Margetuximab Mediates Greater Fc-dependent Anti-tumor Activities than Trastuzumab or Pertuzumab In Vitro

1538

Liqin Liu[1], Yinhua Yang[1], Robert Burns[1], Jonathan Li[2], Haiquan Li[1], Sergey Gorlatov[1], Paul Moore[1], Jeffrey Nordstrom[1]

MacroGenics, Inc., Rockville, MD and Brisbane, CA

http://ir.macrogenics.com/events.cfm

Abstract

Introduction: Margetuximab (M) is an investigational Fc-engineered anti-HER2 monoclonal antibody (mAb) with potential for greater immune-mediated anti-tumor activity than trastuzumab (T). M and T bind the same subdomain IV epitope, but M binds with higher affinity to activating Fc receptor, CD16A (FcγRIIIA), and lower affinity to inhibitory Fc receptor, CD32B (FcγRIIB). Pertuzumab (P) binds to a subdomain II epitope and has the same wild type Fc domain as T.

Purpose: We compared in vitro properties of M, T and P and combinations of M+P and T+P to evaluate Fc domain contributions to anti-HER2 mAb mediated activities.

HER2 binding: By surface plasmon resonance, M, T and P bound to the HER2 extracellular domain with high affinity. Binding affinities of M and T were unchanged if P was pre-bound, indicating lack of interaction. M, T and P exhibited comparable binding to HER2-expressing cell lines.

Inhibition of HER2 ECD shedding: M and T mediated comparable inhibition of HER2 ECD shedding, whereas P was ineffective.

Inhibition of proliferation: Anti-proliferative activities of M and T toward $HER2^{3+}$ cells (N87, SKBR3 or BT474) in the absence or presence of ligands (EGF or HRG1β) were similar, whereas P exhibited weaker activity. In the absence of ligands, M+P or T+P were comparably as active as M or T alone. In presence of ligands, M+P and T+P were generally more active than M or T alone.

ADCC: Antibody-dependent cell-mediated cytotoxicity (ADCC) was evaluated with N87 and SKBR3 target cells ($HER2^{3+}$, expressing a reporter gene to measure viability) and NK effector cells from donors with differing $CD16A^{158}$ genotypes. M was 7- to 84-fold more potent than T, as well as 69- to 744-fold more potent than P. Greater differences generally were seen with effector NK cells bearing the more common $CD16A^{158}$ FF and VF genotypes than with the less common, high affinity $CD16A^{158}$ VV genotype. While addition of P enhanced mean potency of T by 2- to 3-fold, no such effect was seen with M; however, M or M+P was 7- to 25-fold more potent than T+P. Results with JIMT-1 cells ($HER2^{2+}$) were similar, albeit with lower differential than with $HER2^{3+}$ target cells.

NK cell activation: NK cells were monitored by flow cytometry following incubation of peripheral blood mononuclear cells with $HER2^{+}$ target cells and mAbs. M induced greater expression of markers of activation (CD137), cytolytic capability (granzyme B, perforin) and proliferation (Ki67) than T. An anti-HER2 mAb with an inactivated Fc domain was ineffective, indicating that induced changes in NK cells were Fc-dependent.

Conclusion: M and T bind HER2 with high affinity and exhibit anti-proliferative activity that is enhanced by addition of P if ligands (EGF or HRG1β) are present. Relative to T, M mediates superior ADCC with effector cells of all CD16A genotypes and promotes greater NK cell activation and expansion. The M+P combination maintains superior ADCC compared to the T+P combination.

Binding Properties

Margetuximab Has Increased Affinity for Both Allotypes of CD16A (Activating FcγR) and Decreased Affinity for CD32B (Inhibitory FcγR)

Antigen	Antibody	k_a (1/M•s)	k_d (1/s)	K_D (nM)	M/T Affinity Ratio ($1/K_D$)
CD16A 158V[1]	M T	4.7×10^5 4.3×10^5	27×10^{-3} 105×10^{-3}	57 244	4.3
CD16A 158F[1]	M T	5.8×10^5 2.8×10^5	61×10^{-3} 155×10^{-3}	105 554	5.3
CD32B[2]	M T	ND ND	ND ND	87 19	0.2

k_a = association rate constant, k_d = dissociation rate constant, K_D = equilibrium dissociation constant, M = molar, s = second, ND = not determined
[1]Binding of monomeric of human CD16A ECD to antibody captured on immobilized HER2-His
[2]Binding of dimeric human CD32B ECD/aglycosyl Fc fusion to antibody captured on immobilized HER2-His

Comparable Binding to HER2 by Margetuximab (M) and Trastuzumab (T)

- Margetuximab or trastuzumab binding to HER2 is unaffected by prior pertuzumab (P) binding

Antigen	Antibody	k_a (1/M•s)	k_d (1/s)	K_D (nM)
HER2[1]	M	3.2×10^5	10.0×10^{-4}	3.1
	M (+ P)[2]	3.5×10^5	10.0×10^{-4}	2.9
	T	3.7×10^5	9.5×10^{-4}	2.6
	T (+ P)[2]	4.2×10^5	11.0×10^{-4}	2.6
	P	2.5×10^5	1.6×10^{-4}	0.6

k_a = association rate constant, k_d = dissociation rate constant, K_D = equilibrium dissociation constant, M = molar, s = second
[1]Binding of antibodies to human HER2 (ECD)-His protein captured on anti-penta-His surface
[2]Binding of M or T measured after P was preloaded

Comparable Binding to SKBR-3 ($HER2^{3+}$) Cells by Margetuximab (M), Trastuzumab (T) and Pertuzumab (P)

- Margetuximab or trastuzumab binding to $HER2^{+}$ cells is unaffected by pertuzumab

