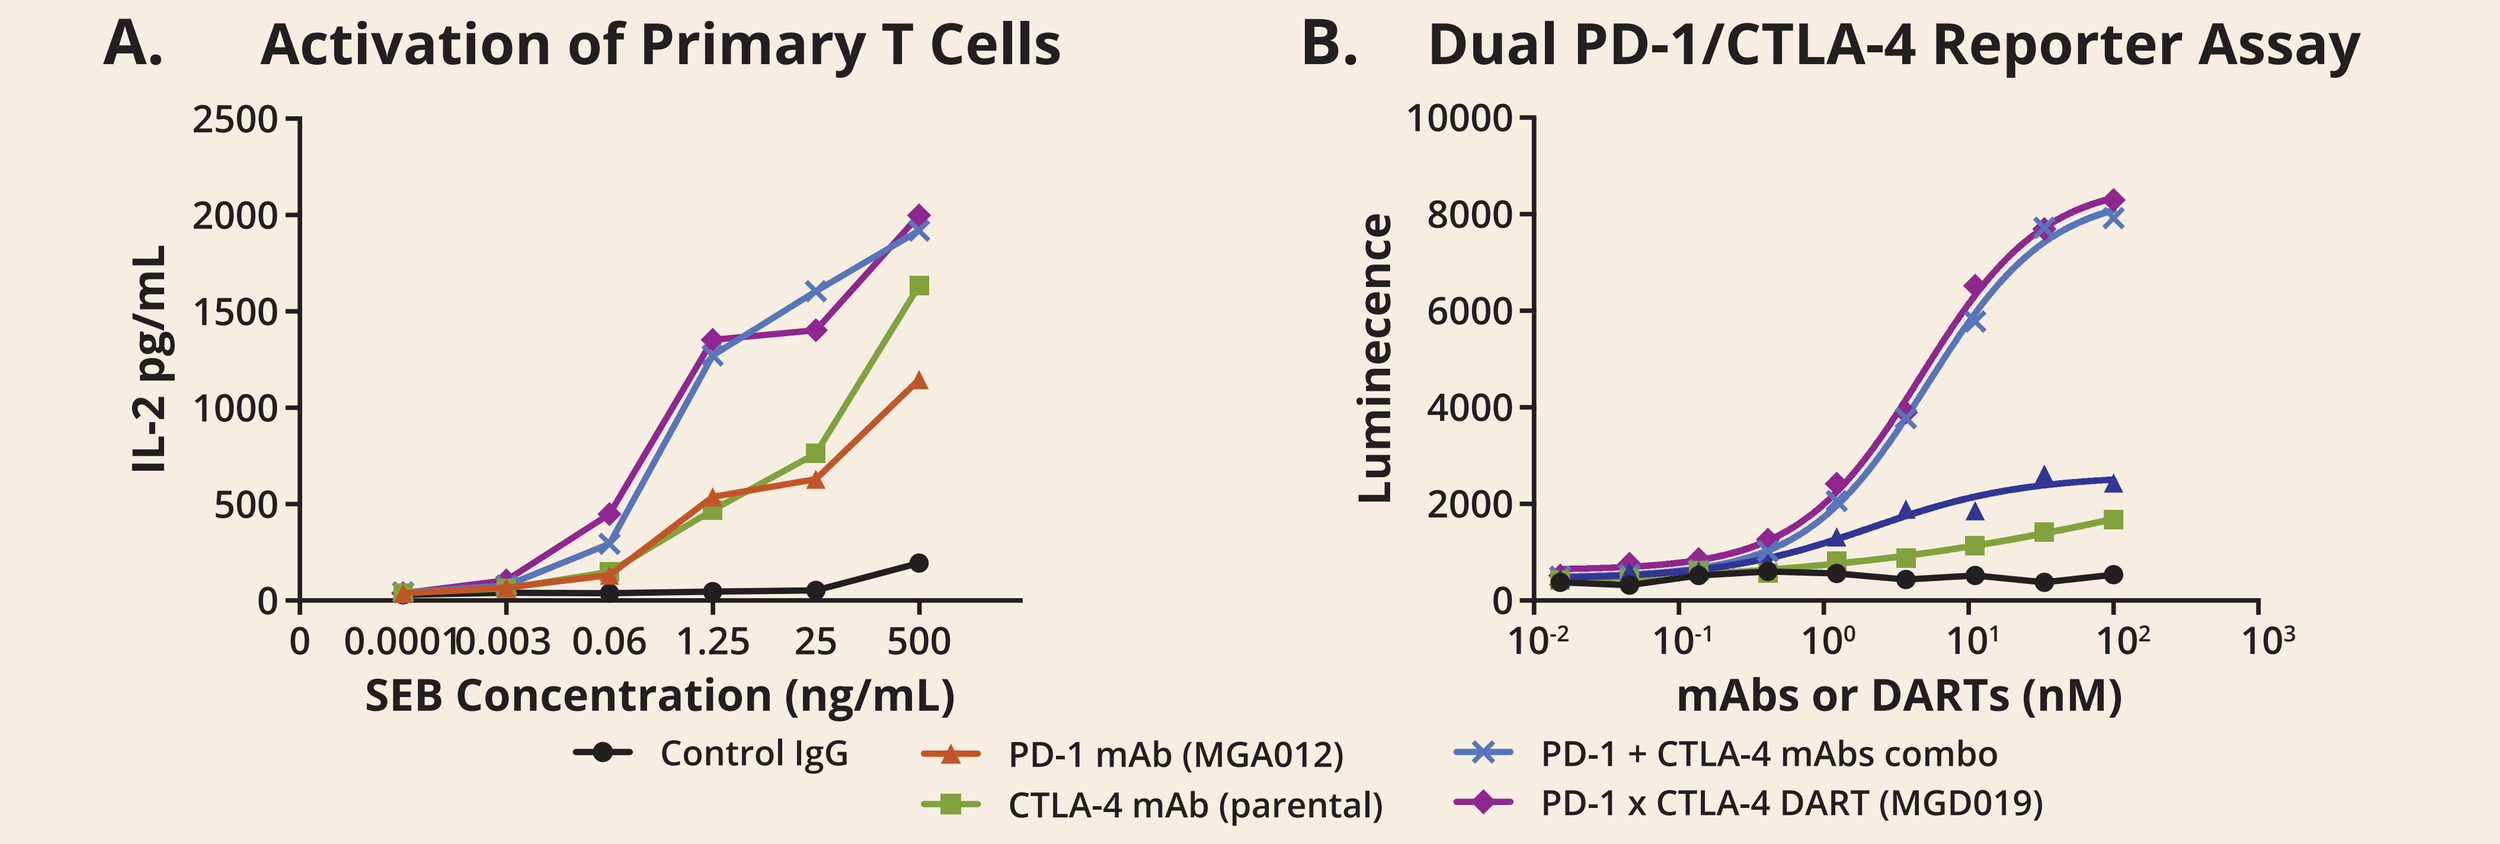
A. Activation of Primary T Cells
B. Dual PD-1/CTLA-4 Reporter Assay
IL-2 pg/mL
SEB Concentration (ng/mL)
Luminecence
mAbs or DARTs (nM)
Control IgG
PD-1 mAb (MGA012)
CTLA-4 mAb (parental)
PD-1 + CTLA-4 mAbs combo
PD-1 x CTLA-4 DART (MGD019)

