Anti-B7-H3 Antibody-Drug Conjugates as Potential Therapeutics for Solid Cancer

Deryk Loo, Juniper Scribner, Thomas Son, Jeff Hooley, Tim Hotaling, Michael Chiechi, Pam Li, Anushka De Costa, Yan Chen, Ann Easton, Francine Chen, Bhaswati Barat, Valentina Ciccarone, James Tamura, Mark Kubik, Scott Koenig, Syd Johnson, Paul A. Moore, and Ezio Bonvini

MacroGenics, Inc., Rockville, MD and South San Francisco, CA

1201

http://ir.macrogenics.com/events.cfm

Abstract

Introduction: Monoclonal antibodies (mAbs) were generated via a target-unbiased approach based on intact cell immunization with cell lines, fetal progenitor cells, and cancer stem cells. An immunohistochemical (IHC) screen for cancer-specific candidates identified a panel of anti-B7-H3 (CD276) mAbs with highly differential tumor-versus-normal tissue binding. B7-H3 expression was observed in tumor epithelium as well as tumor-associated vasculature and stroma. Consistent with our findings, B7-H3 has been reported to be overexpressed in a growing number of solid cancers, including breast, lung, pancreatic, prostate, kidney, and colon cancer, as well as melanoma and glioblastoma. Furthermore, overexpression of B7-H3 has been correlated with disease severity and poor outcome in a number of these cancer types. A humanized version of an anti-B7-H3 mAb engineered with an enhanced Fc domain (enoblituzumab or MGA271[1]) and a humanized Dual-Affinity Re-Targeting (DART®) protein that recognizes both B7-H3 and CD3 and redirects T cells to kill B7-H3-expressing cells (MGD009) are being investigated in Phase 1 clinical studies. In this nonclinical study, we evaluated the therapeutic potential of anti-B7-H3 antibody-drug conjugates (ADCs) toward B7-H3-expressing solid cancers.

Methods: A panel of anti-B7-H3 mAbs was screened for internalization and a subset of mAbs that were efficiently internalized by tumor cells was identified. These mAbs were converted to ADCs via chemical conjugation; in vitro and in vivo activity studies were then conducted with a range of tumor cell lines representing human cancer types that overexpress B7-H3.

Results: The anti-B7-H3 ADCs exhibited specific, dose-dependent cytotoxicity toward B7-H3-positive tumor cell lines in vitro, including breast, lung, ovarian, pancreatic, and prostate cancer lines, with IC_{50} values generally in the sub-nM range. Cytotoxicity was not observed with cell lines lacking B7-H3 expression. The anti-B7-H3 ADCs exhibited potent antitumor activity in vivo, resulting in tumor stasis and tumor regression in mice bearing B7-H3-positive human breast, lung, and ovarian tumor xenografts.

Conclusion: Anti-B7-H3 ADCs exhibited dose-dependent cytotoxicity in vitro and potent antitumor activity in vivo toward a range of B7-H3-expressing tumor cell lines representing cancer types that overexpress B7-H3. Our findings demonstrate that ADCs targeting B7-H3 may serve as potential therapeutics for B7-H3-expressing solid cancers.

Introduction

- mAbs were generated via immunization of mice with viable human fetal progenitor cells or tumor initiating/cancer stem-like cells (CSLCs)
- An IHC screen for cancer-specific mAbs identified a panel of anti-B7-H3 (CD276) reactive mAbs with highly differential tumor-versus-normal tissue binding
- Strong B7-H3 expression was observed in tumor epithelium of a large range of solid cancers, as well as in tumor-associated vasculature and stroma
- A subset of anti-B7-H3 mAbs were efficiently internalized by tumor cells in vitro. Together with their favorable tumor-versus-normal tissue reactivity, this subset was selected for evaluation for an ADC approach
- **Objective:** Evaluate the therapeutic potential of anti-B7-H3 ADCs toward solid cancer by conducting in vitro and in vivo activity studies with anti-B7-H3 ADCs across a range of tumor cell lines representing human cancer types that overexpress B7-H3

Targeting B7-H3 via Multiple Mechanisms of Action

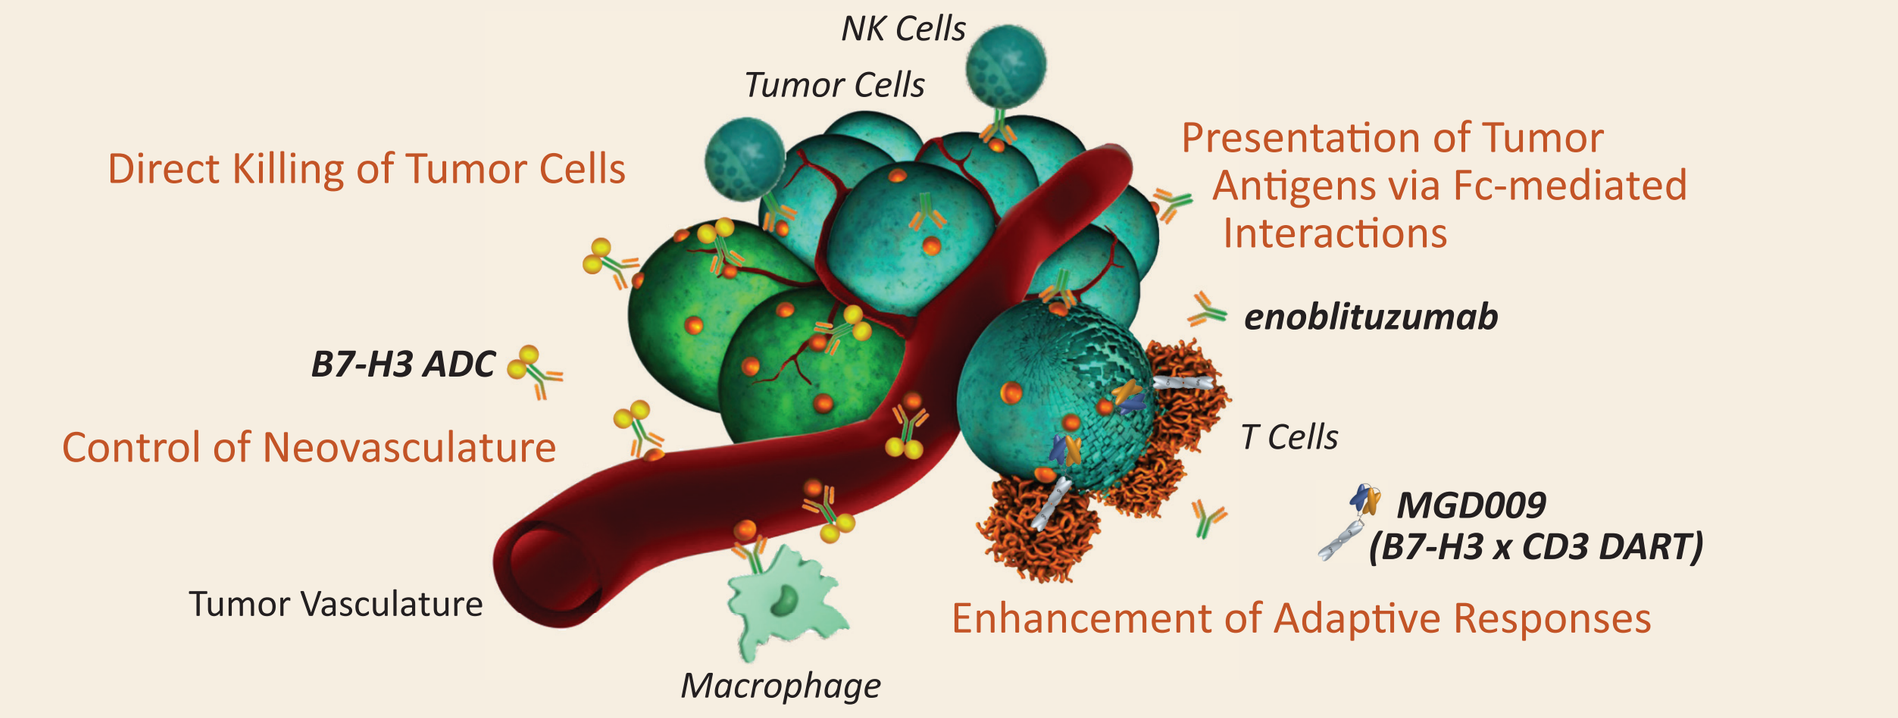
NK Cells
Tumor Cells
Direct Killing of Tumor Cells
Presentation of Tumor Antigens via Fc-mediated Interactions
enoblituzumab
B7-H3 ADC
Control of Neovasculature
T Cells
MGD009 (B7-H3 x CD3 DART)
Tumor Vasculature
Enhancement of Adaptive Responses
Macrophage