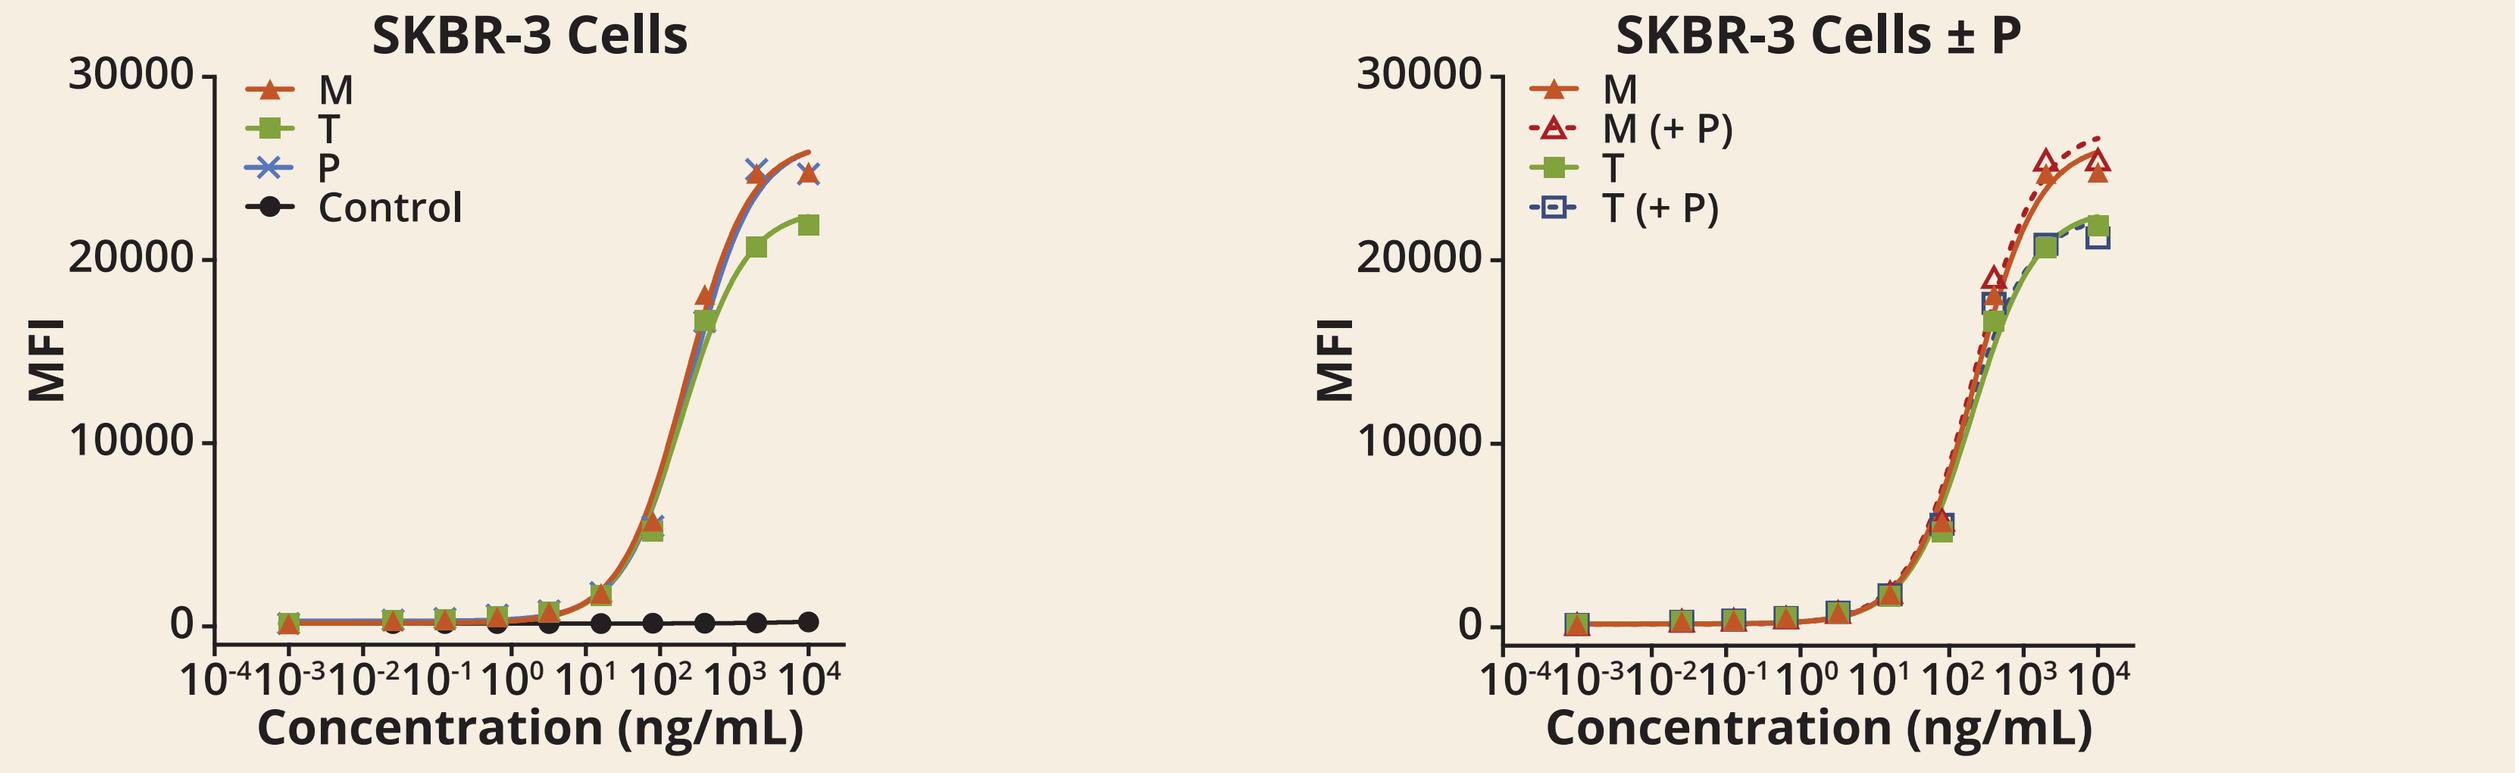

SKBR-3 Cells
M
T
P
Control
MFI
Concentration (ng/mL)
SKBR-3 Cells ± P
M
M (+ P)
T
T (+ P)

Binding of Alexa-Fluor-488-labeled M, T or P.

Binding of Alexa-Fluor-488-labeled M or T in the absence or presence of unlabeled P.

Fc-independent Activities

Comparable Inhibition of HER2 Extracellular Domain (ECD) Shedding by Margetuximab and Trastuzumab