A. PBMC were activated by SEB in the presence of 10 μg/mL of the indicated molecules. IL-2 secretion was measured at 96h by ELISA. **B.** Expression of IL-2 reporter cassette in Jurkat cells co-expressing PD-1 and CTLA-4 (Promega) co-incubated with APC in the presence of the indicated molecules.

Characterization of MGD019-Enhanced T-cell Activation

Tumor Microenvironment Models

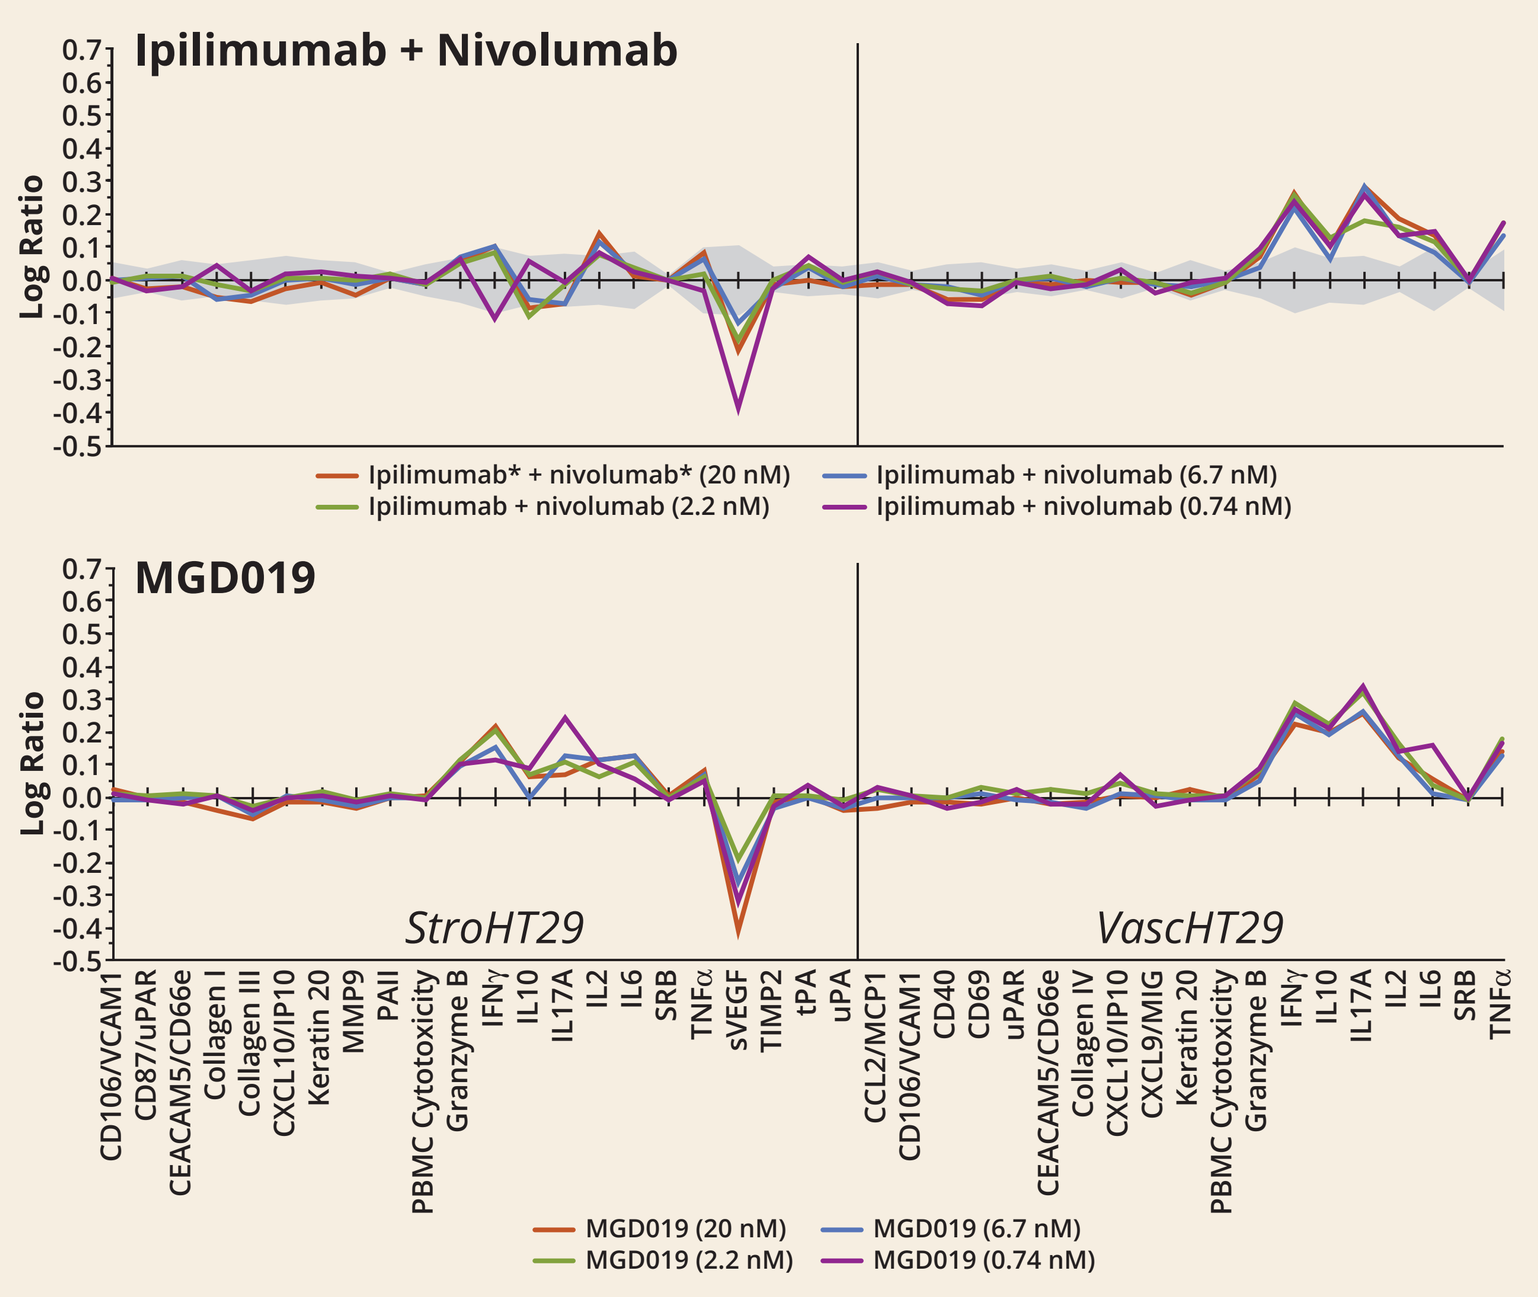
Ipilimumab + Nivolumab
MGD019
Log Ratio
StroHT29
VascHT29
Ipilimumab* + nivolumab* (20 nM)
Ipilimumab + nivolumab (2.2 nM)
Ipilimumab + nivolumab (6.7 nM)
Ipilimumab + nivolumab (0.74 nM)
MGD019 (20 nM)
MGD019 (2.2 nM)
MGD019 (6.7 nM)
MGD019 (0.74 nM)