Methods

- mAb generation
 - Immunizations with intact viable fetal progenitor and tumor initiating/CSLCs were performed as previously described[2]
- Internalization assay
 - Internalization was performed in a 5-day assay using saporin-conjugated anti-mouse Fab at 1:1 or 10:1 Fab-ZAP:Test mAb ratio, as per manufacturer's protocol (Advanced Targeting Systems)
- ADC generation
 - Anti-B7-H3 mAbs (chimeric human IgG1) were converted to ADCs via cysteine-conjugation to the cleavable auristatin E linker/payload vc-MMAE to an average drug-to-antibody ratio of 4.5-4.7 (Concortis Biosystems)
- In vitro cytotoxicity
 - 7-day in vitro cytotoxicity was quantified using Alamar Blue reagent, according to manufacturer's protocol (BioRad)
- In vivo activity
 - Tumor cells (5 x 10^6) were implanted subcutaneously into the flank of CD1 nude mice. When tumors reached ~150 mm^3, mice were randomized and test articles were administered intraperitoneally. Tumors were measured twice weekly by orthogonal measurements with electronic calipers, with tumor volumes calculated as: (length x width x height)/2
- Pharmacokinetics
 - Non-tumor bearing CD1 nude mice were administered test articles intraperitoneally at a single dose of 5 mg/kg. Blood samples were collected over the course of 10 days and sandwich ELISAs were performed on sera to quantify total mAb and intact ADC concentrations

Antibody/Target Discovery Platform

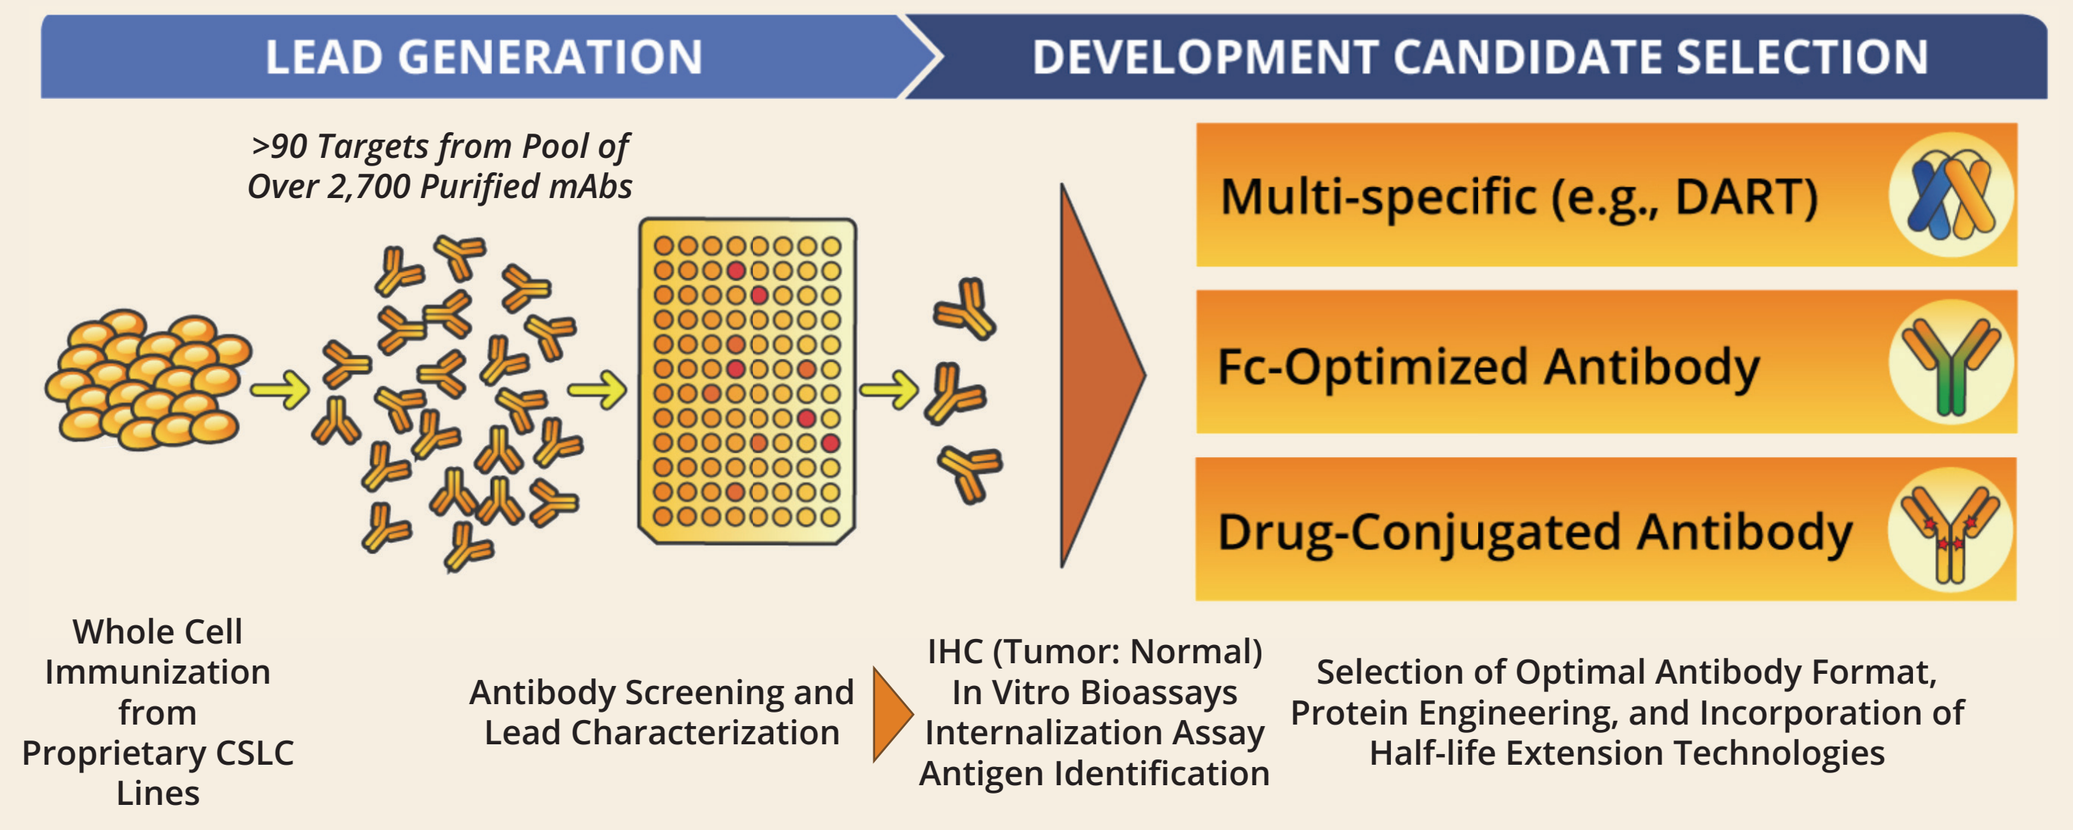
LEAD GENERATION
DEVELOPMENT CANDIDATE SELECTION
>90 Targets from Pool of Over 2,700 Purified mAbs
Multi-specific (e.g., DART)
Fc-Optimized Antibody
Drug-Conjugated Antibody
Whole Cell Immunization from Proprietary CSLC Lines
Antibody Screening and Lead Characterization
IHC (Tumor: Normal) In Vitro Bioassays Internalization Assay Antigen Identification
Selection of Optimal Antibody Format, Protein Engineering, and Incorporation of Half-life Extension Technologies