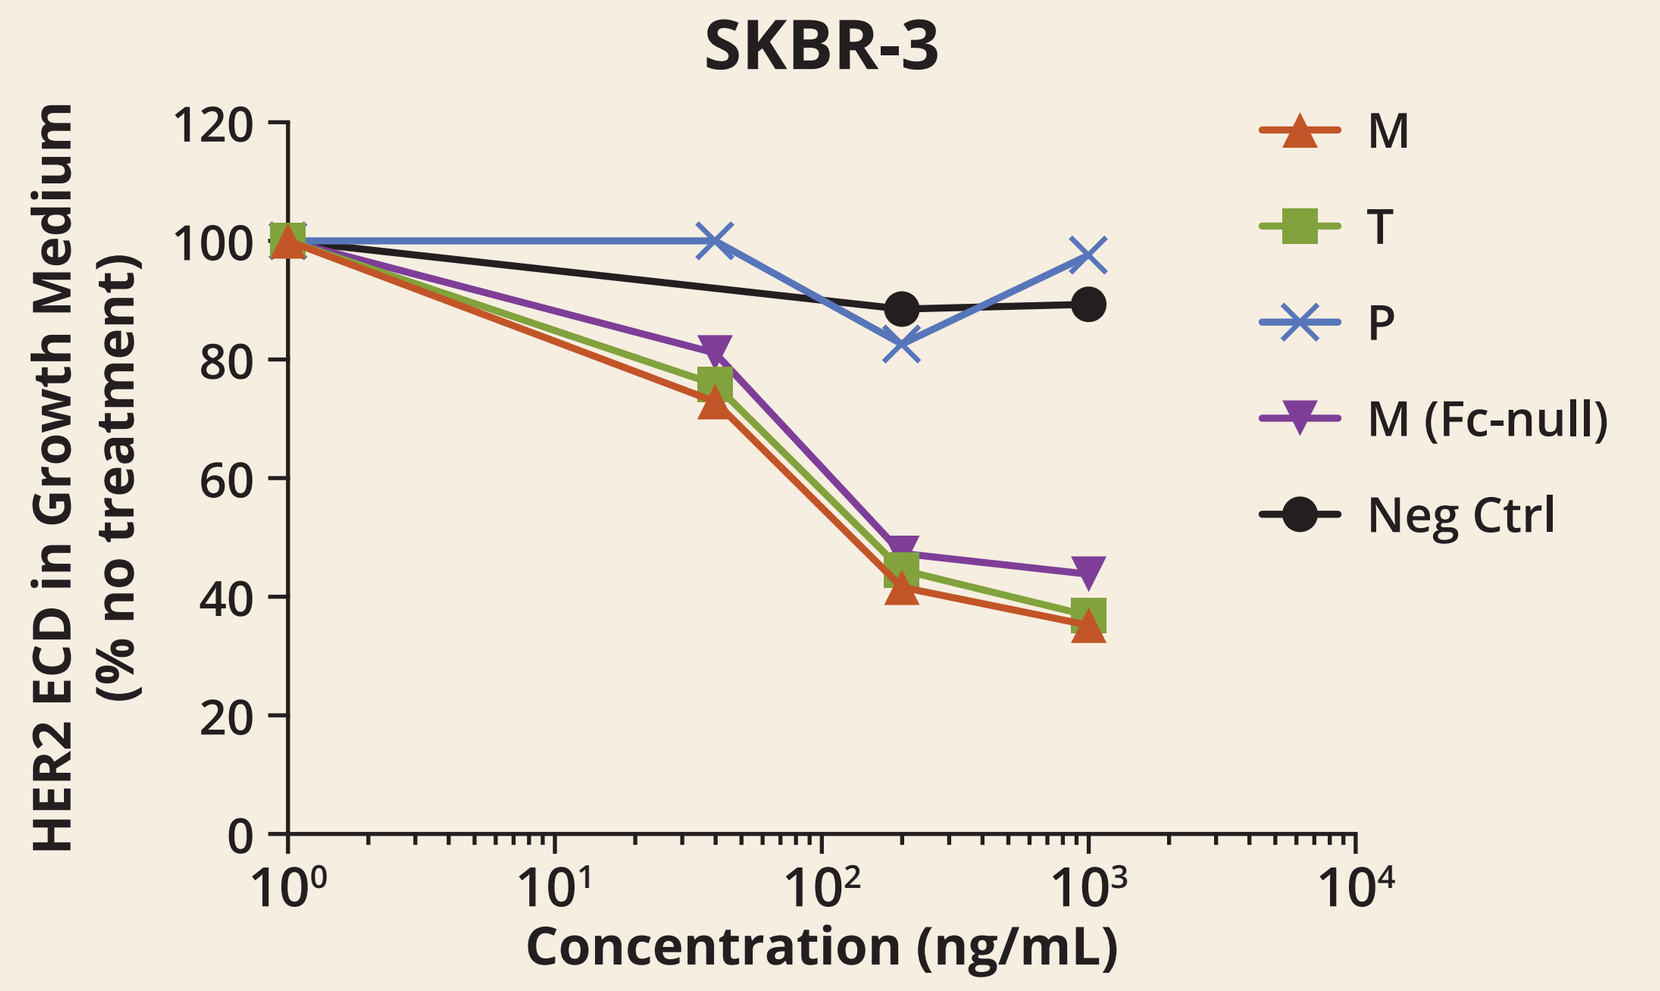

SKBR-3
HER2 ECD in Growth Medium (% no treatment)
Concentration (ng/mL)
M
T
P
M (Fc-null)
Neg Ctrl

- M, T and Fc-null (aglycosylated) variant of M block HER2 ECD shedding
- Pertuzumab (P) does not block HER2 ECD shedding

Anti-proliferative Activities of Margetuximab and Trastuzumab Are Improved by Combination with Pertuzumab