DiscoverX StroHT29 and VascHT29 TME models: HT-29 colorectal cells were cultured with stromal cells and PBMCs to recapitulate a tumor microenviroment (DiscoverX). Protein expression was measured following treatment with mAbs or MGD019. *Replicas of nivolumab and ipilimumab were constructed on the basis of the published sequences.

Transcriptional Changes in Allo-activated T Cells

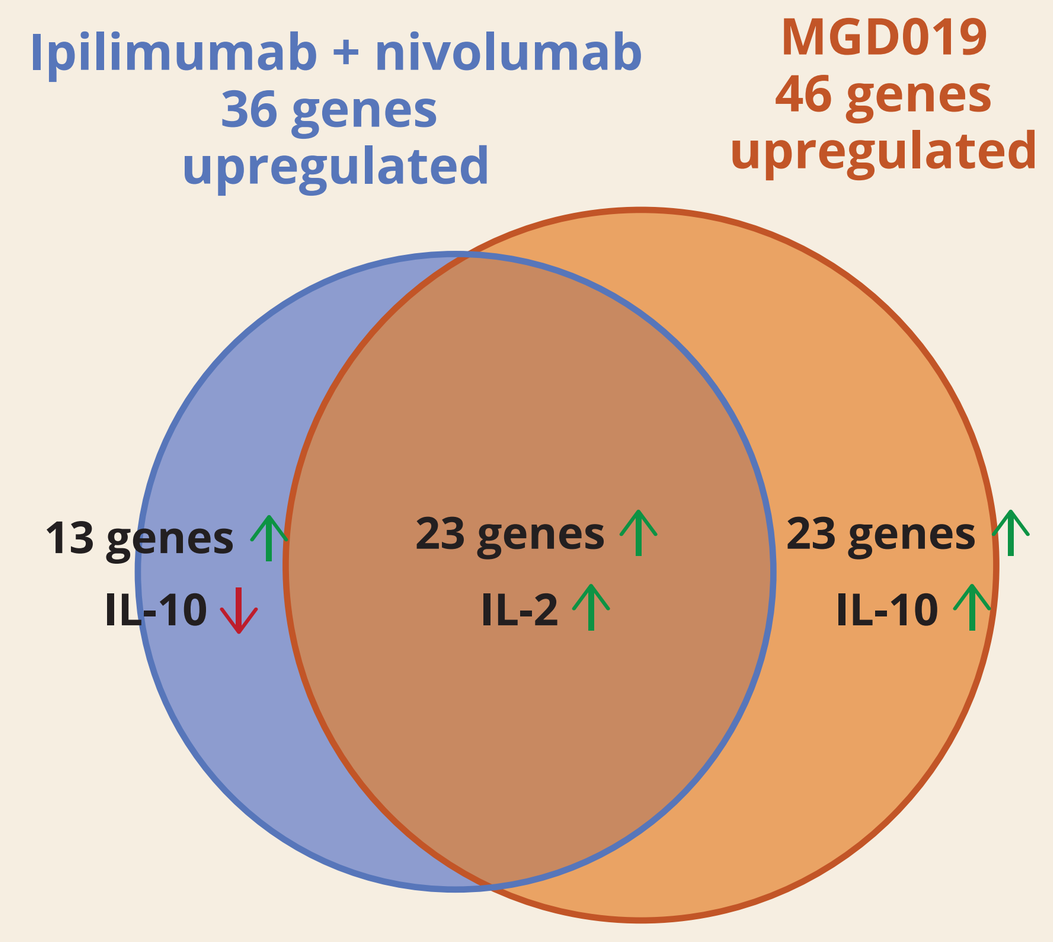
Ipilimumab + nivolumab
36 genes
upregulated
MGD019
46 genes
upregulated
13 genes ↑
IL-10 ↓
23 genes ↑
IL-2 ↑
23 genes ↑
IL-10 ↑

Transcriptional changes in MLR assay: T cells were re-isolated from 72h MLR assay after treatment with 1 μg/mL each ipilimumab and nivolumab or MGD019 at 1 μg/mL, and RNAseq was performed using Illumina HiSeq platform. Gene alignment, filtering for 750 immune-related genes and differential expression analysis were done using Strand NGS software. Genes were clustered based on upregulation in both donors.