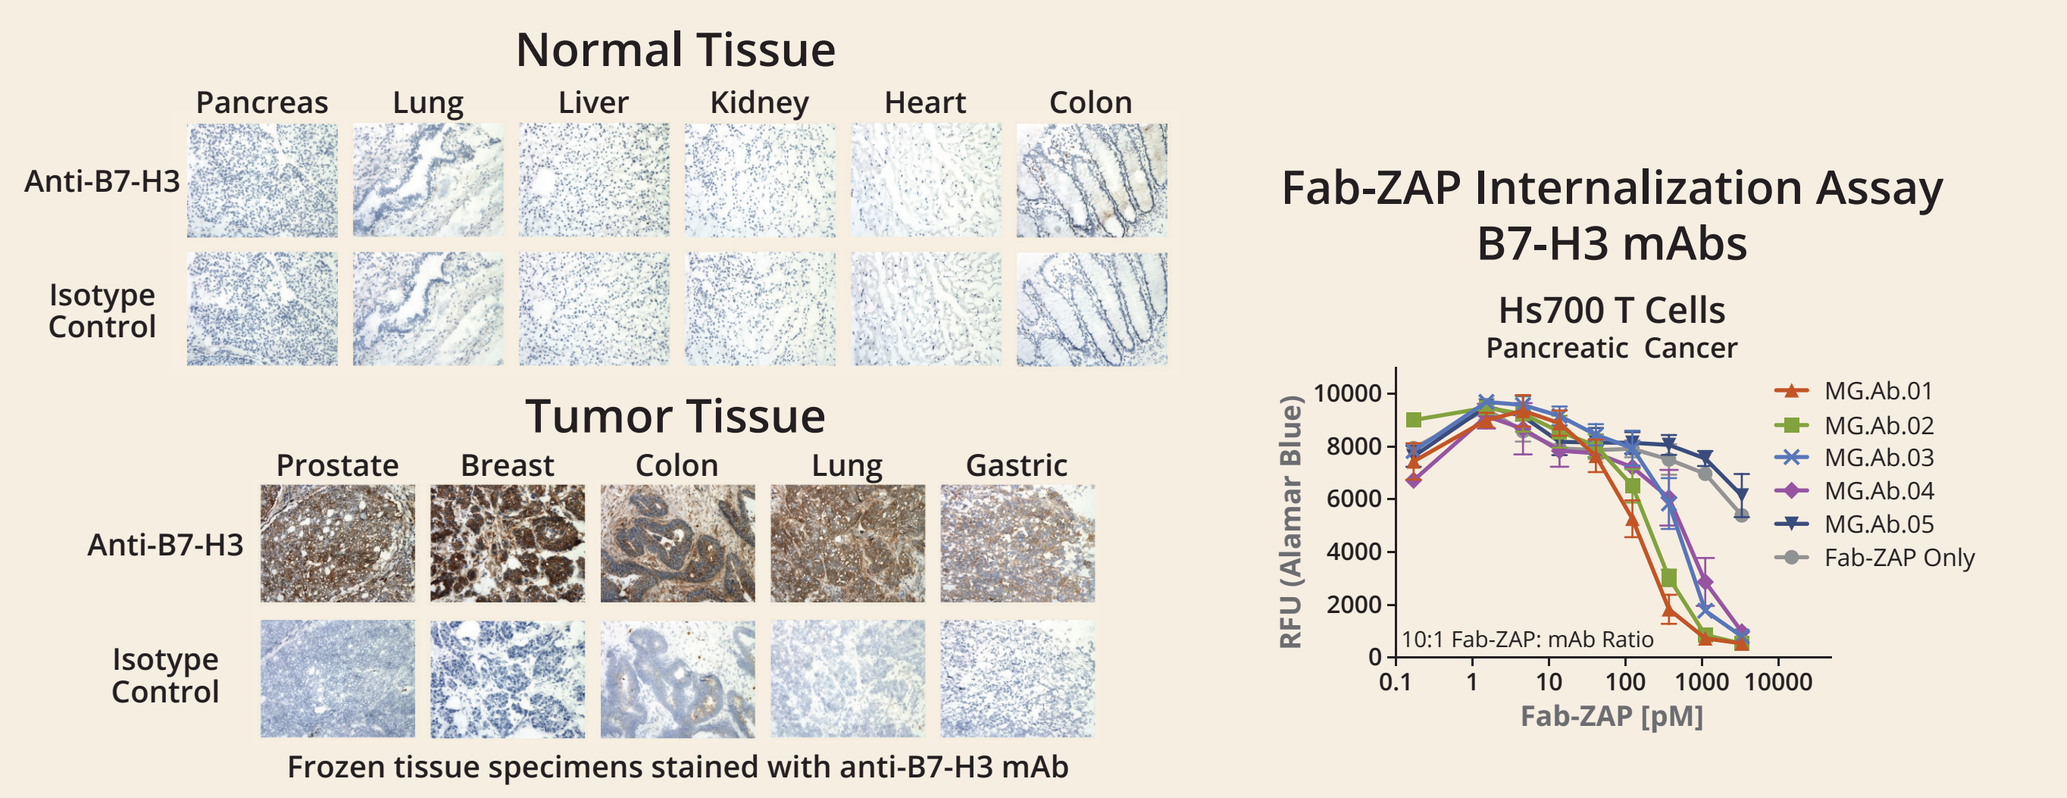
Normal Tissue
Pancreas
Lung
Liver
Kidney
Heart
Colon
Anti-B7-H3
Isotype Control
Tumor Tissue
Prostate
Breast
Colon
Lung
Gastric
Frozen tissue specimens stained with anti-B7-H3 mAb
Fab-ZAP Internalization Assay
B7-H3 mAbs
Hs700 T Cells
Pancreatic Cancer
RFU (Alamar Blue)
10:1 Fab-ZAP: mAb Ratio
Fab-ZAP [pM]
MG.Ab.01
MG.Ab.02
MG.Ab.03
MG.Ab.04
MG.Ab.05
Fab-ZAP Only

- Panel of tumor-specific B7-H3 mAbs identified
- Subset of B7-H3 mAbs that are efficiently internalized by tumor cells in vitro selected for further evaluation