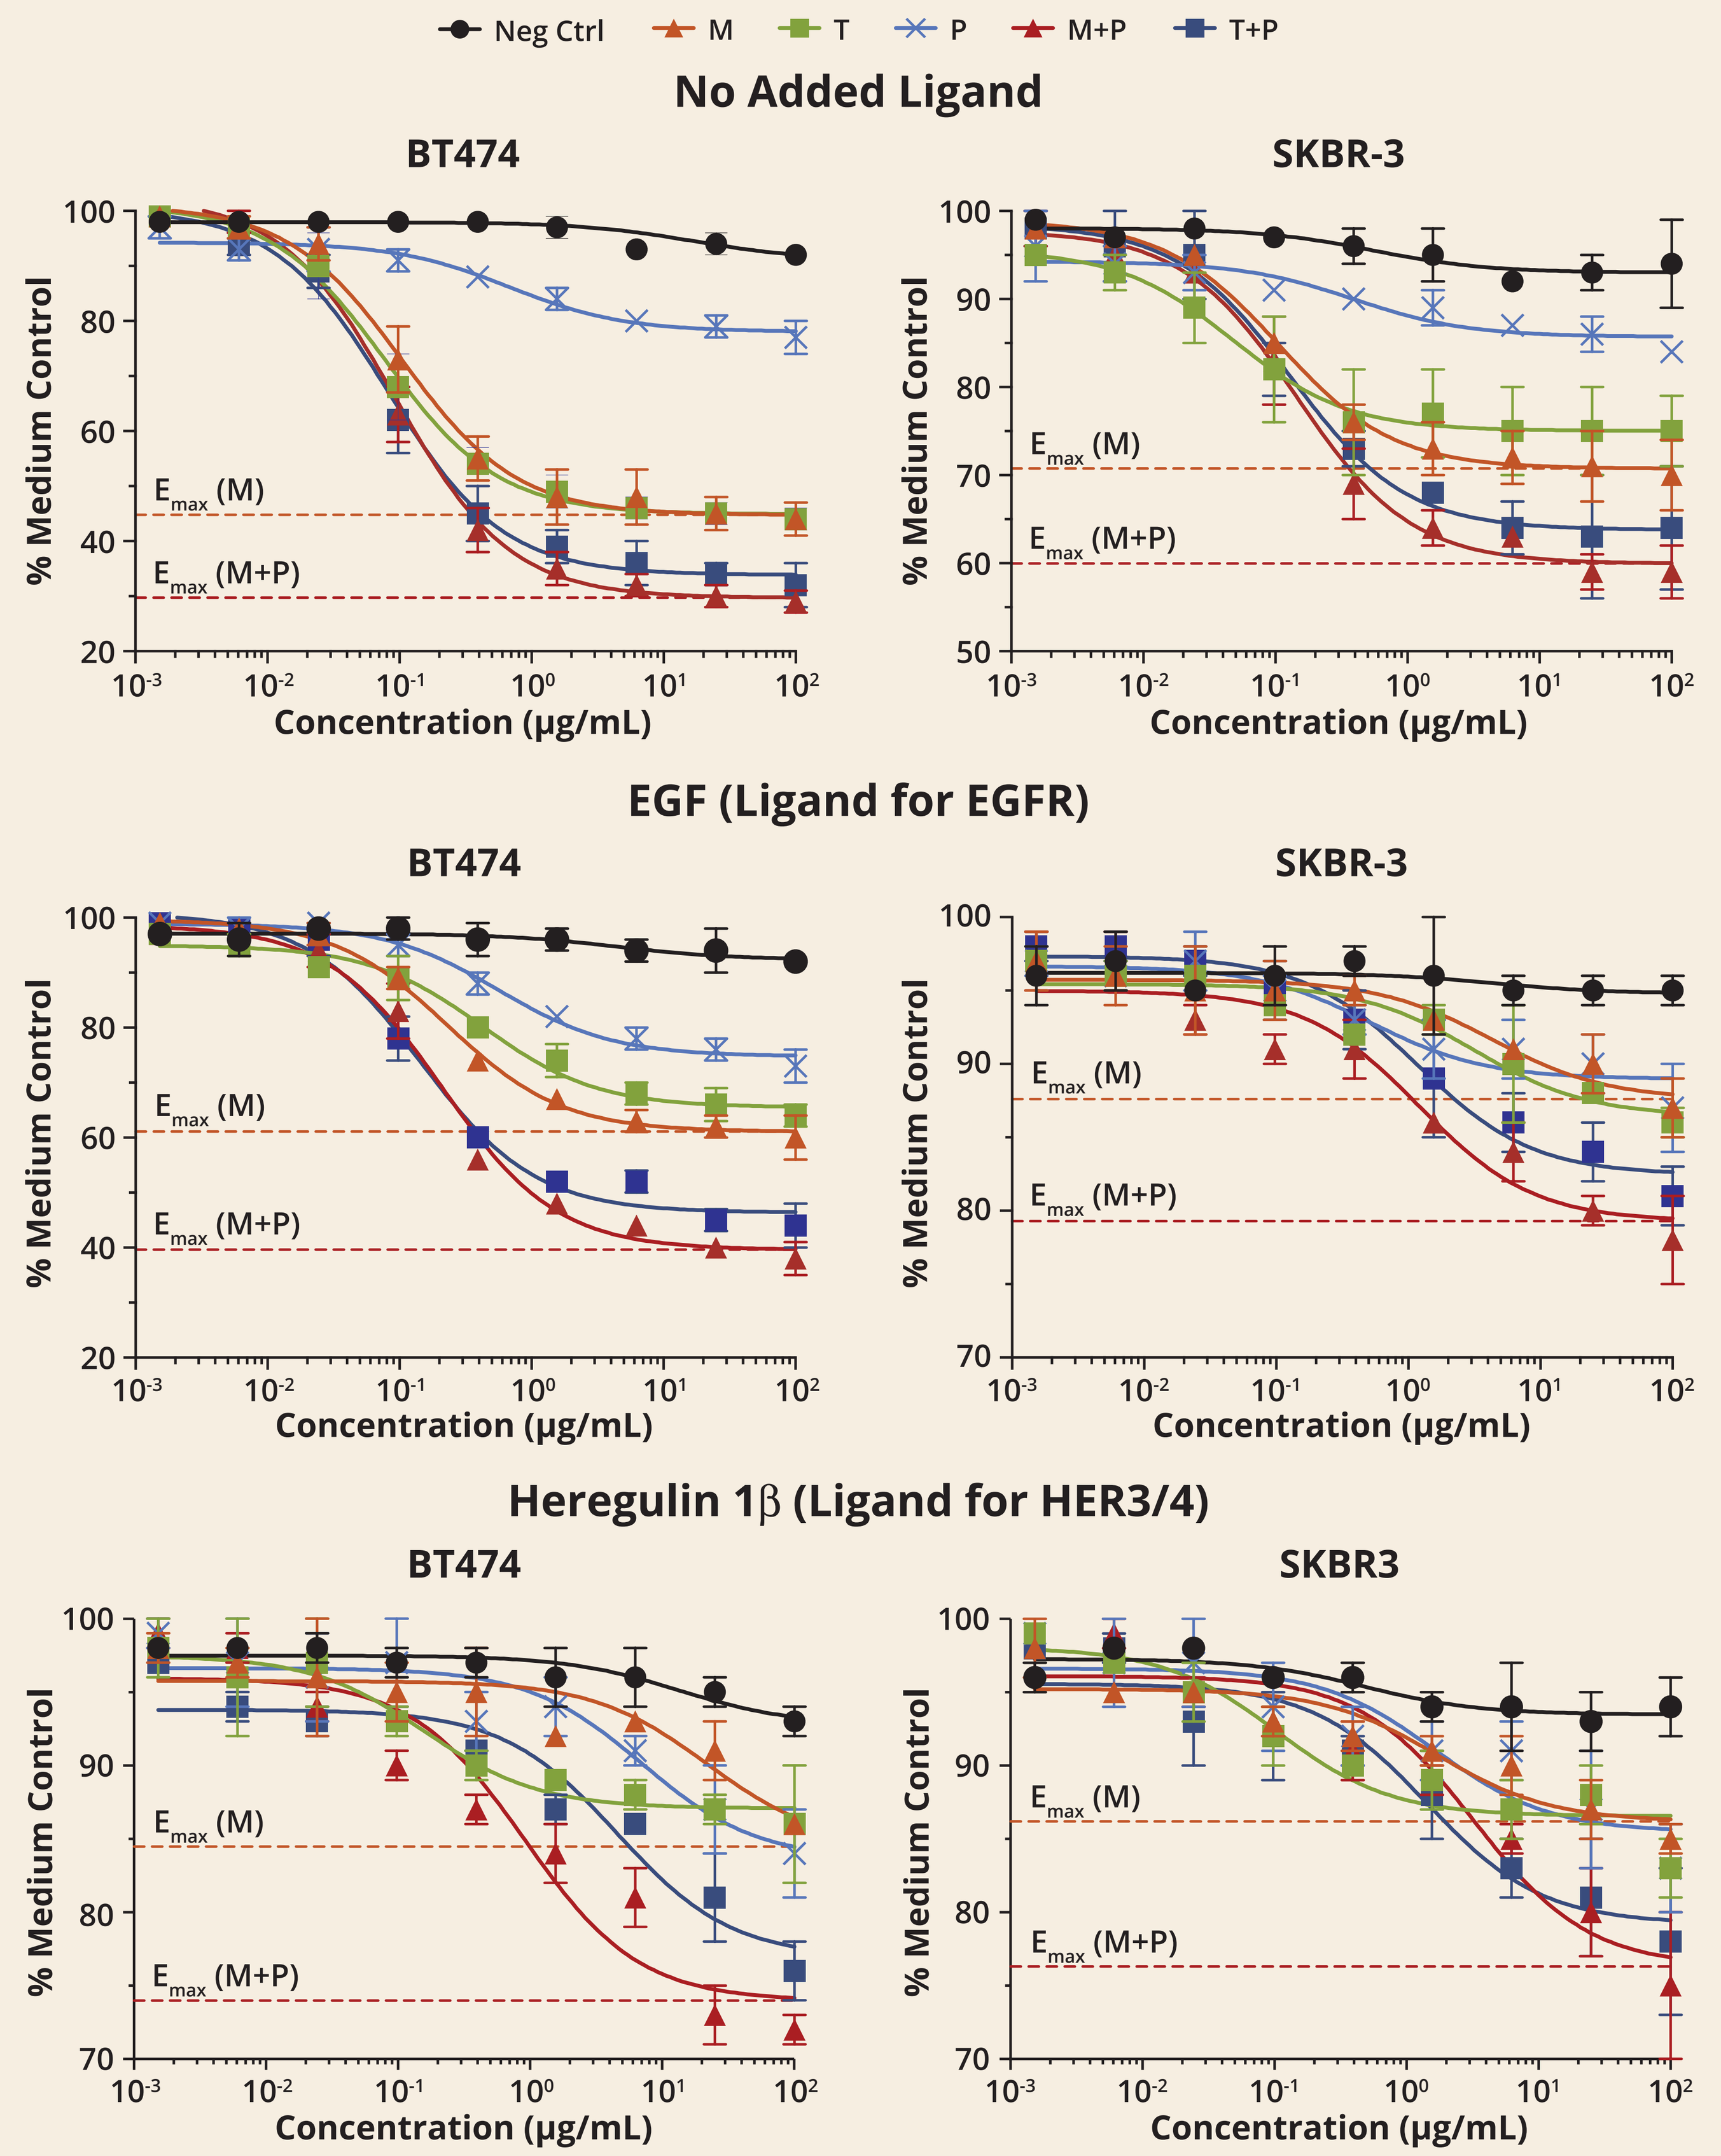

Neg Ctrl
M
T
P
M+P
T+P
No Added Ligand
BT474
SKBR-3
EGF (Ligand for EGFR)
BT474
SKBR-3
Heregulin 1β (Ligand for HER3/4)
BT474
SKBR3
% Medium Control
Emax (M)
Emax (M+P)
Concentration (µg/mL)

Dashed lines indicate the maximal level of inhibition of proliferation (E_{max}) by M alone or M+P combined.