MGD019 GLP Toxicology Study

Dose Level (mg/kg)	Dose Route	Dose Days	No. of Animals: Terminal Necropsy (Week 4)	No. of Animals: Recovery Necropsy (Week 14)
0 (vehicle)	IV infusion for 30 minutes	1, 8, 15, 22	3M / 3F	2M / 2F
10			3M / 3F	2M / 2F
40			3M / 3F	2M / 2F
100			3M / 3F	2M / 2F

Finding	MGD019 (PD-1xCTLA-4)			MGA012 (PD-1)
	10 mg/kg	40 mg/kg	100 mg/kg	≥100 mg/kg
Adverse clinical signs	-	-	-	-
Body weight loss	-	-	-	-
Increased spleen weight	+	++	++	-
Lymphoid hyperplasia in spleen	-	+	++	-
GI tract inflammation	-	-	-	-
Circulating cytokines	-	-	-	-
T cell proliferation (Ki67+)	+	++	++	+/-

- MGD019 is relatively well-tolerated compared to prior reported PD1+CTLA4 mAb combination[1]

MGD019 Pharmacokinetics and Receptor Occupancy

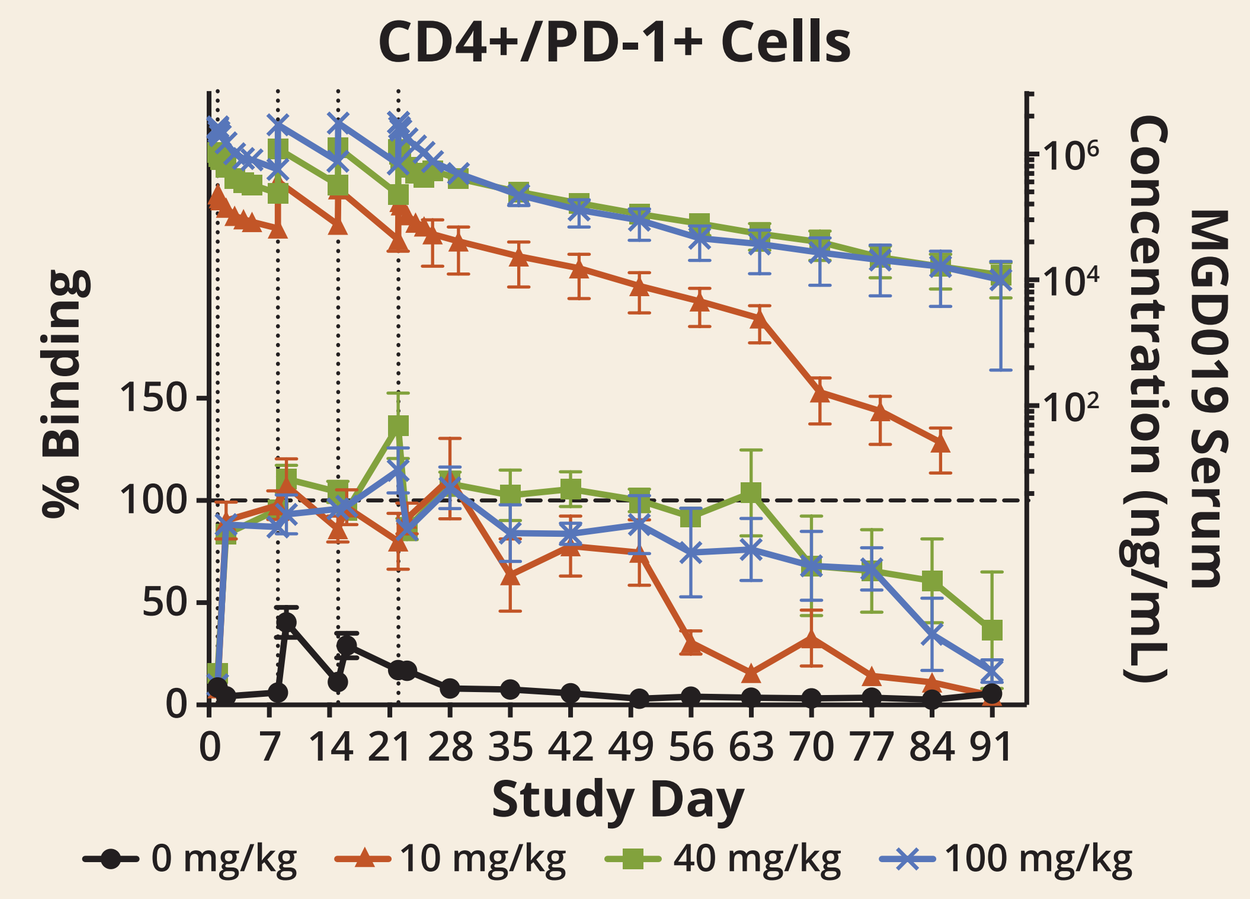
CD4+/PD-1+ Cells
% Binding
MGD019 Serum Concentration (ng/mL)
Study Day
0 mg/kg
10 mg/kg
40 mg/kg
100 mg/kg

- MGD019 demonstrates good pharmacokinetics consistent with IgG4 Fc backbone
- MGD019 receptor occupancy correlate with its serum concentration

Cynomolgus monkey (3F/3M) were infused with 10, 40 or 100 mg/kg/dose MGD019 at Day 1, 8, 15, and 22. Serum concentration was measured by ELISA (right axis) and receptor occupancy was measured by flow cytometry (left axis).