Results

B7-H3 Is Highly Expressed on a Range of Solid Cancers

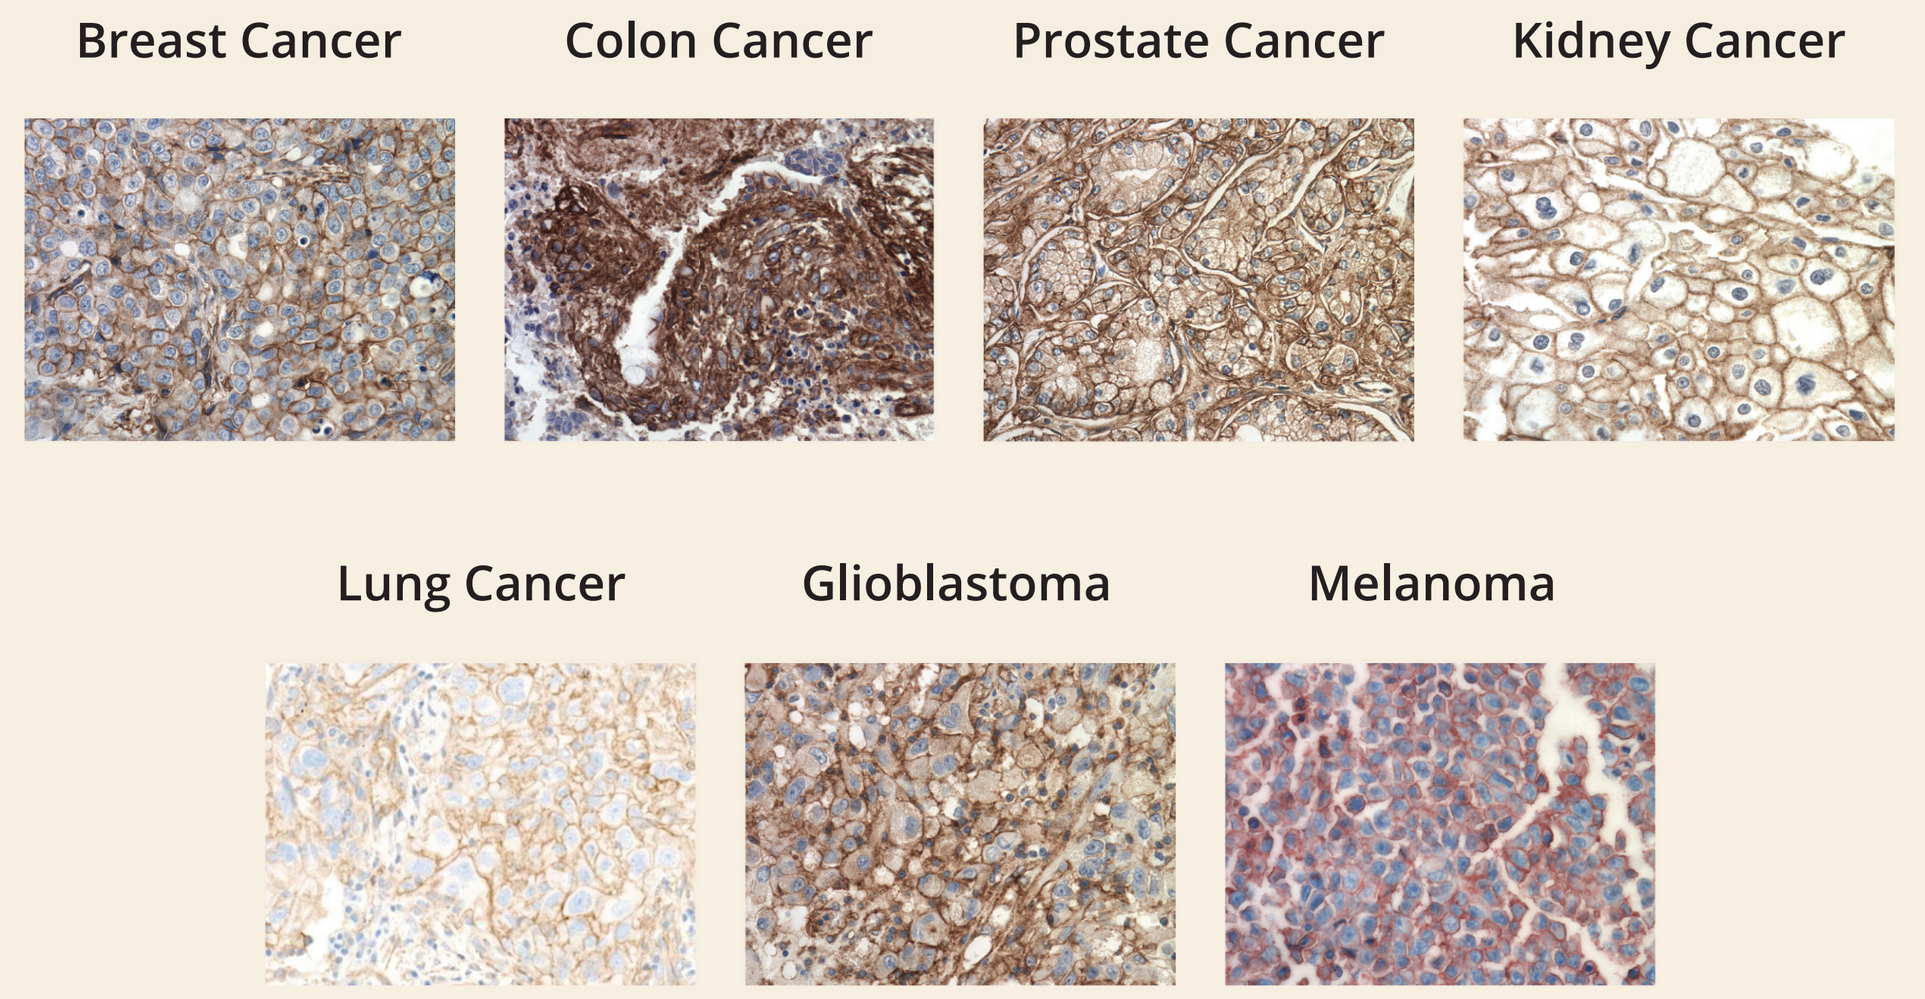
Breast Cancer
Colon Cancer
Prostate Cancer
Kidney Cancer
Lung Cancer
Glioblastoma
Melanoma

FFPE tumor specimens stained with R&D goat pAb

- Membrane expression of B7-H3 on tumor epithelium and tumor-associated vasculature

IHC Summary of Tumor Samples Screened

Cancer Type	B7-H3 Total Positive		B7-H3 2+ or Above	
Head and Neck	19/19	100%	19/19	100%
Kidney Cancer	77/78	99%	75/78	96%
Glioblastoma	65/66	98%	63/66	95%
Thyroid Cancer	34/35	97%	33/35	94%
Melanoma	66/70	94%	32/70	46%
Prostate Cancer	88/99	89%	51/99	52%
Pancreatic Cancer	69/78	88%	45/78	58%
Lung Cancer	226/272	83%	211/272	78%
Ovarian Cancer	59/79	75%	36/79	46%
Breast Cancer	119/164	73%	115/164	70%
Bladder Cancer	14/20	70%	9/20	45%