Fc-dependent Activities

Margetuximab (M) Mediates ADCC with Greater Potency than Trastuzumab (T) or Pertuzumab (P)

ADCC of SKBR-3/GF Cells by NK Cells of Different $CD16A^{158}$ Genotypes (E:T = 2:1)

- SKBR-3/GF cells constitutively express luciferase, which allows specific measurement of target cell viability

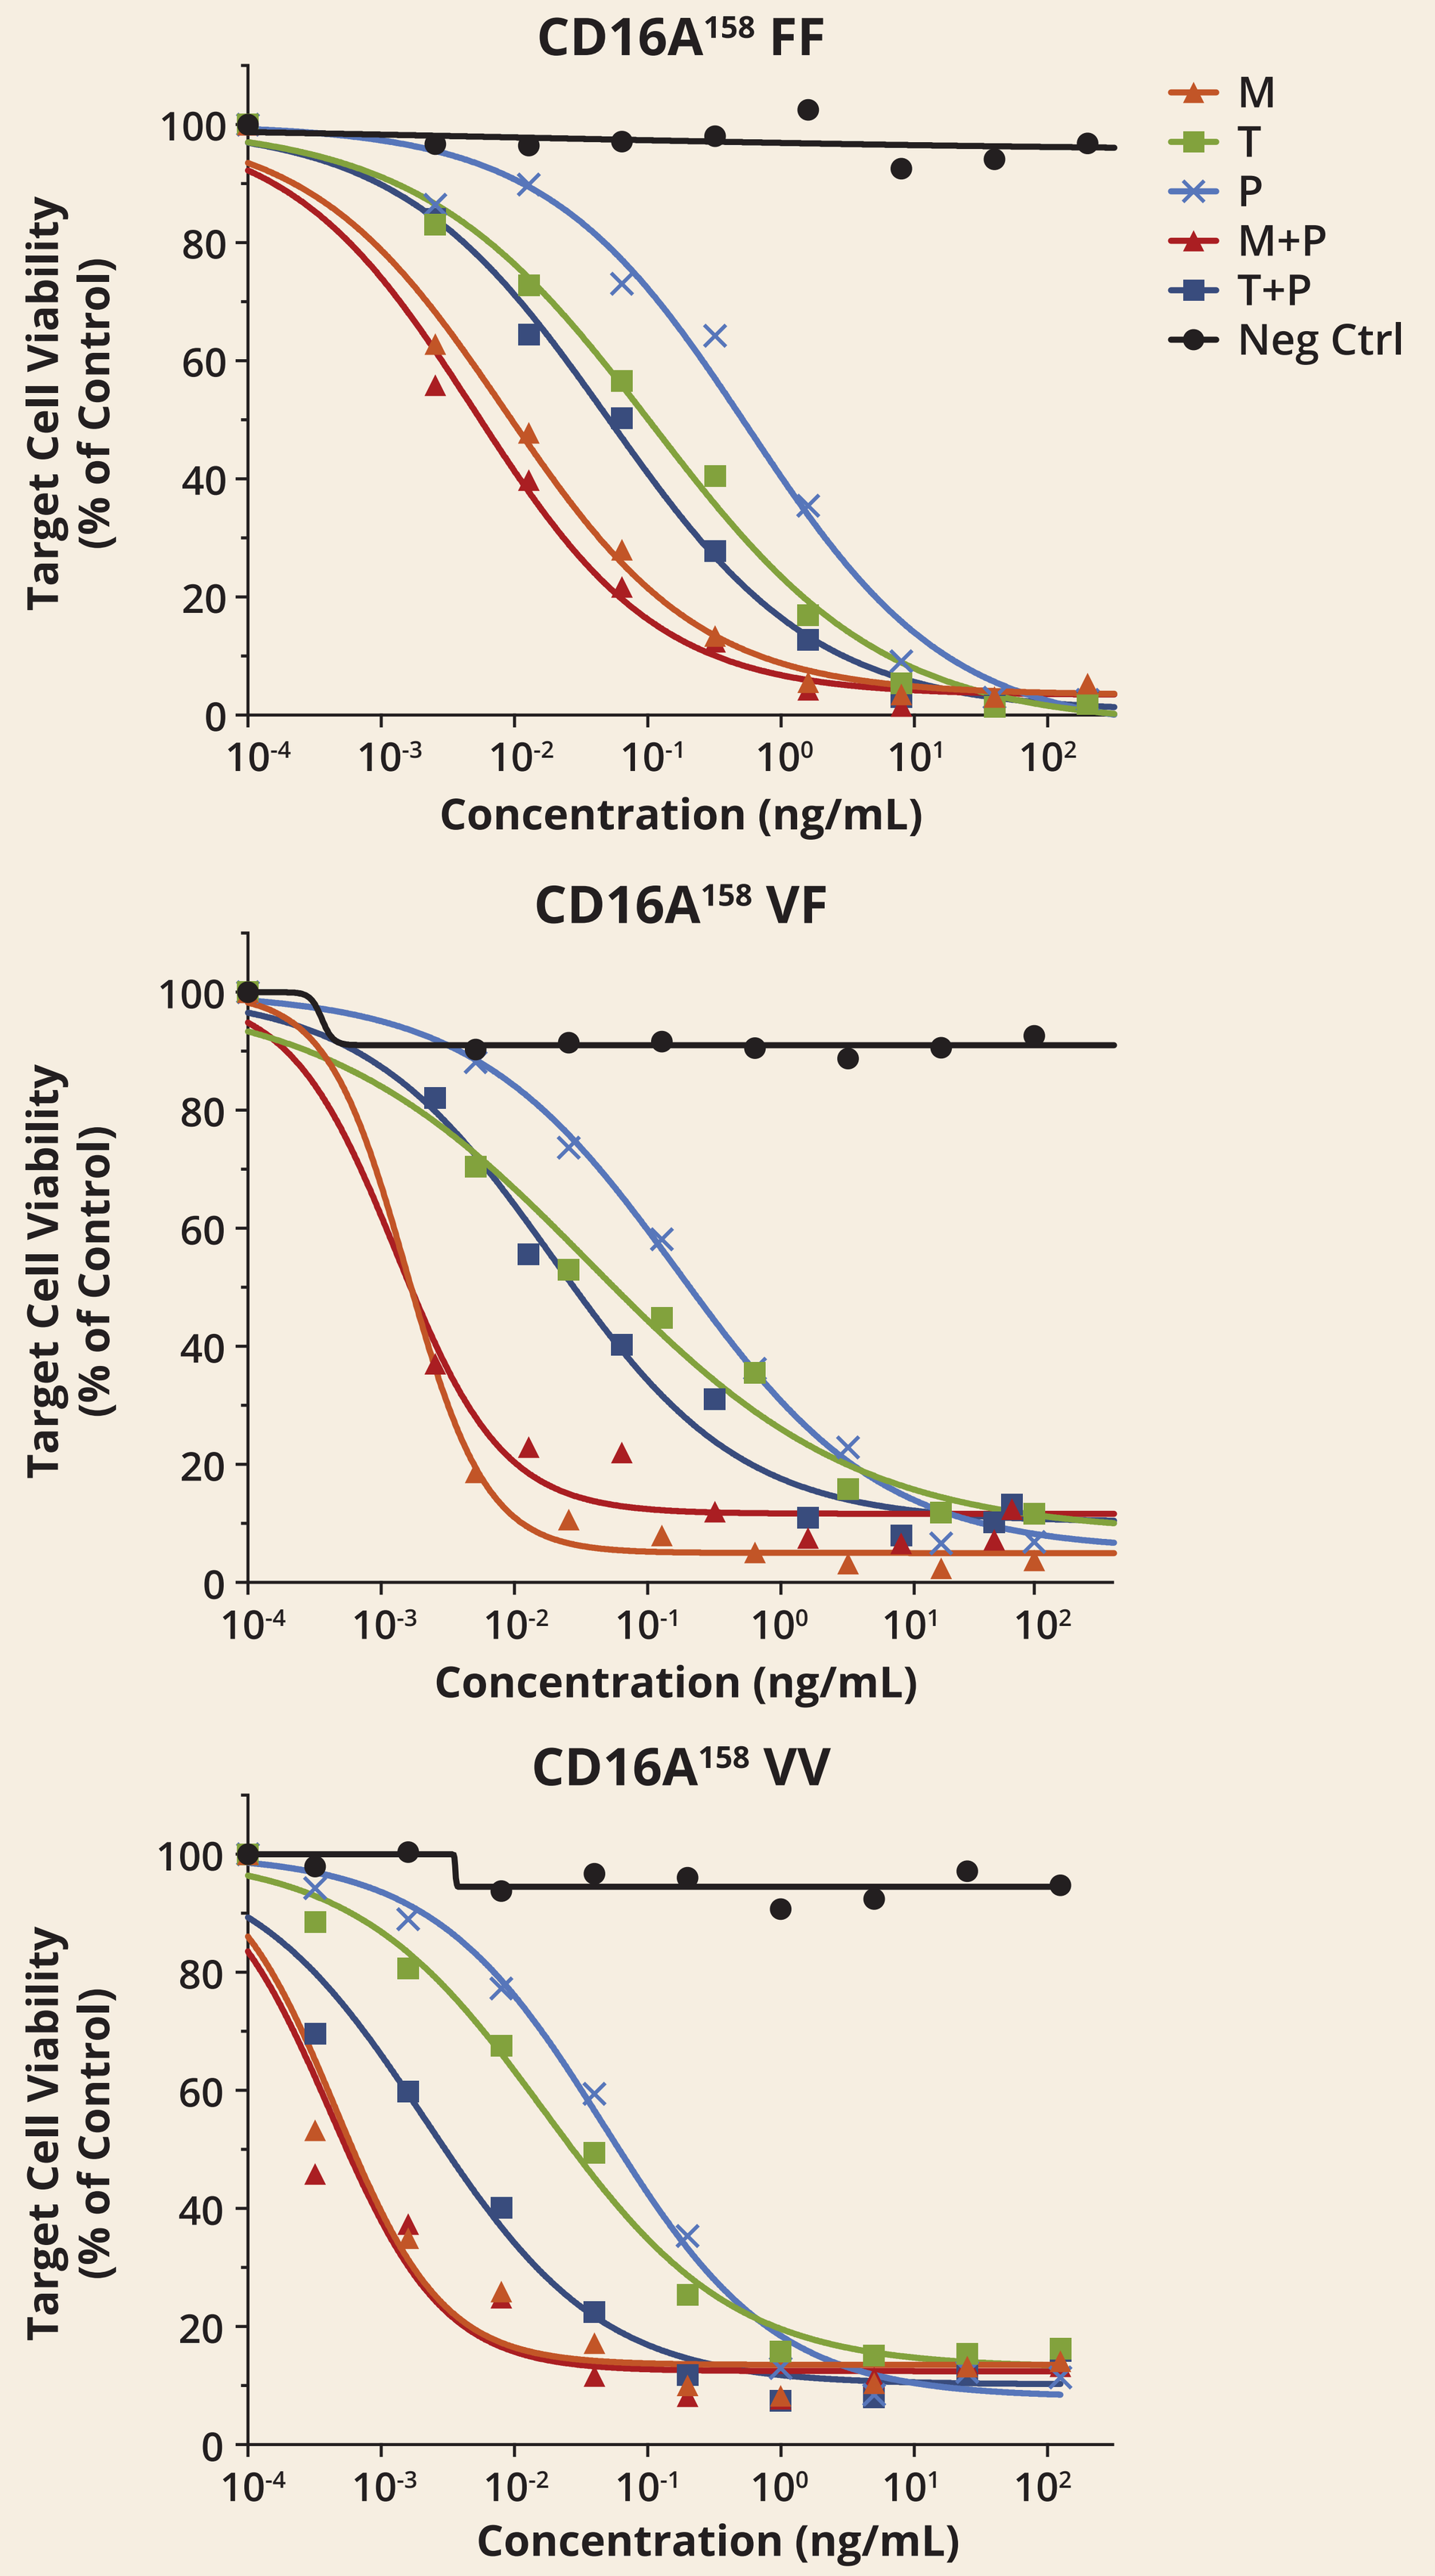

CD16A158 FF
CD16A158 VF
CD16A158 VV
M
T
P
M+P
T+P
Neg Ctrl
Target Cell Viability (% of Control)
Concentration (ng/mL)

ADCC EC_{50} Values Obtained with NK Cells of Different CD16A Genotypes

- For each NK cell donor:
 - M is always more potent (lower EC50) than T, which is always more potent than P
 - Combination of M + P is always more potent than combination of T + P
- Similar results obtained with N87/GF and JIMT-1/GF target cells