MGD019 Supports Homeostatic Proliferation of T Cells In Vivo

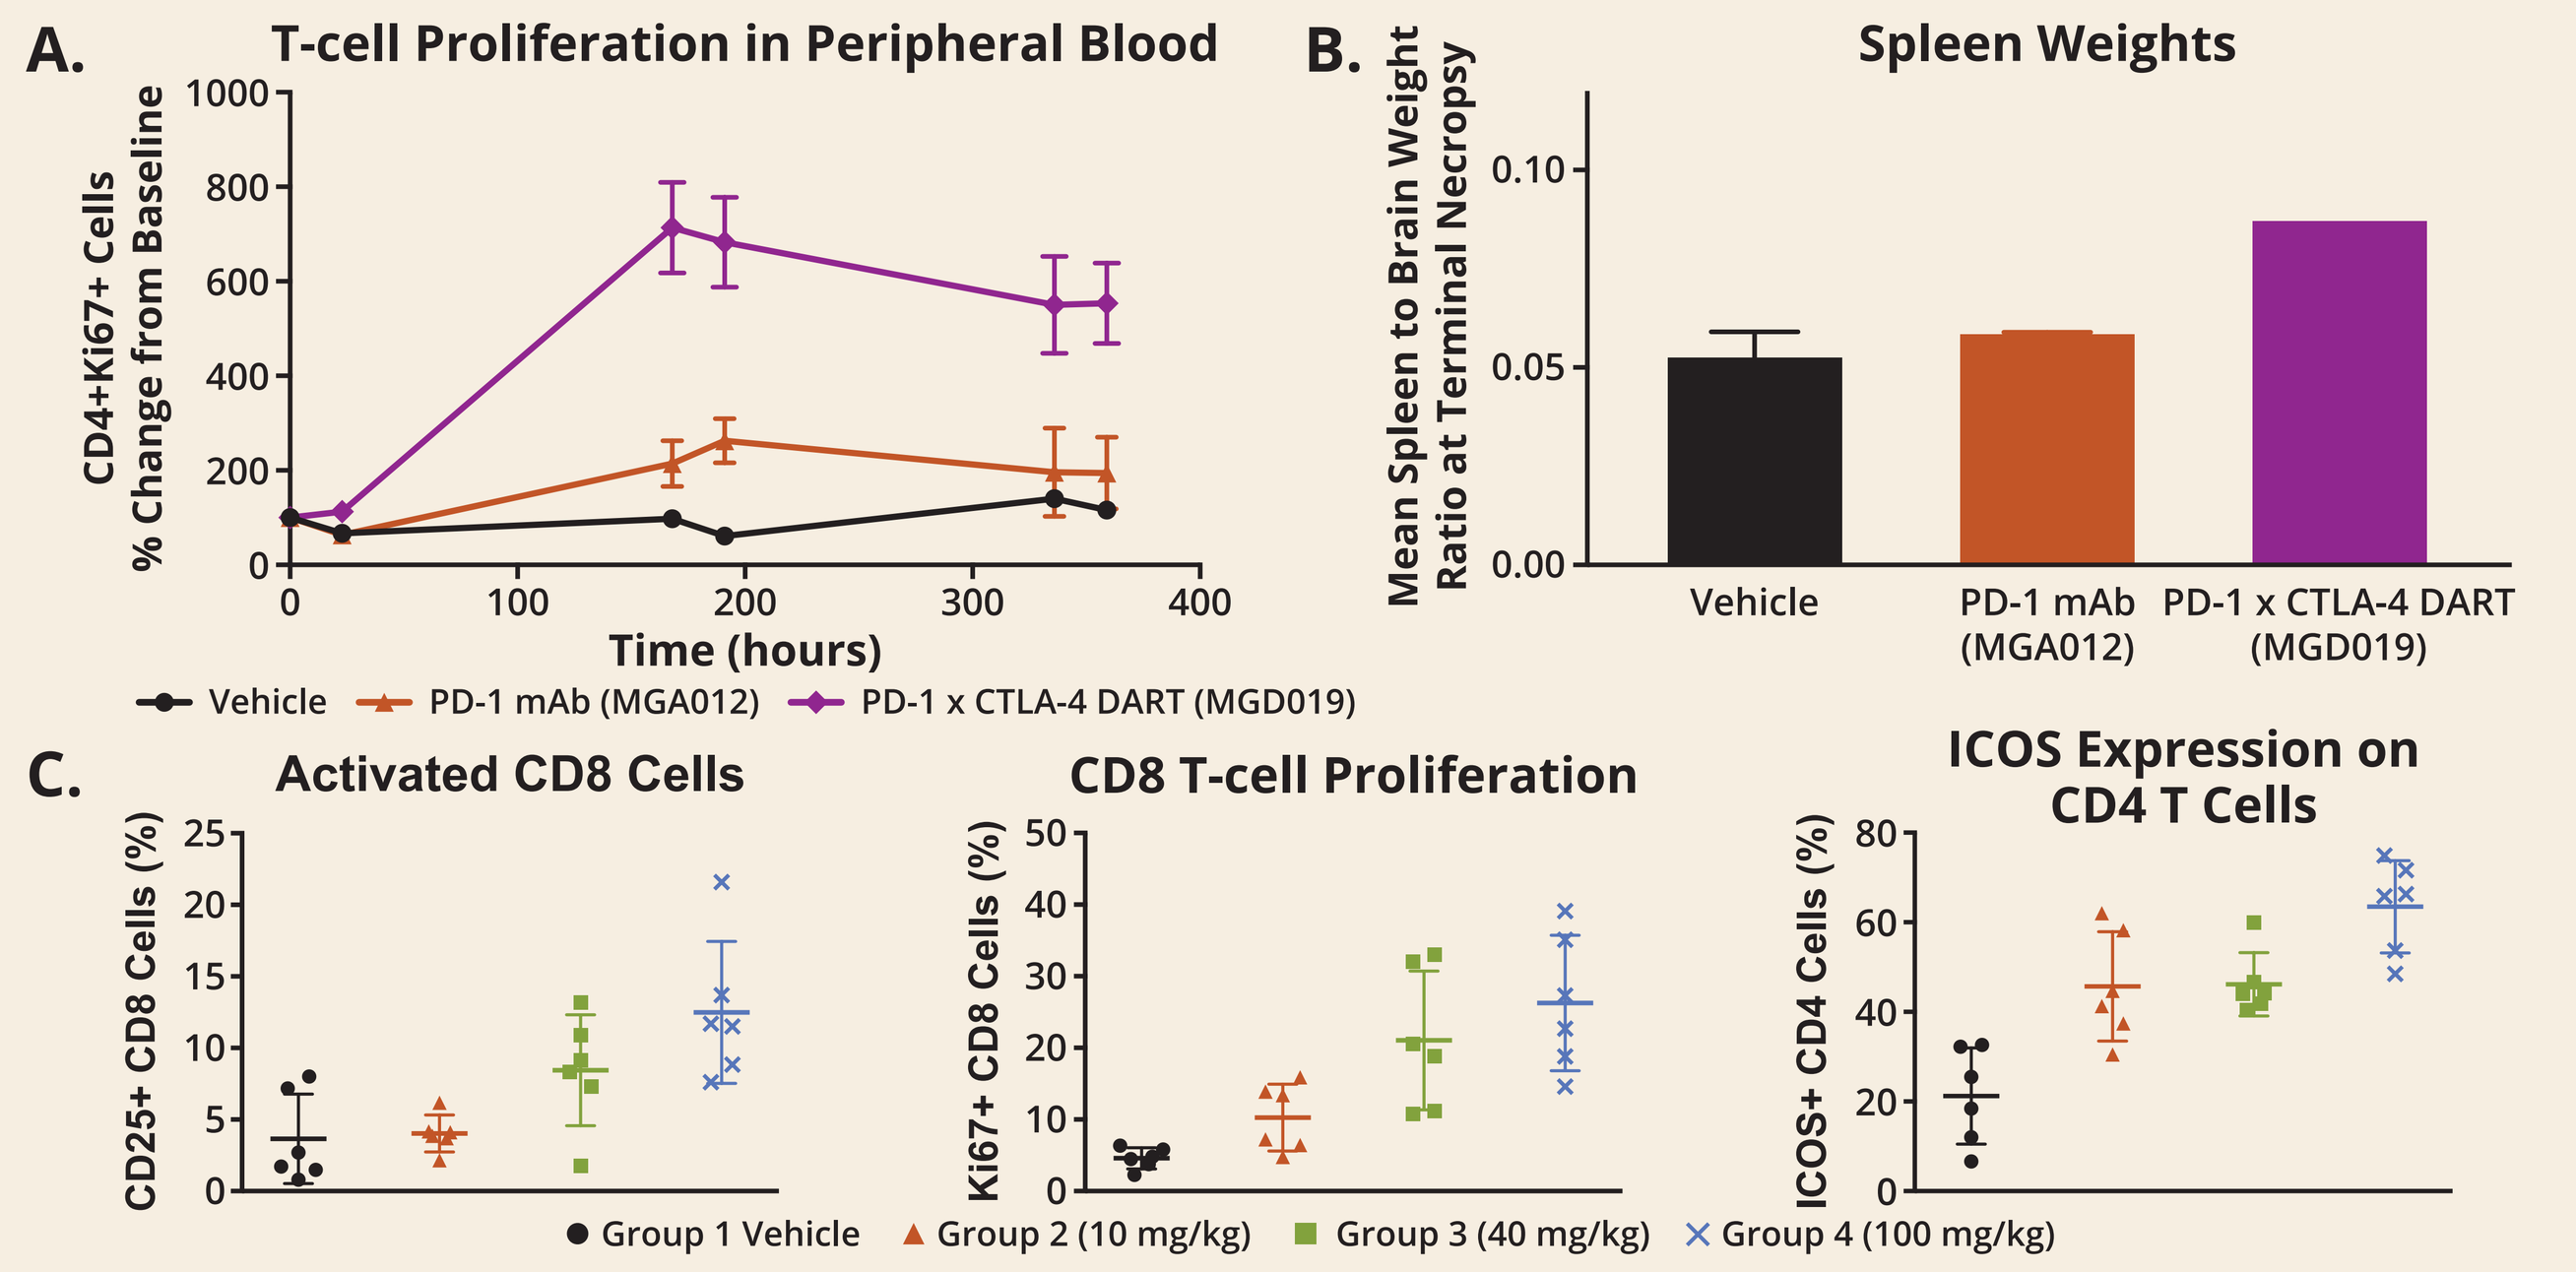
A. T-cell Proliferation in Peripheral Blood
CD4+Ki67+ Cells % Change from Baseline
Time (hours)
Vehicle
PD-1 mAb (MGA012)
PD-1 x CTLA-4 DART (MGD019)
B. Spleen Weights
Mean Spleen to Brain Weight Ratio at Terminal Necropsy
C. Activated CD8 Cells
CD8 T-cell Proliferation
ICOS Expression on CD4 T Cells
CD25+ CD8 Cells (%)
Ki67+ CD8 Cells (%)
ICOS+ CD4 Cells (%)
Group 1 Vehicle
Group 2 (10 mg/kg)
Group 3 (40 mg/kg)
Group 4 (100 mg/kg)