- B7-H3 is strongly expressed at a high frequency across a broad range of tumors

B7-H3 ADCs Exhibit Potent In Vitro Cytotoxicity

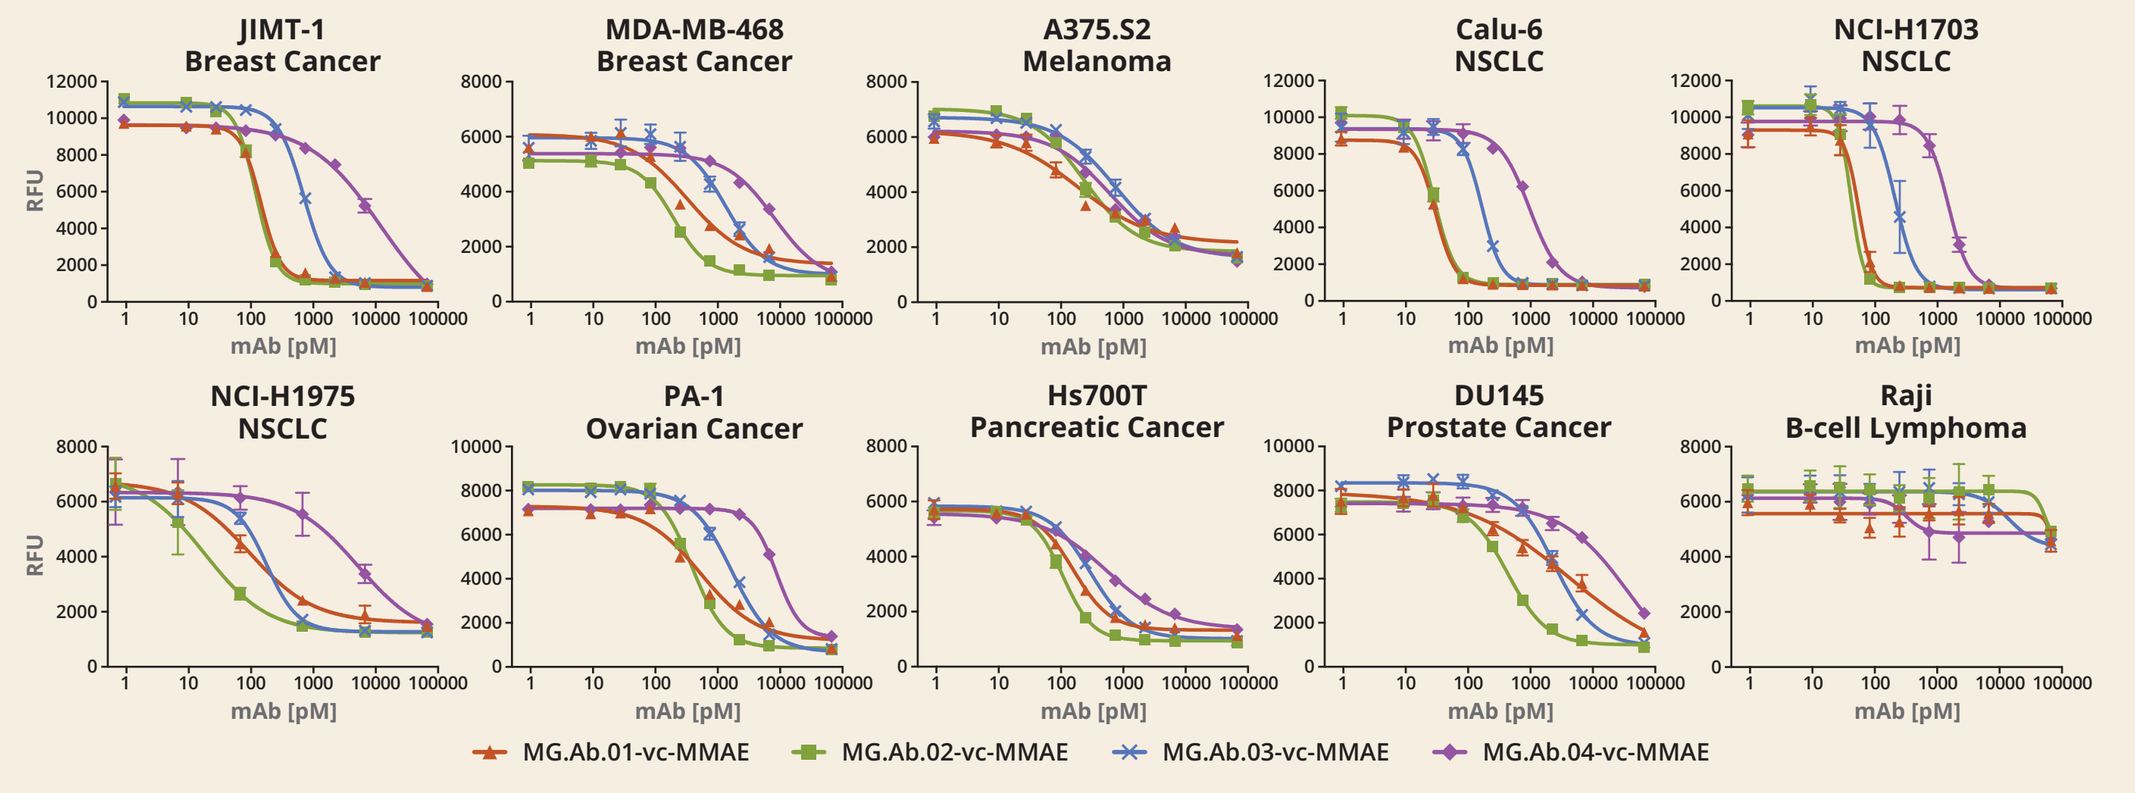
JIMT-1 Breast Cancer
MDA-MB-468 Breast Cancer
A375.S2 Melanoma
Calu-6 NSCLC
NCI-H1703 NSCLC
NCI-H1975 NSCLC
PA-1 Ovarian Cancer
Hs700T Pancreatic Cancer
DU145 Prostate Cancer
Raji B-cell Lymphoma
RFU
mAb [pM]
MG.Ab.01-vc-MMAE
MG.Ab.02-vc-MMAE
MG.Ab.03-vc-MMAE
MG.Ab.04-vc-MMAE

- Potency observed against a range of B7-H3-positive tumor lines

IC_{50} (pM)*	Breast Cancer		Melanoma	Non-Small Cell Lung Cancer			Ovarian Cancer	Pancreatic Cancer	Prostate Cancer
	JIMT-1	MDA-MB-468	A375.S2	Calu-6	NCI-H1703	NCI-H1975	PA-1	Hs700T	DU145
Antibody Binding Sites**	1.1e6	4.2e5	7.5e5	8.5e5	8.1e5	4.8e5	6.1e5	2.1e6	2.4e5
MG.Ab.01-vc-MMAE	221	352	153	59	90	31	555	159	3770
MG.Ab.02-vc-MMAE	124	201	267	30	43	16	409	109	465
MG.Ab.03-vc-MMAE	735	1383	887	171	219	162	1795	303	2587
MG.Ab.04-vc-MMAE	9100	8095	703	995	1517	26976	8326	607	20153

*Alamar Blue cytotoxicity assay. **Antibody Binding Sites determined by Bangs QFACS Kit.

In Vivo Activity: MDA-MB-468 Breast Cancer

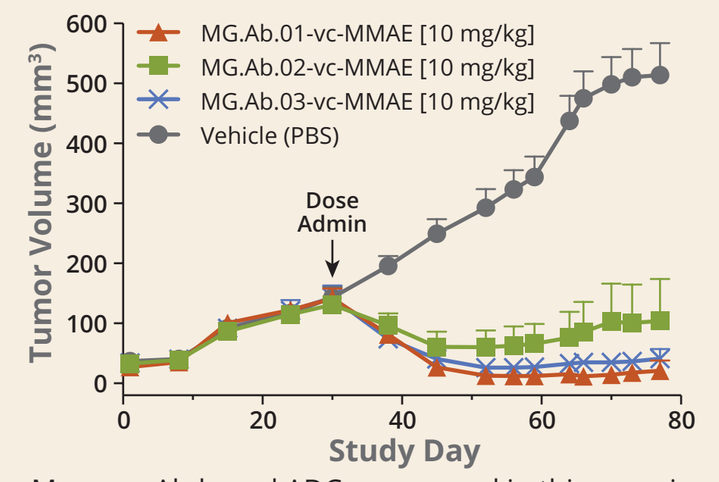
Tumor Volume (mm³)
MG.Ab.01-vc-MMAE [10 mg/kg]
MG.Ab.02-vc-MMAE [10 mg/kg]
MG.Ab.03-vc-MMAE [10 mg/kg]
Vehicle (PBS)
Dose Admin
Study Day

Mouse mAb-based ADCs were used in this experiment.