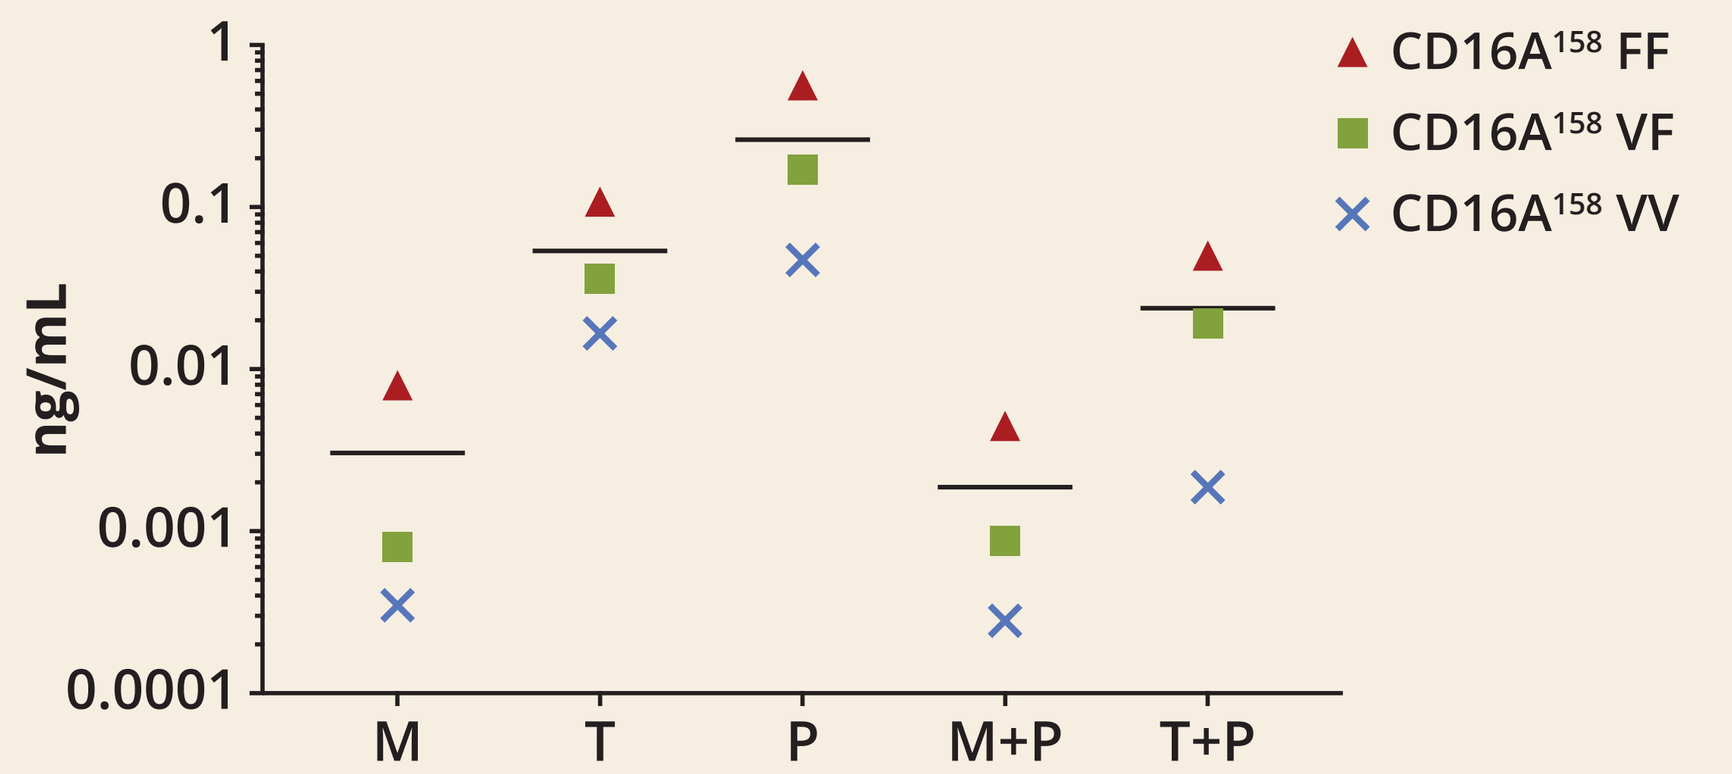

CD16A158 FF
CD16A158 VF
CD16A158 VV
ng/mL
M
T
P
M+P
T+P

Black bar = mean EC_{50} value.

Margetuximab (M) Induces Greater Activation and Proliferation of NK Cells than Trastuzumab (T)