A. Cynomolgus monkeys were infused IV Q1W for 3 weeks with 75 mg/kg MGD019 (3M/3F) and, in a separate study, 100 mg/kg MGA012 (anti-PD-1, 2M/2F). Ki67 expression was quantified by flow cytometry. **B.** Spleen weights at terminal necropsy were calculated as fraction of brain weight. **C.** Cynomolgus monkey were injected weekly with indicated amounts of MGD019. Shortly after 4th injection, splenocytes of necropsied animals were analyzed for expression of CD25, Ki-67, and ICOS.

MGD019 Induces Memory T Cells In Vivo

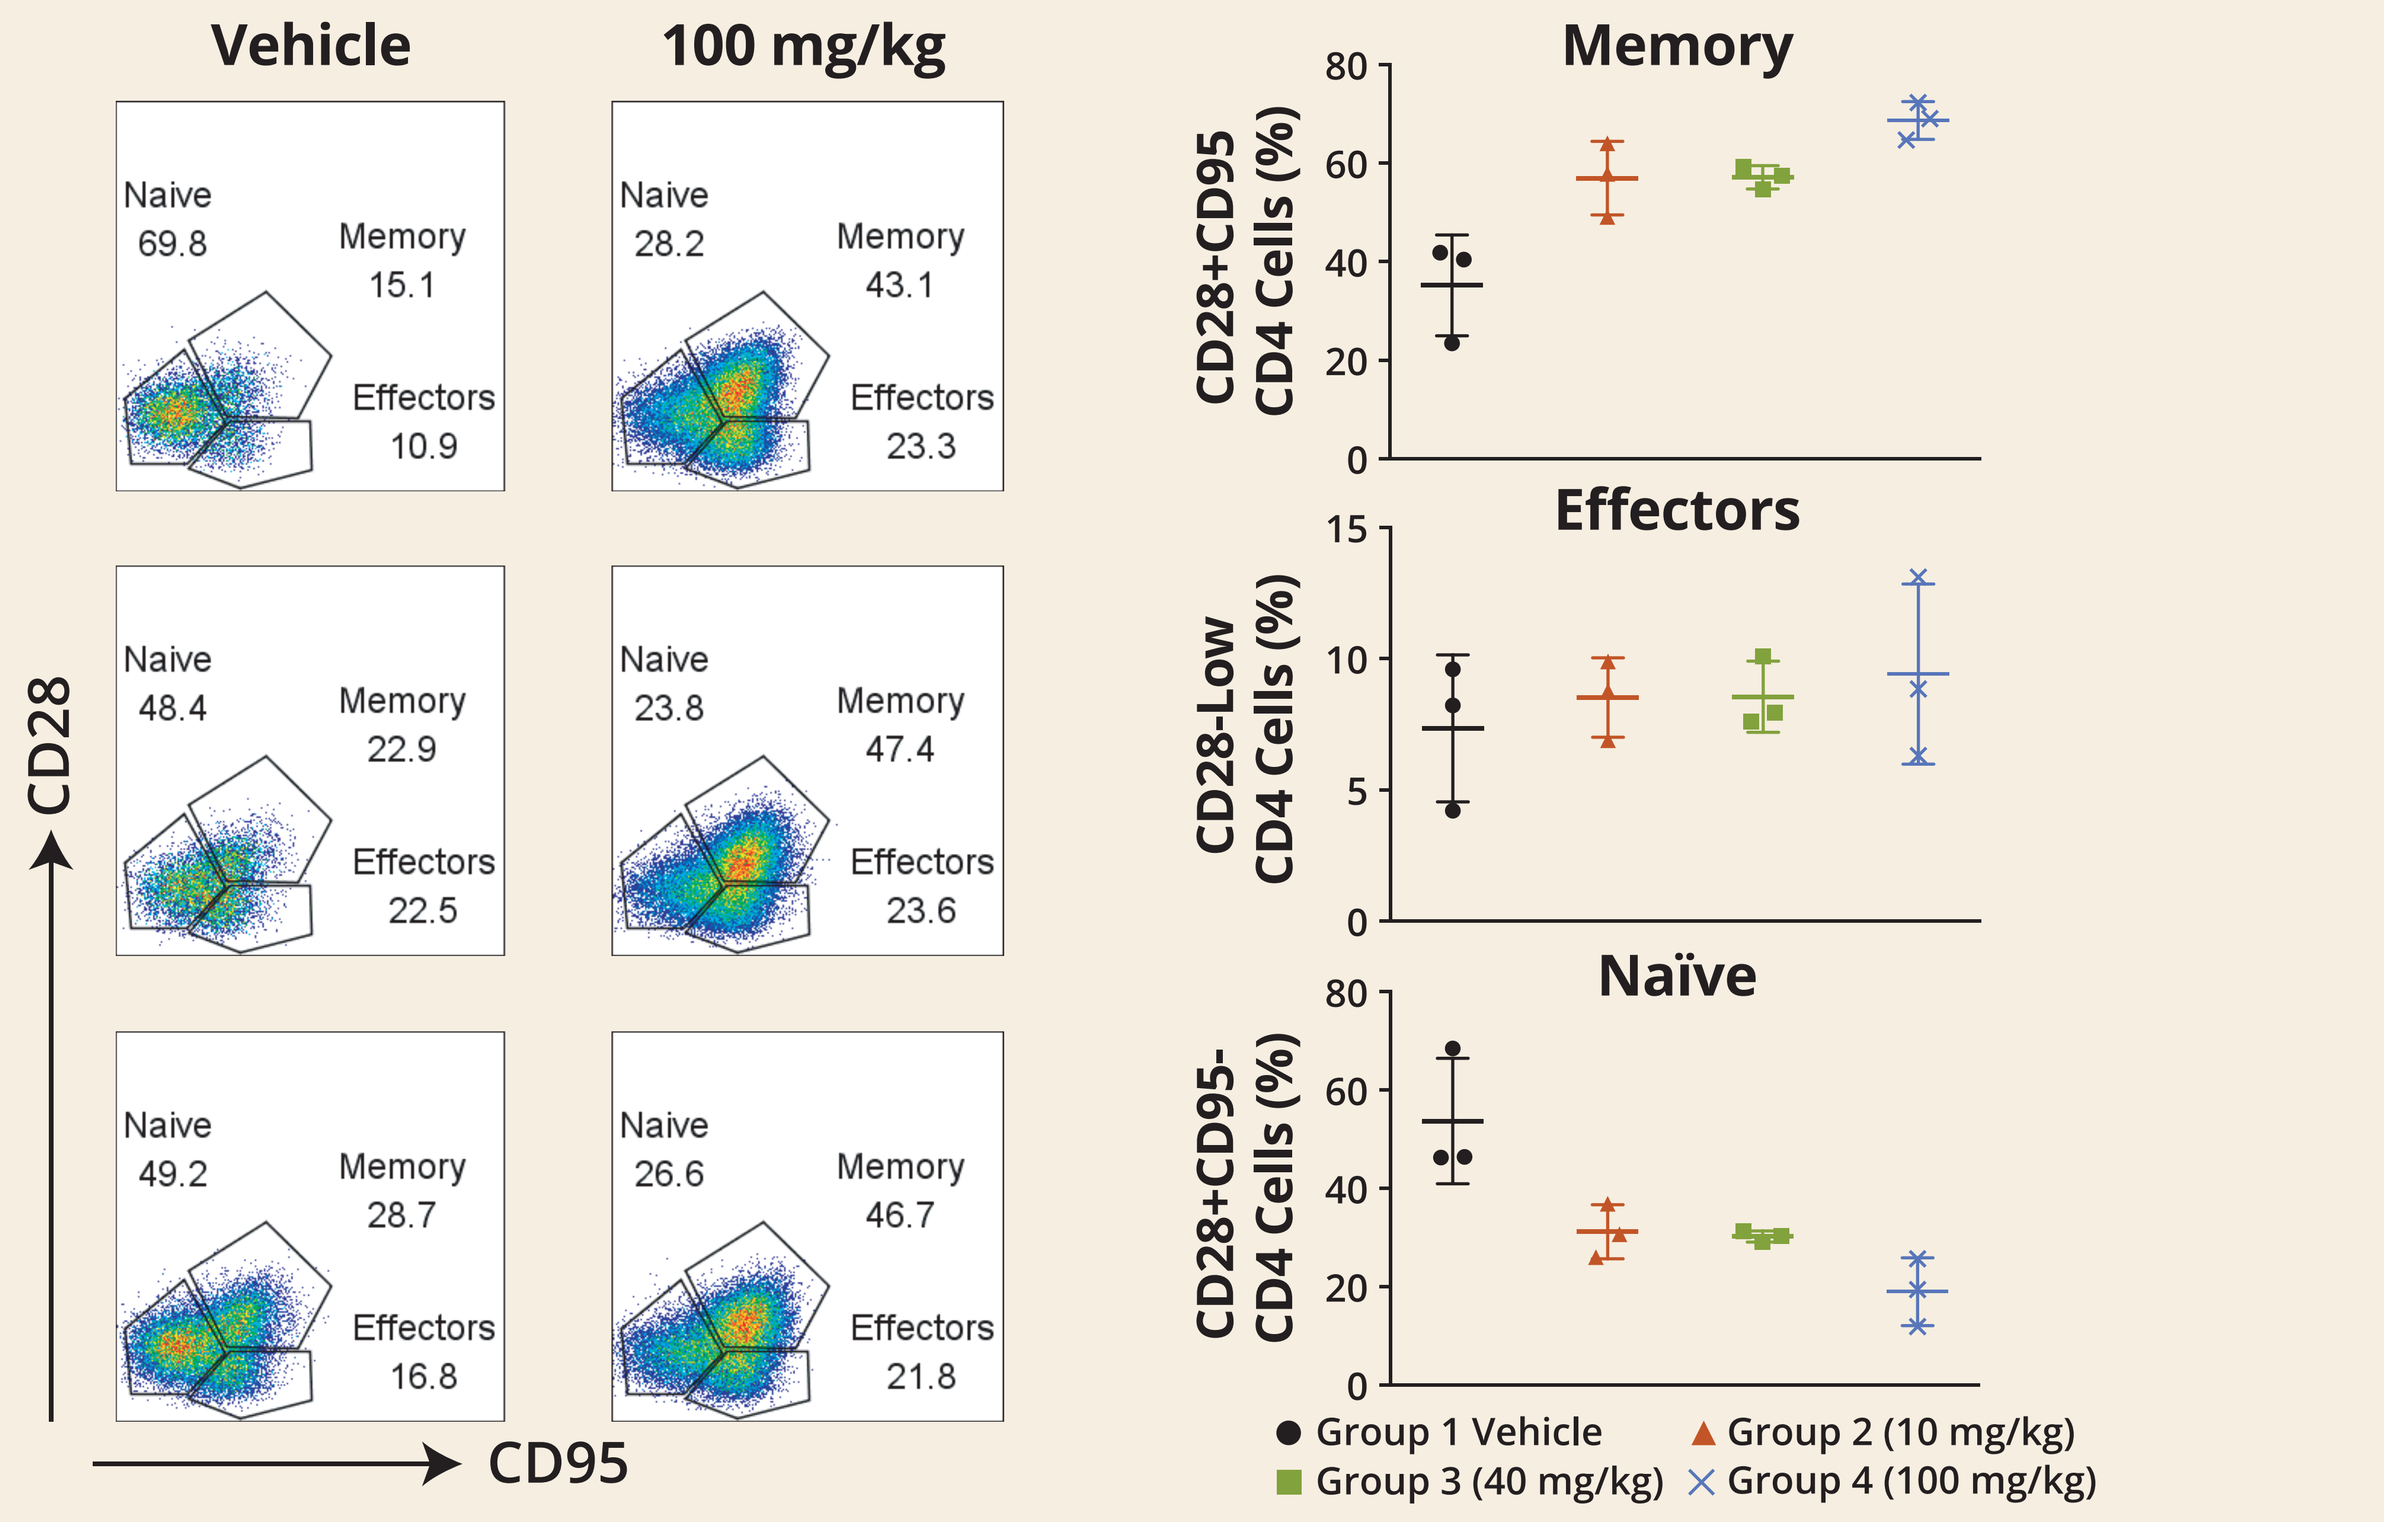
Vehicle
100 mg/kg
Naive
Memory
Effectors
CD28
CD95
Memory
Effectors
Naïve
CD28+CD95 CD4 Cells (%)
CD28-Low CD4 Cells (%)
CD28+CD95- CD4 Cells (%)
Group 1 Vehicle
Group 2 (10 mg/kg)
Group 3 (40 mg/kg)
Group 4 (100 mg/kg)