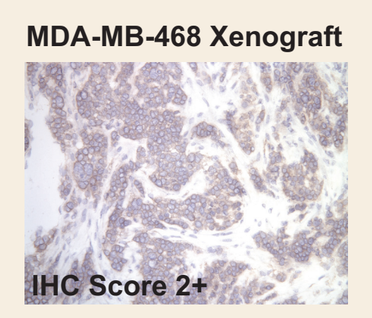
MDA-MB-468 Xenograft
IHC Score 2+

Treatment	IP Dose (mg/kg)	Schedule (Dosed Day 30)	T/C (%)	CR
MG.Ab.01-vc-MMAE	10	qdx1	4	6/7
MG.Ab.02-vc-MMAE	10	qdx1	20	4/7
MG.Ab.03-vc-MMAE	10	qdx1	8	1/7

T/C (%) = Percentage of tumor size relative to vehicle
CR = Tumor volume ≤5 mm^3 during study

In Vivo Activity: NCI-H1703 Non-Small Cell Lung Cancer

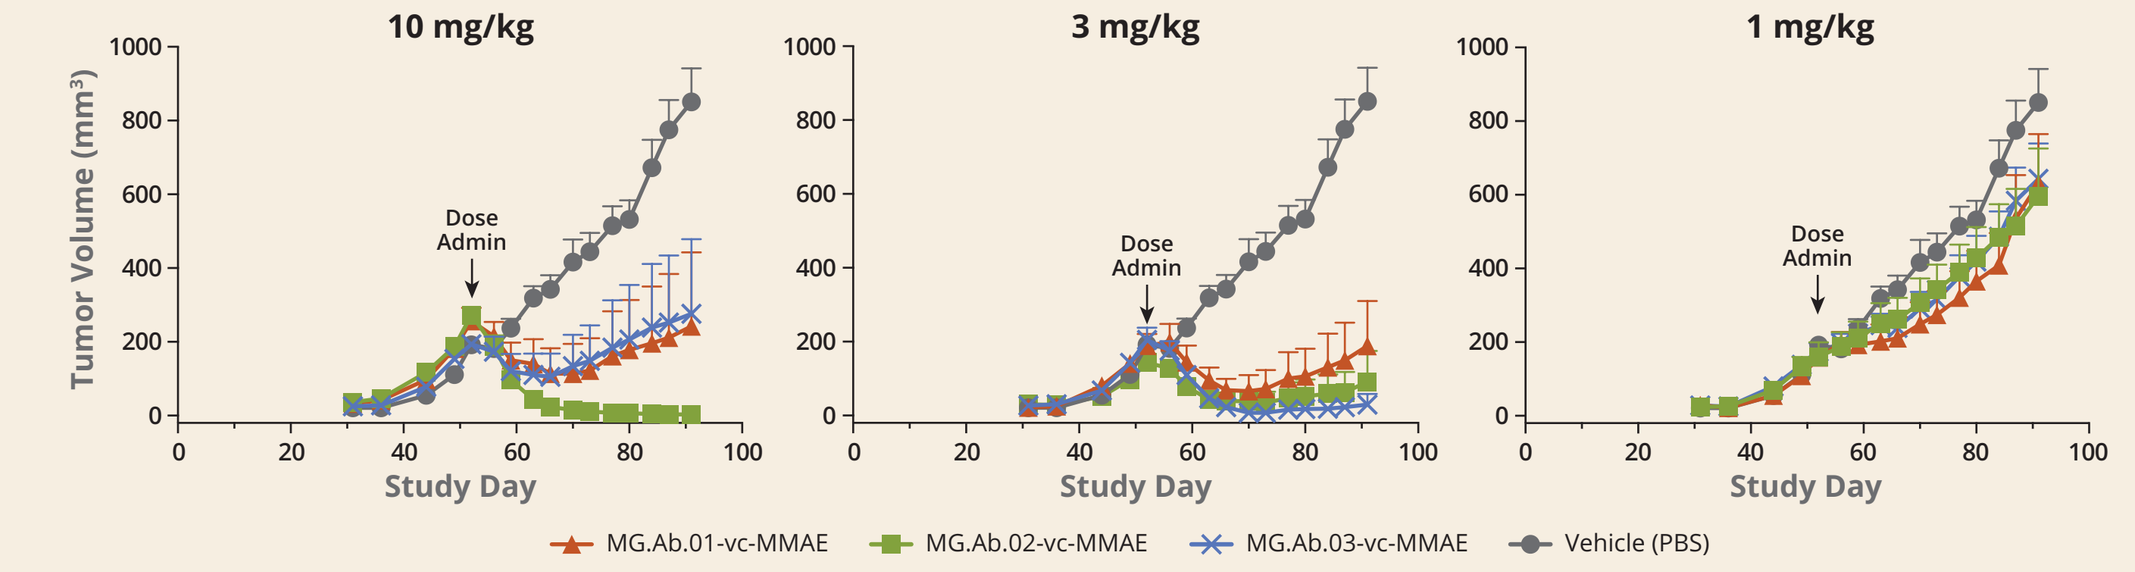
10 mg/kg
3 mg/kg
1 mg/kg
Tumor Volume (mm³)
Dose Admin
Study Day
MG.Ab.01-vc-MMAE
MG.Ab.02-vc-MMAE
MG.Ab.03-vc-MMAE
Vehicle (PBS)

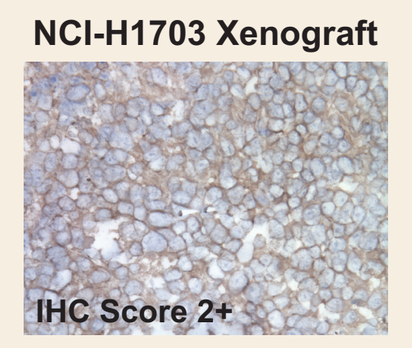
NCI-H1703 Xenograft
IHC Score 2+

Treatment	IP Dose (mg/kg)	Schedule (Dosed Day 52)	T/C (%)	CR
MG.Ab.01-vc-MMAE	10	qdx1	28	5/7
MG.Ab.01-vc-MMAE	3	qdx1	22	3/7
MG.Ab.01-vc-MMAE	1	qdx1	74	0/7
MG.Ab.02-vc-MMAE	10	qdx1	0	6/7
MG.Ab.02-vc-MMAE	3	qdx1	11	5/7
MG.Ab.02-vc-MMAE	1	qdx1	70	0/7
MG.Ab.03-vc-MMAE	10	qdx1	32	5/7
MG.Ab.03-vc-MMAE	3	qdx1	4	6/7
MG.Ab.03-vc-MMAE	1	qdx1	76	0/7

T/C (%) = Percentage of tumor size relative to vehicle
CR = Tumor volume ≤5 mm^3 during study