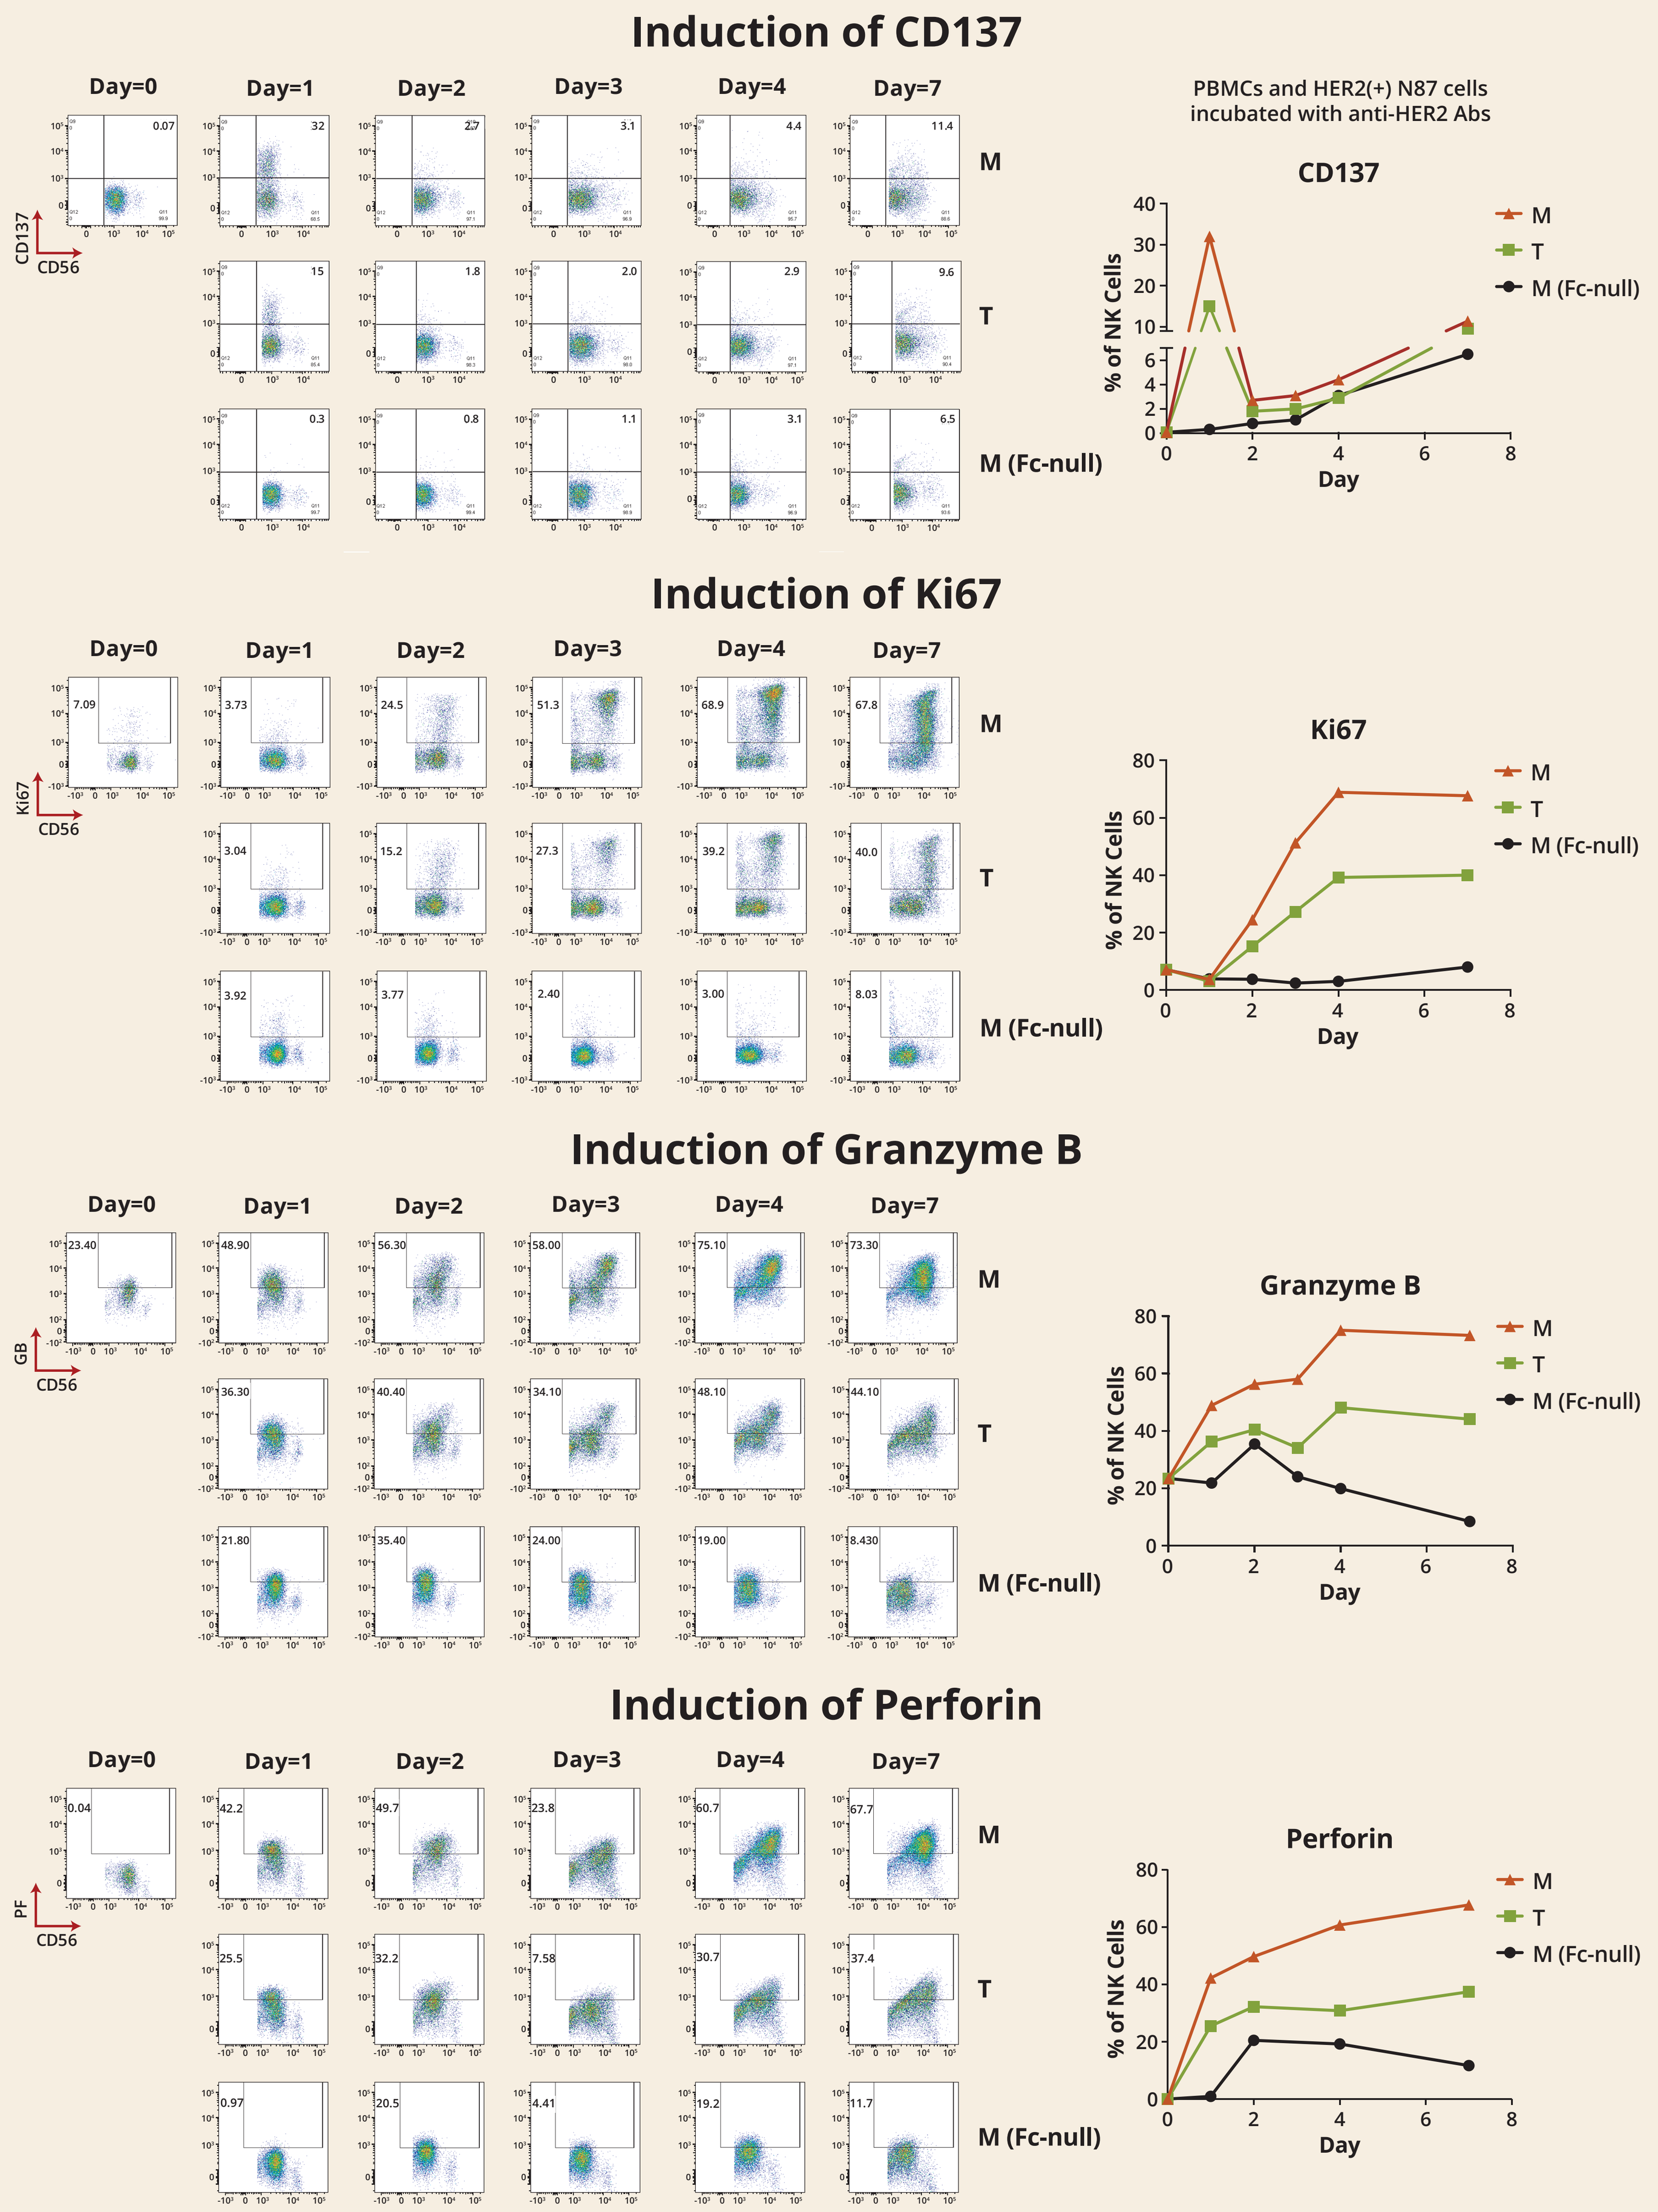

Induction of CD137
Day=0
Day=1
Day=2
Day=3
Day=4
Day=7
M
T
M (Fc-null)
CD137
CD56
PBMCs and HER2(+) N87 cells incubated with anti-HER2 Abs
CD137
% of NK Cells
Day
Induction of Ki67
Ki67
Induction of Granzyme B
GB
Granzyme B
Induction of Perforin
PF
Perforin

Conclusions

Binding Properties	CD16A (activating FcγR)	M > T
	CD32B (inhibitory FcγR)	M < T
	HER2 (in absence or presence of P)	M ≈ T
Fc-independent Activities	HER2 ECD shedding inhibition	M ≈ T
	Anti-proliferation (in absence or presence of EGF or heregulin)	M ≈ T M+P ≈ T+P
Fc-dependent Activities	ADCC (with effector cells of all $CD16A^{158}$ genotypes)	M > T > P M+P > T+P
	NK cell activation and expansion	M > T

- Margetuximab mediates ADCC in vitro with greater potency than trastuzumab and promotes greater NK cell activation and expansion
- Combination of margetuximab + pertuzumab mediates ADCC in vitro with greater potency than combination of trastuzumab + pertuzumab

Presented at the American Association for Cancer Research Annual Meeting 2019, March 29–April 3, 2019, Atlanta, GA

©2019 MacroGenics, Inc. All rights reserved.