Cynomolgus monkeys were injected weekly with indicated amounts of MGD019. Shortly after 4th injection, T cells of necropsied animals were analyzed for expression of CD28 and CD95 by flow cytometry.

MGD019 Phase 1 Study (Started 2H2018)

An ongoing Phase 1, open-label study will characterize safety, dose-limiting toxicities (DLTs), and maximum tolerated/administered dose (MTD/MAD) of MGD019. Dose escalation will occur in a 3+3+3 design in patients with advanced solid tumors of any histology. Once the MTD/MAD is determined, a Cohort Expansion Phase will be enrolled to further characterize safety and initial anti-tumor activity in patients with specific tumor types anticipated to be sensitive to dual checkpoint blockade.

Conclusions

- T cells co-expressing PD-1 and CTLA-4 are more prevalent in tumors compared to healthy tissues
- MGD019 binds to and blocks its targets with increased activity on dual PD-1/CTLA-4-expressing cells
- MGD019 induces a unique transcriptional activation pattern partially overlapping that induced by the combination of individual mAbs
- In cynomolgus monkeys, MGD019 demonstrates IgG4-like PK and a good safety profile similar to that observed with PD-1 blockade alone, while demonstrating biological effects of CTLA-4 antagonism

Reference

1. Selby M. et al. Preclinical Development of Ipilimumab and Nivolumab Combination Immunotherapy: Mouse Tumor Models, In Vitro Functional Studies, and Cynomolgus Macaque Toxicology. PLoS One. 2016 Sep 9;11(9):e0161779:

Group	M/F	Treatment	Dose (mg/kg)	Diarrhea (n/N)	Mean SpleenWeight (g) Day 30 M/F	Mean SpleenWeight (g) Day 59 M/F	Spleen Pathology (n/N)	Gastrointestinal Pathology (n/N)
1	5/5	Saline control	—	0/10	3.9/2.8	3.5/3.7	0/6	0/6
2	5/5	Nivolumab + ipilimumab	10 3	2/10	4.0/3.6	4.3/2.4	2/6	2/6
3	5/5	Nivolumab + ipilimumab	50 10	4/10	6.1/4.47	7.5/3.2	4/5	3/5

Presented at the 2019 Keystone Symposia Conference, "Uncovering Mechanisms of Immune-Based Therapy in Cancer and Autoimmunity", February 18–22, 2019, Breckenridge, Colorado

©2019 MacroGenics, Inc. All rights reserved.