In Vivo Activity: PA-1 Ovarian Cancer

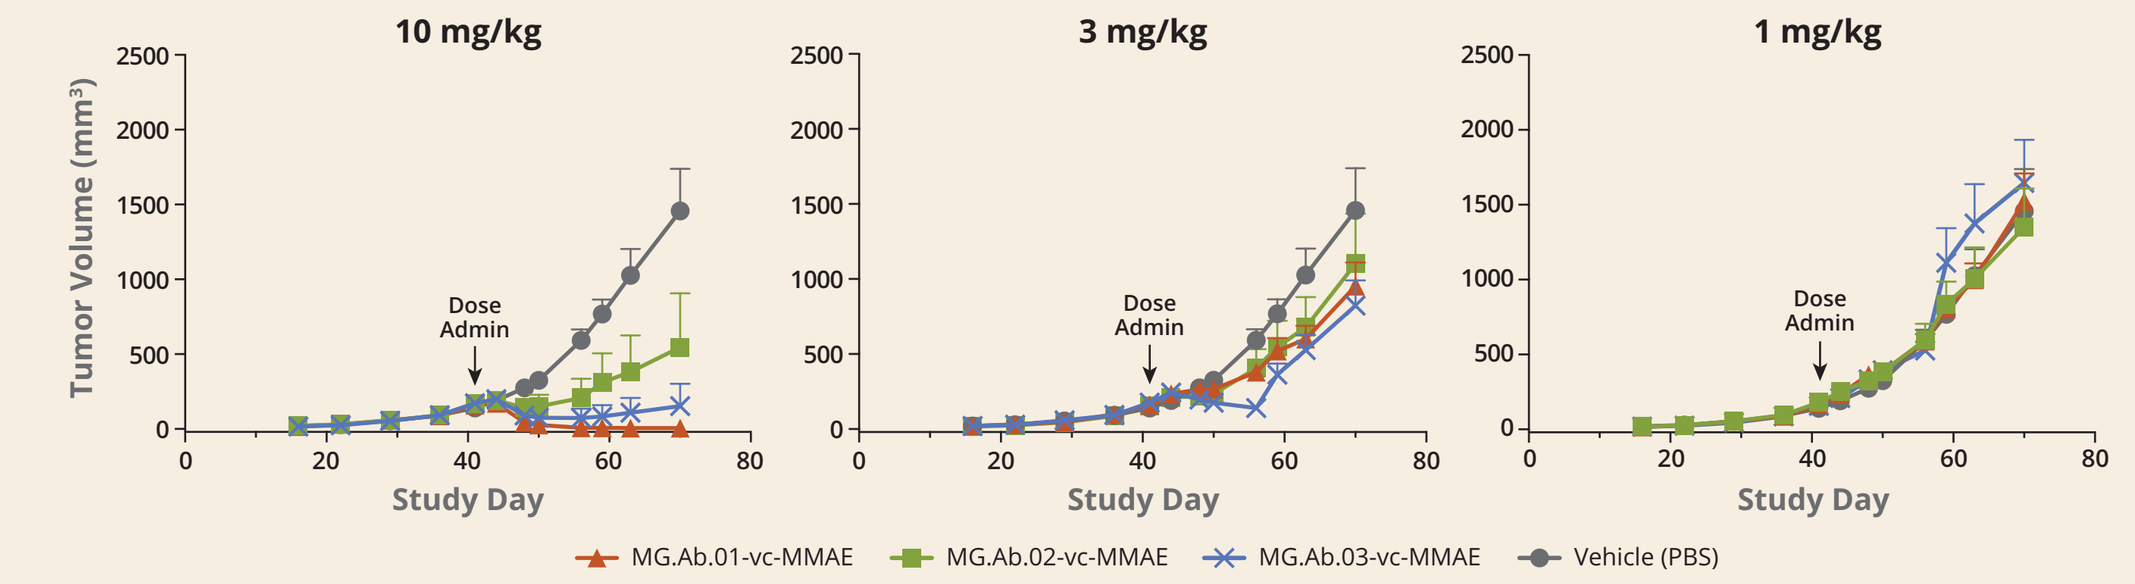
10 mg/kg
3 mg/kg
1 mg/kg
Tumor Volume (mm³)
Dose Admin
Study Day
MG.Ab.01-vc-MMAE
MG.Ab.02-vc-MMAE
MG.Ab.03-vc-MMAE
Vehicle (PBS)

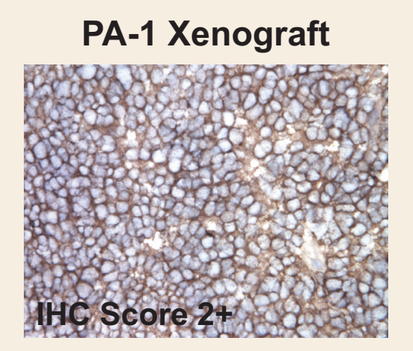
PA-1 Xenograft
IHC Score 2+

Treatment	IP Dose (mg/kg)	Schedule (Dosed Day 42)	T/C (%)	CR
MG.Ab.01-vc-MMAE	10	qdx1	0	6/7
MG.Ab.01-vc-MMAE	3	qdx1	65	0/7
MG.Ab.01-vc-MMAE	1	qdx1	105	0/7
MG.Ab.02-vc-MMAE	10	qdx1	37	3/7
MG.Ab.02-vc-MMAE	3	qdx1	76	1/7
MG.Ab.02-vc-MMAE	1	qdx1	93	0/7
MG.Ab.03-vc-MMAE	10	qdx1	11	5/7
MG.Ab.03-vc-MMAE	3	qdx1	57	1/7
MG.Ab.03-vc-MMAE	1	qdx1	113	0/7

T/C (%) = Percentage of tumor size relative to vehicle
CR = Tumor volume ≤5 mm^3 during study

In Vivo Activity: A375.S2 Melanoma

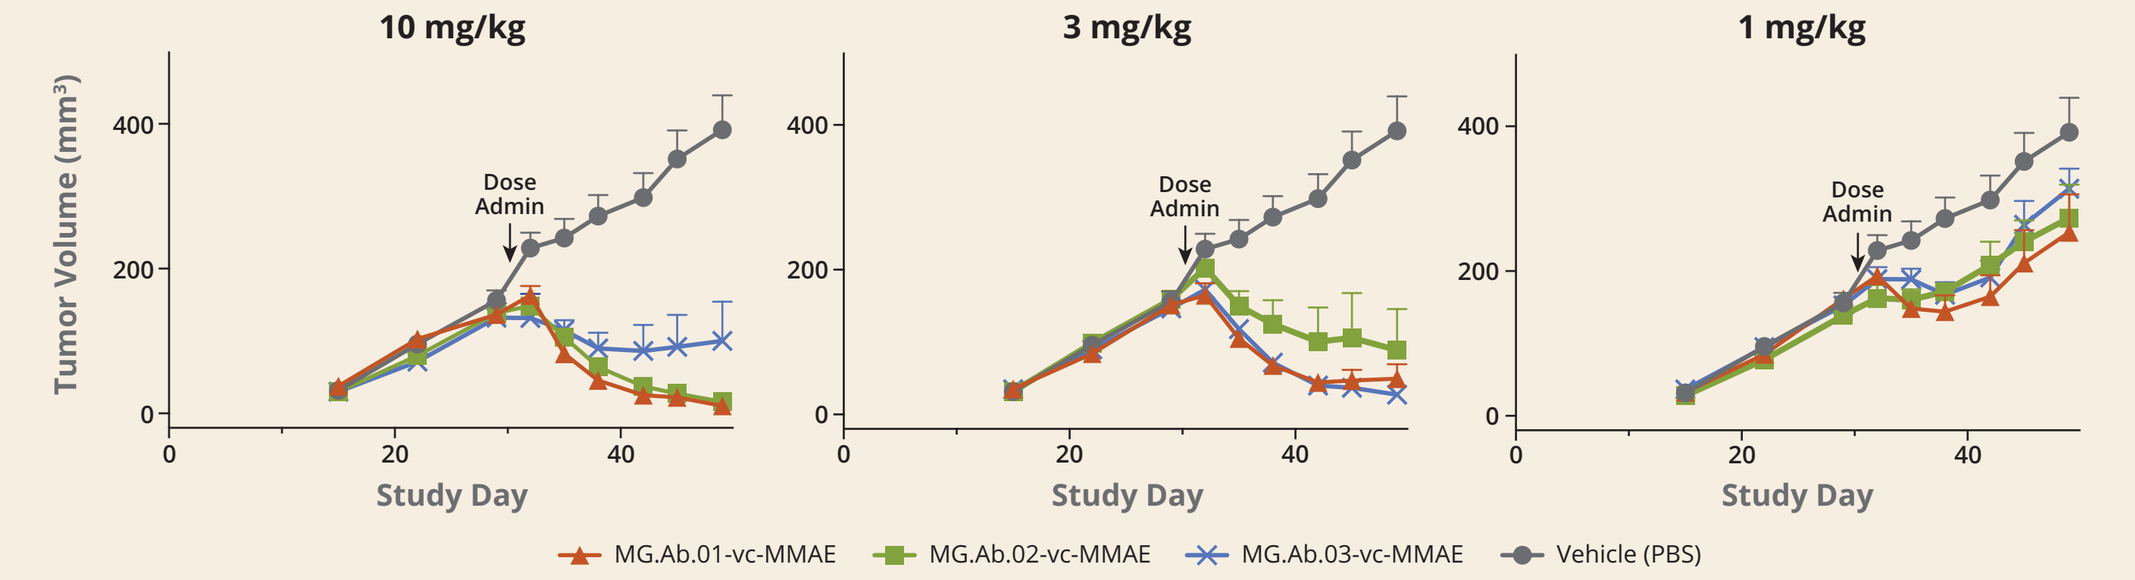
10 mg/kg
3 mg/kg
1 mg/kg
Tumor Volume (mm³)
Dose Admin
Study Day
MG.Ab.01-vc-MMAE
MG.Ab.02-vc-MMAE
MG.Ab.03-vc-MMAE
Vehicle (PBS)

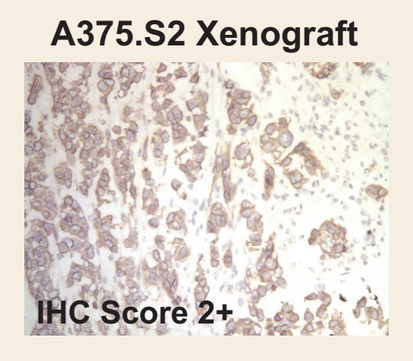
A375.S2 Xenograft
IHC Score 2+

Treatment	IP Dose (mg/kg)	Schedule (Dosed Day 30)	T/C (%)	CR
MG.Ab.01-vc-MMAE	10	qdx1	3	5/7
MG.Ab.01-vc-MMAE	3	qdx1	13	1/7
MG.Ab.01-vc-MMAE	1	qdx1	65	0/7
MG.Ab.02-vc-MMAE	10	qdx1	4	1/7
MG.Ab.02-vc-MMAE	3	qdx1	23	0/7
MG.Ab.02-vc-MMAE	1	qdx1	70	0/7
MG.Ab.03-vc-MMAE	10	qdx1	26	2/7
MG.Ab.03-vc-MMAE	3	qdx1	7	0/7
MG.Ab.03-vc-MMAE	1	qdx1	80	0/7

T/C (%) = Percentage of tumor size relative to vehicle
CR = Tumor volume ≤5 mm^3 during study

Pharmacokinetics of B7-H3 ADCs

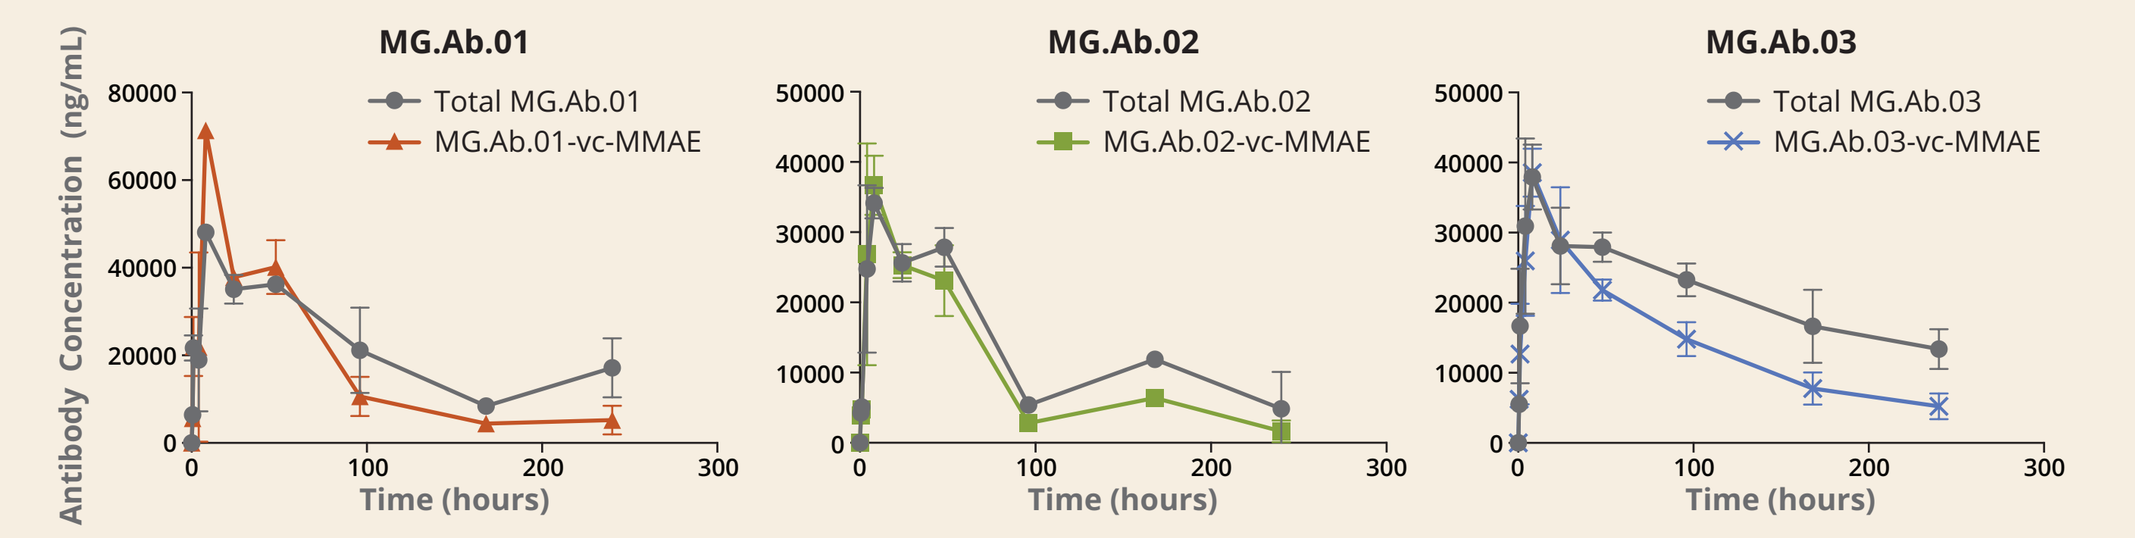
Antibody Concentration (ng/mL)
MG.Ab.01
Total MG.Ab.01
MG.Ab.01-vc-MMAE
MG.Ab.02
Total MG.Ab.02
MG.Ab.02-vc-MMAE
MG.Ab.03
Total MG.Ab.03
MG.Ab.03-vc-MMAE
Time (hours)

Antibody	Total Antibody $t_{1/2}$ (h)	Total Antibody AUC (h*µg/mL)	Intact ADC $t_{1/2}$ (h)	Intact ADC AUC (h*µg/mL)
MG.Ab.01-vc-MMAE	114.1	4,796	58.9	4,033
MG.Ab.02-vc-MMAE	75.9	2,699	52.6	2,202
MG.Ab.03-vc-MMAE	177.2	5,162	87.3	3,502

- Auristatin E-conjugated B7-H3 ADCs are highly stable in mice

Conclusions

- B7-H3 is overexpressed in a wide range of solid cancers, including breast, lung, pancreatic, head and neck, prostate, kidney, and colon cancer, as well as in glioblastoma and melanoma
- Anti-B7-H3 auristatin E ADCs:
 - Exhibited specific, dose-dependent cytotoxicity toward a wide range of B7-H3-positive tumor cell lines in vitro
 - Demonstrated significant antitumor activity toward B7-H3-positive mouse tumor xenograft models of breast, lung, and ovarian cancers, as well as melanoma
 - Showed typical pharmacokinetics in mice, with a half-life of ~2.2-3.6 days for the intact ADC

ADCs targeting B7-H3 may serve as potential therapeutics for the treatment of B7-H3-expressing malignancies

References

1. Loo D, et al. Clin Cancer Res. 2012 Jul 15;18(14):3834-45. **2.** Loo D, et al. Mol Cancer Ther. 2007 Mar;6(3):856-65.

Acknowledgements

ADCs were provided by Concortis Biosystems, San Diego, CA

Presented at the 2016 American Association for Cancer Research Annual Meeting, April 16–20, 2016, New Orleans, Louisiana

©2016 MacroGenics, Inc. All rights reserved.